Discovery of activities via statistical clustering of fixation patterns

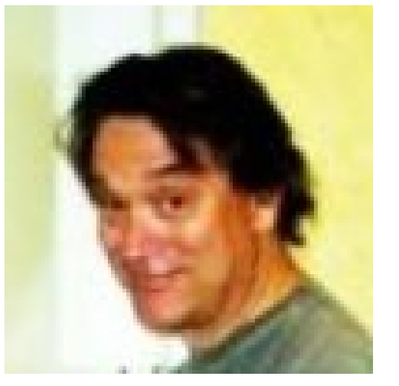

Jeffrey B. Mulligan, Human Systems Integration Division, NASA Ames Research Center

Can we infer activities from eye movements?

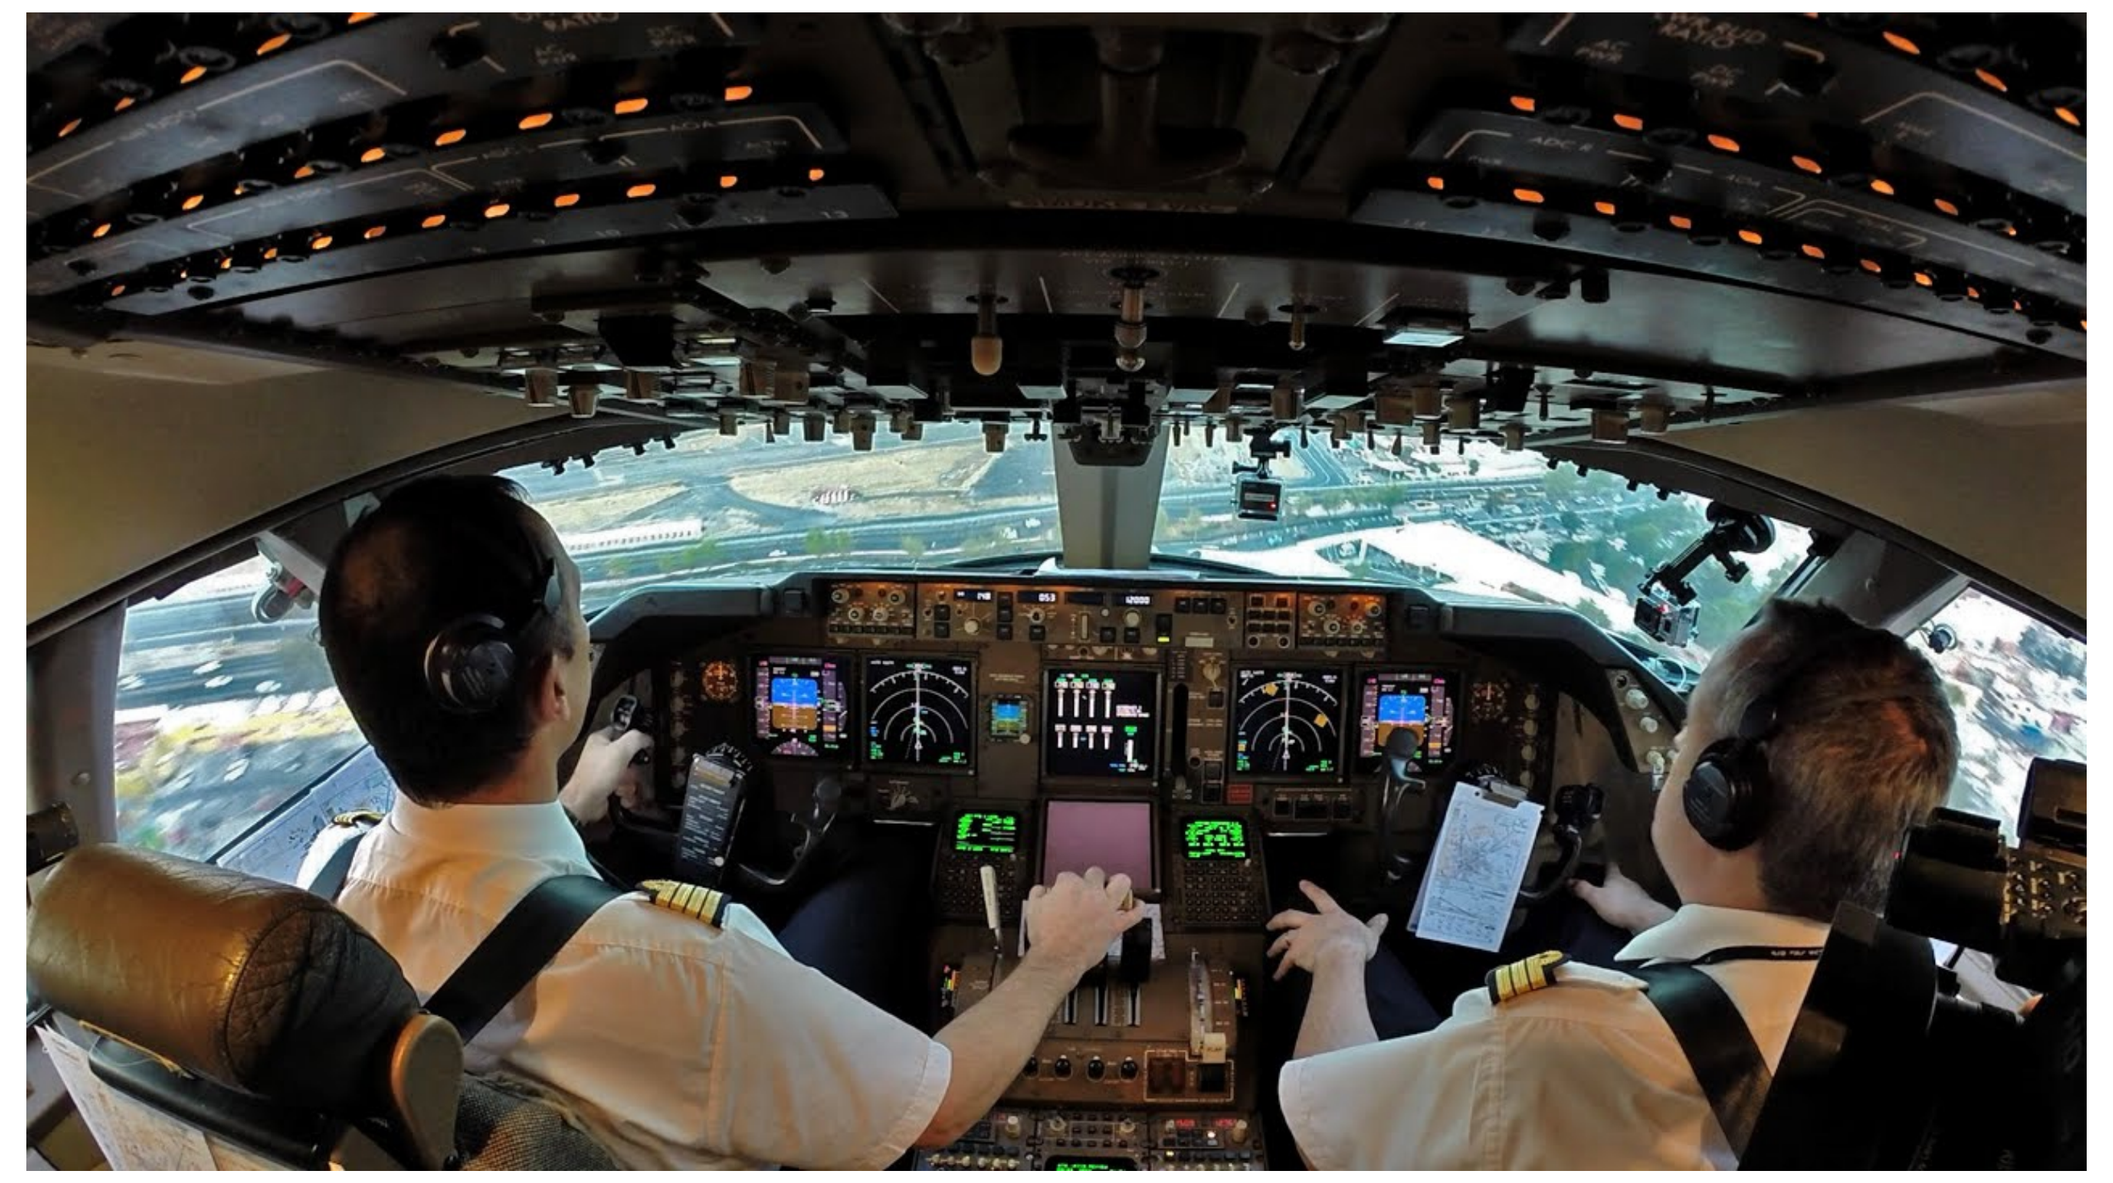

Image of Boeing 747 cockpit from http://www.worldairroutes.com/AF747.html

Can we infer pilot activities from scan path data? We would like to be able to know whether the pilots are paying attention to the energy state of the aircraft, and other safety-critical parameters.

The raw data: symbol time series, of Area-Of-Interest (AOI) labels

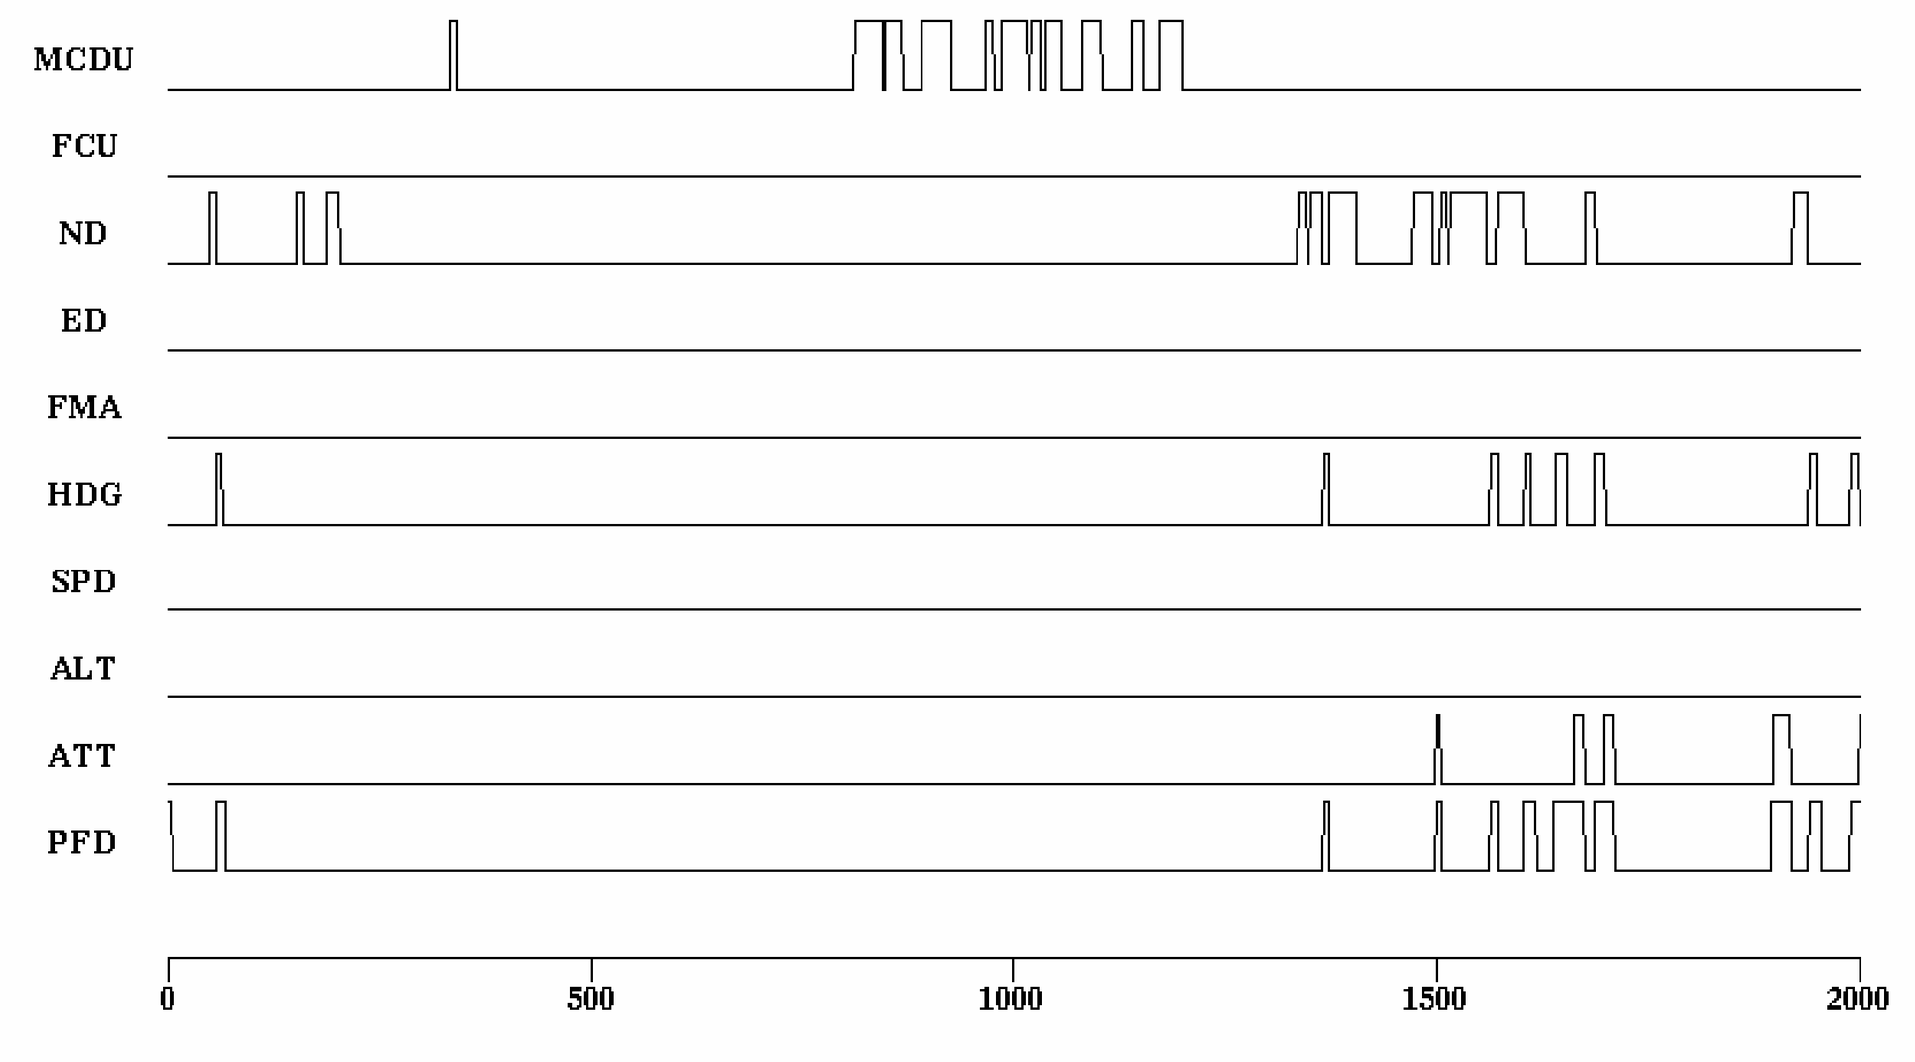
MCDU
FCU
ND
ED
FMA
HDG
SPD
ALT
ATT
PFD
0
500
1000
1500
2000
frame index

The raw data can be represented as a matrix of time-series data, with one row for each AOI. A value of 1 indicates a fixation at the corresponding AOI at a given time. Averaging or "blurring" in the time domain transforms the columns to proportions of time spent at each AOI during the integration window.

Modeling activities as a first-order Markov process

$$P(\ s(t+1) = i \mid s(t) = j\) = m_{ij}$$

$$P(\ s(t+2) = i \mid s(t) = j\) = \sum_{k=1}^{N} m_{ik} m_{kj}$$

Examples of a random activity with 10 AOIs

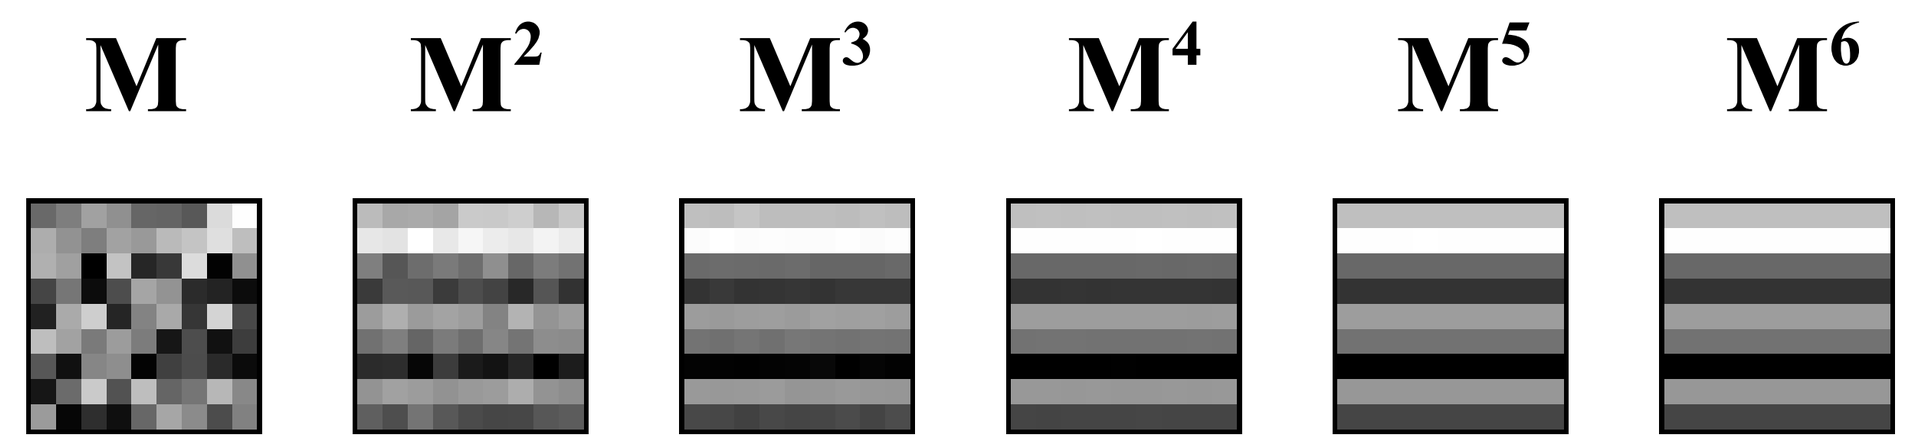
M
M2
M3
M4
M5
M6

Clustering results for three window sizes

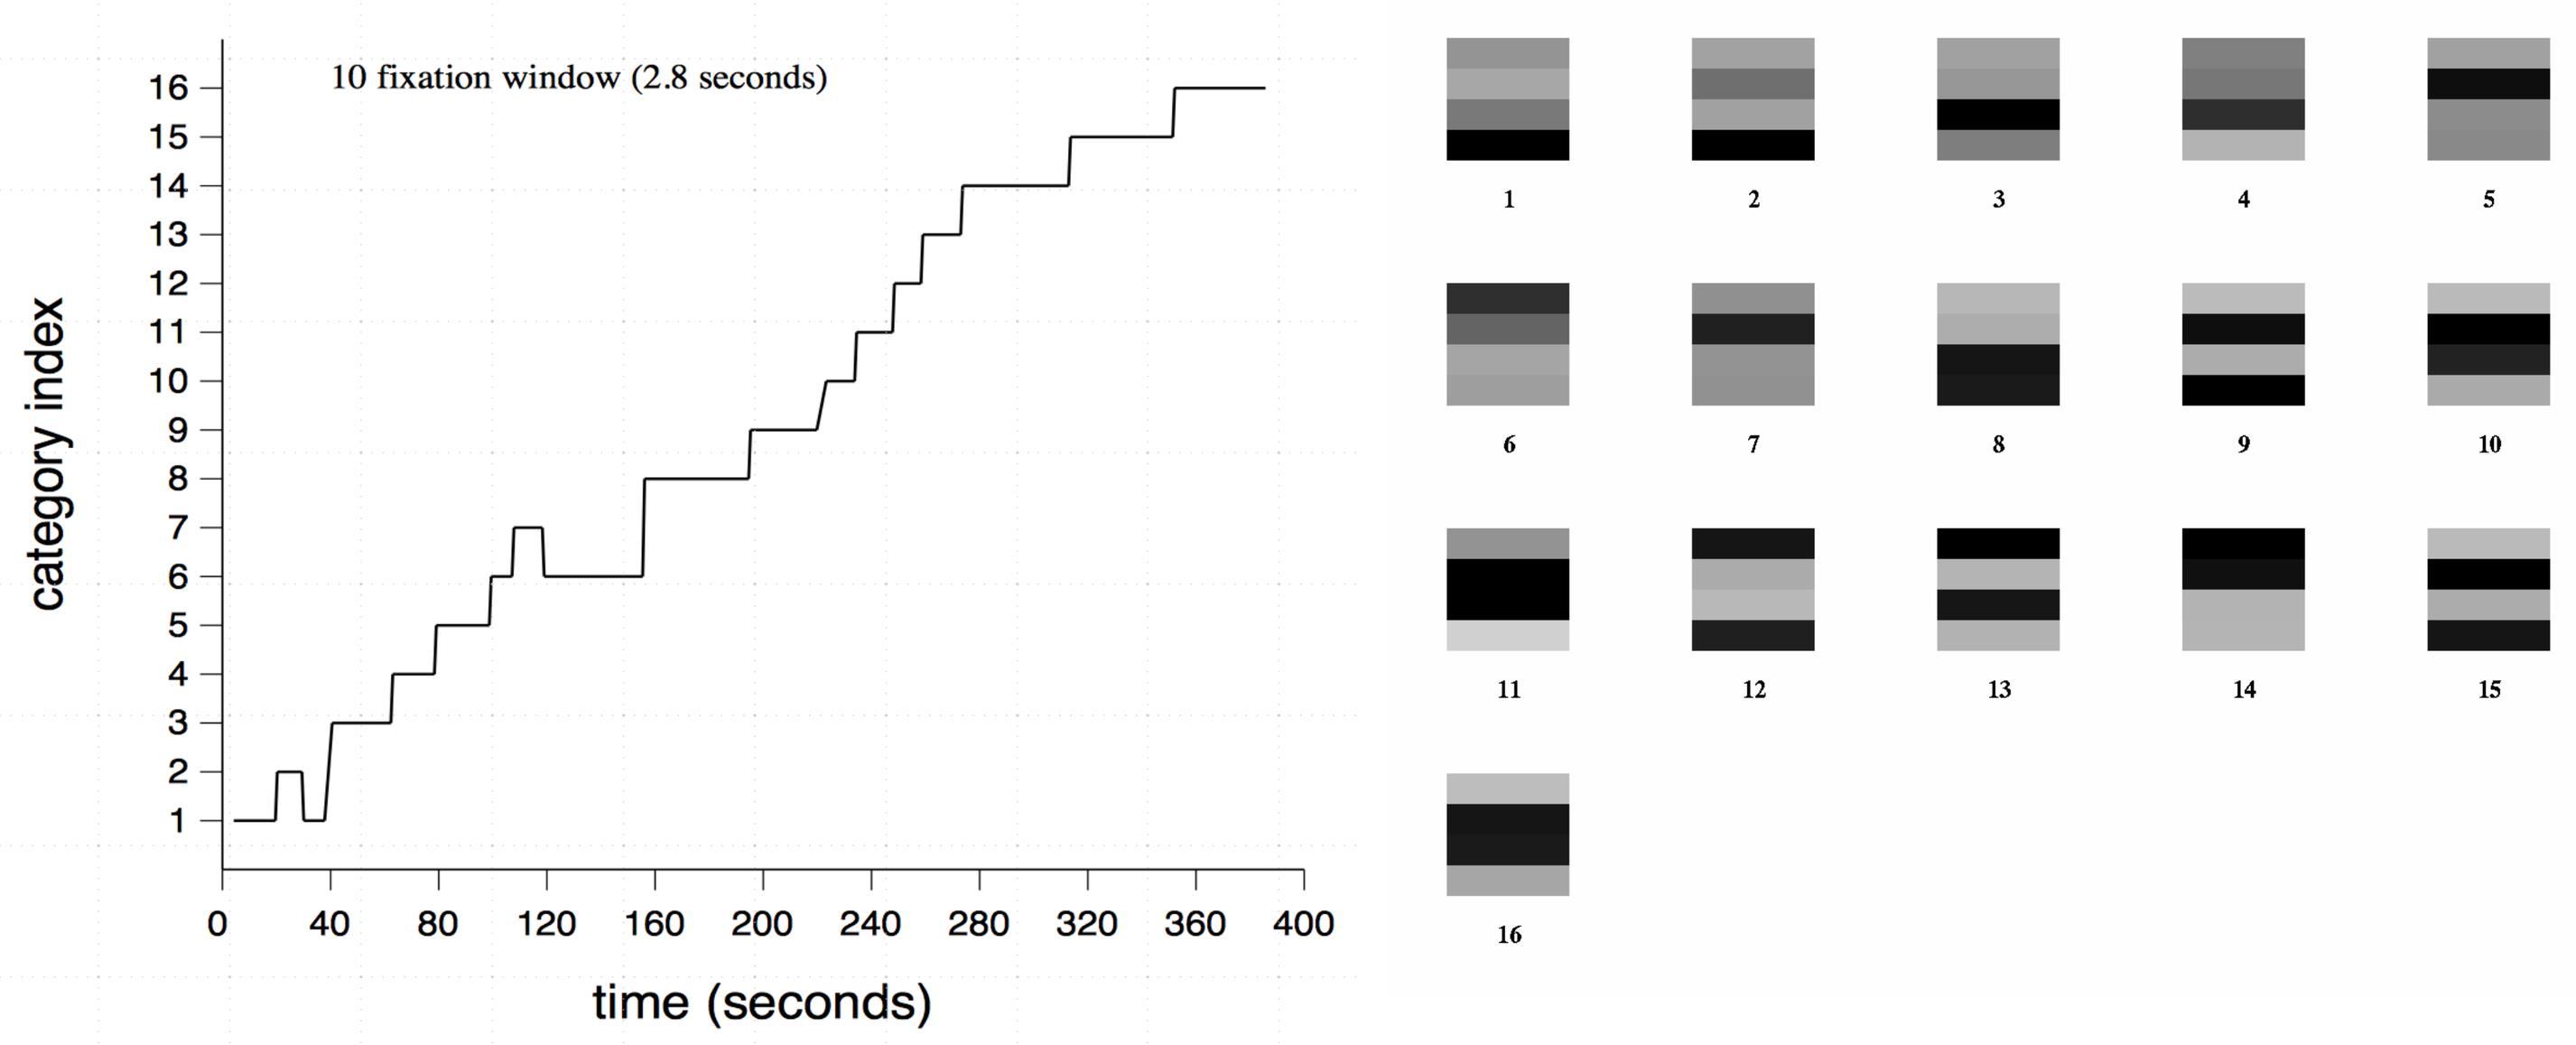
10 fixation window (2.8 seconds)
category index
time (seconds)

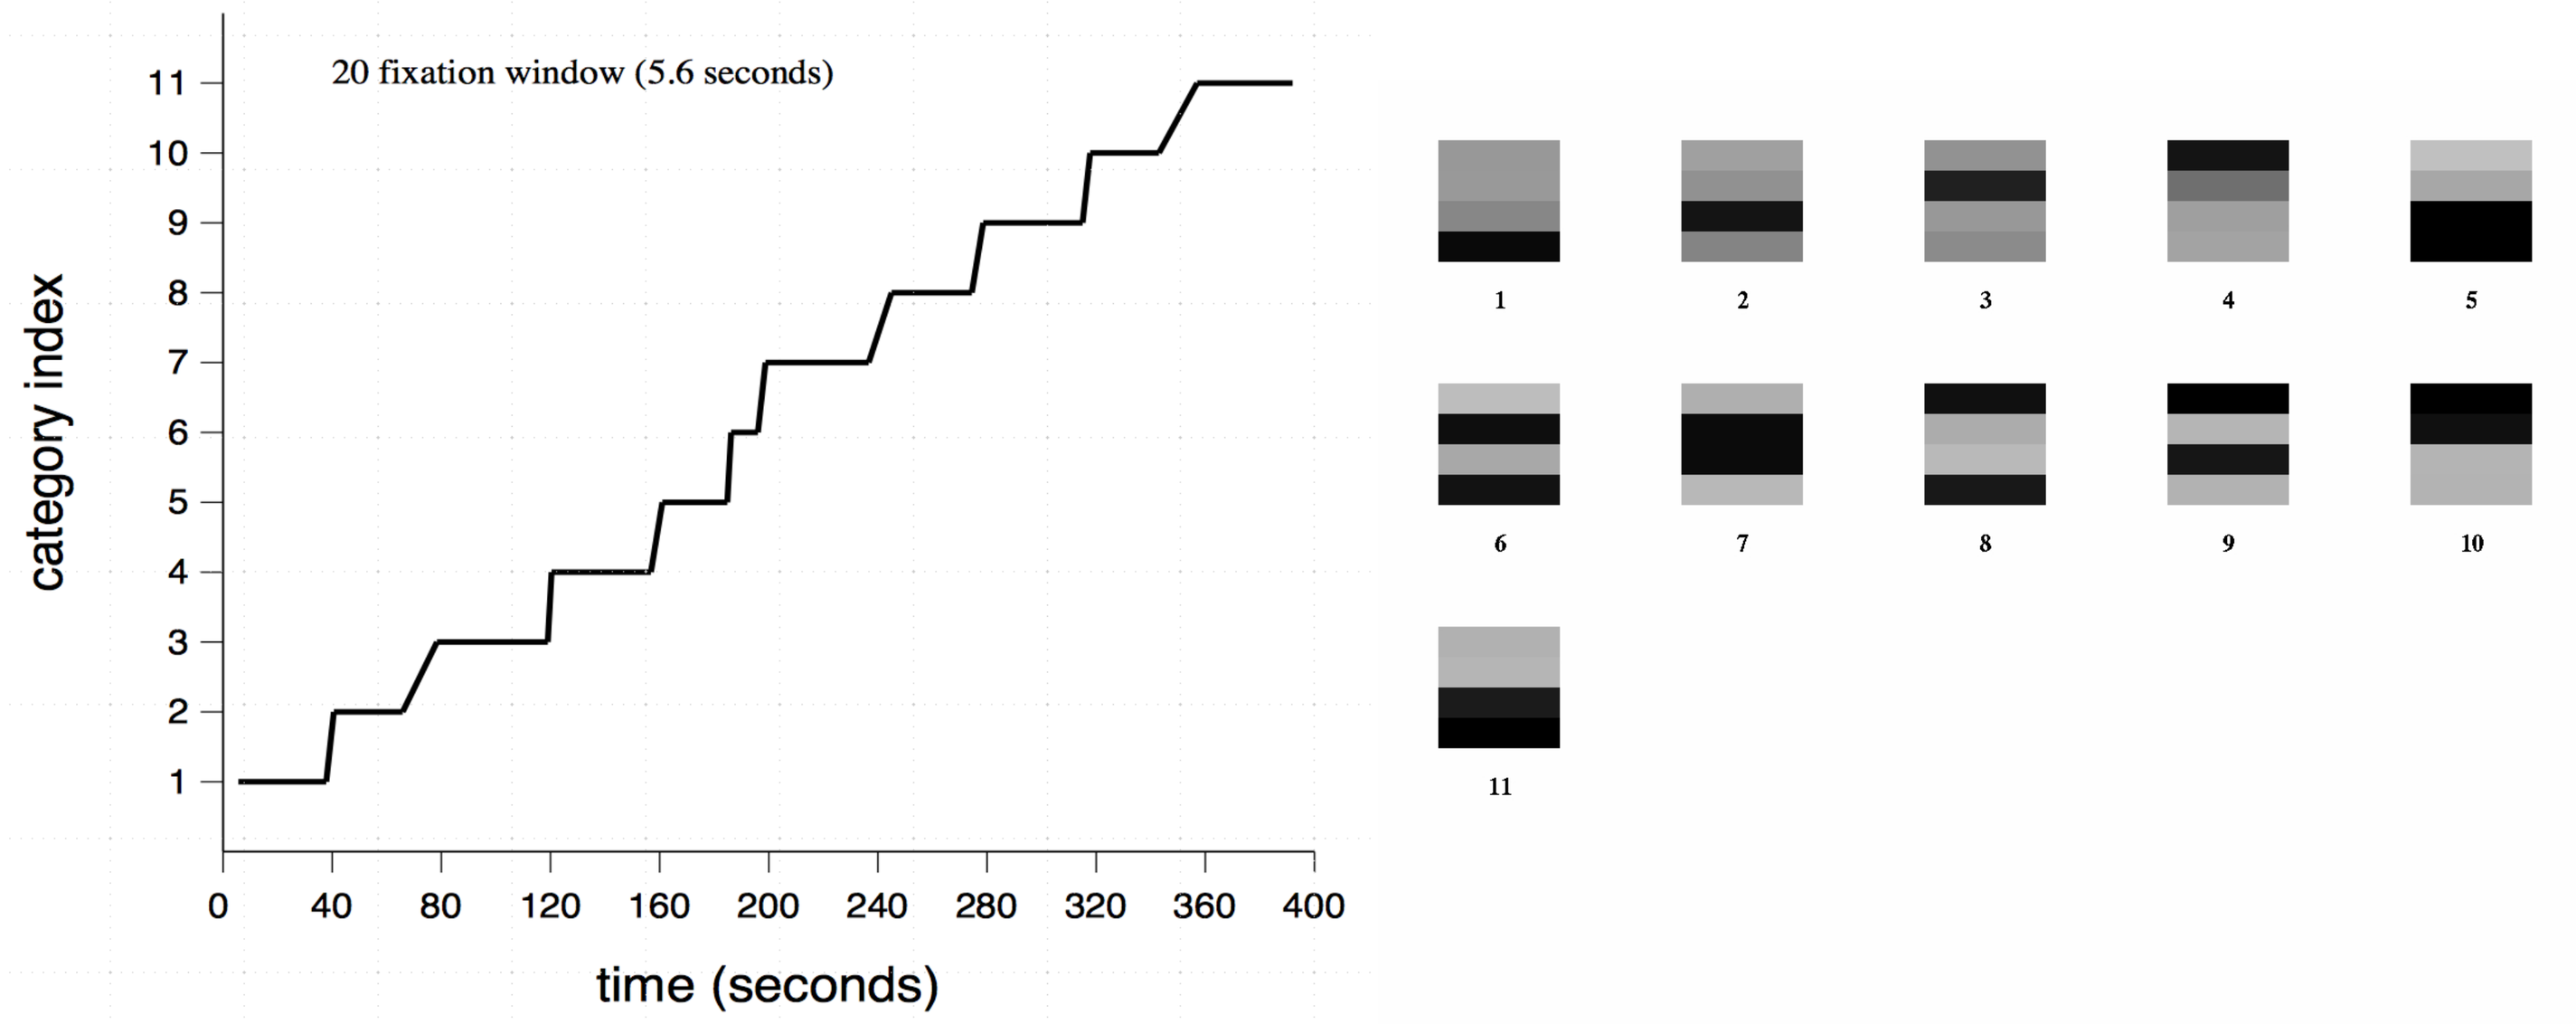
20 fixation window (5.6 seconds)
category index
time (seconds)

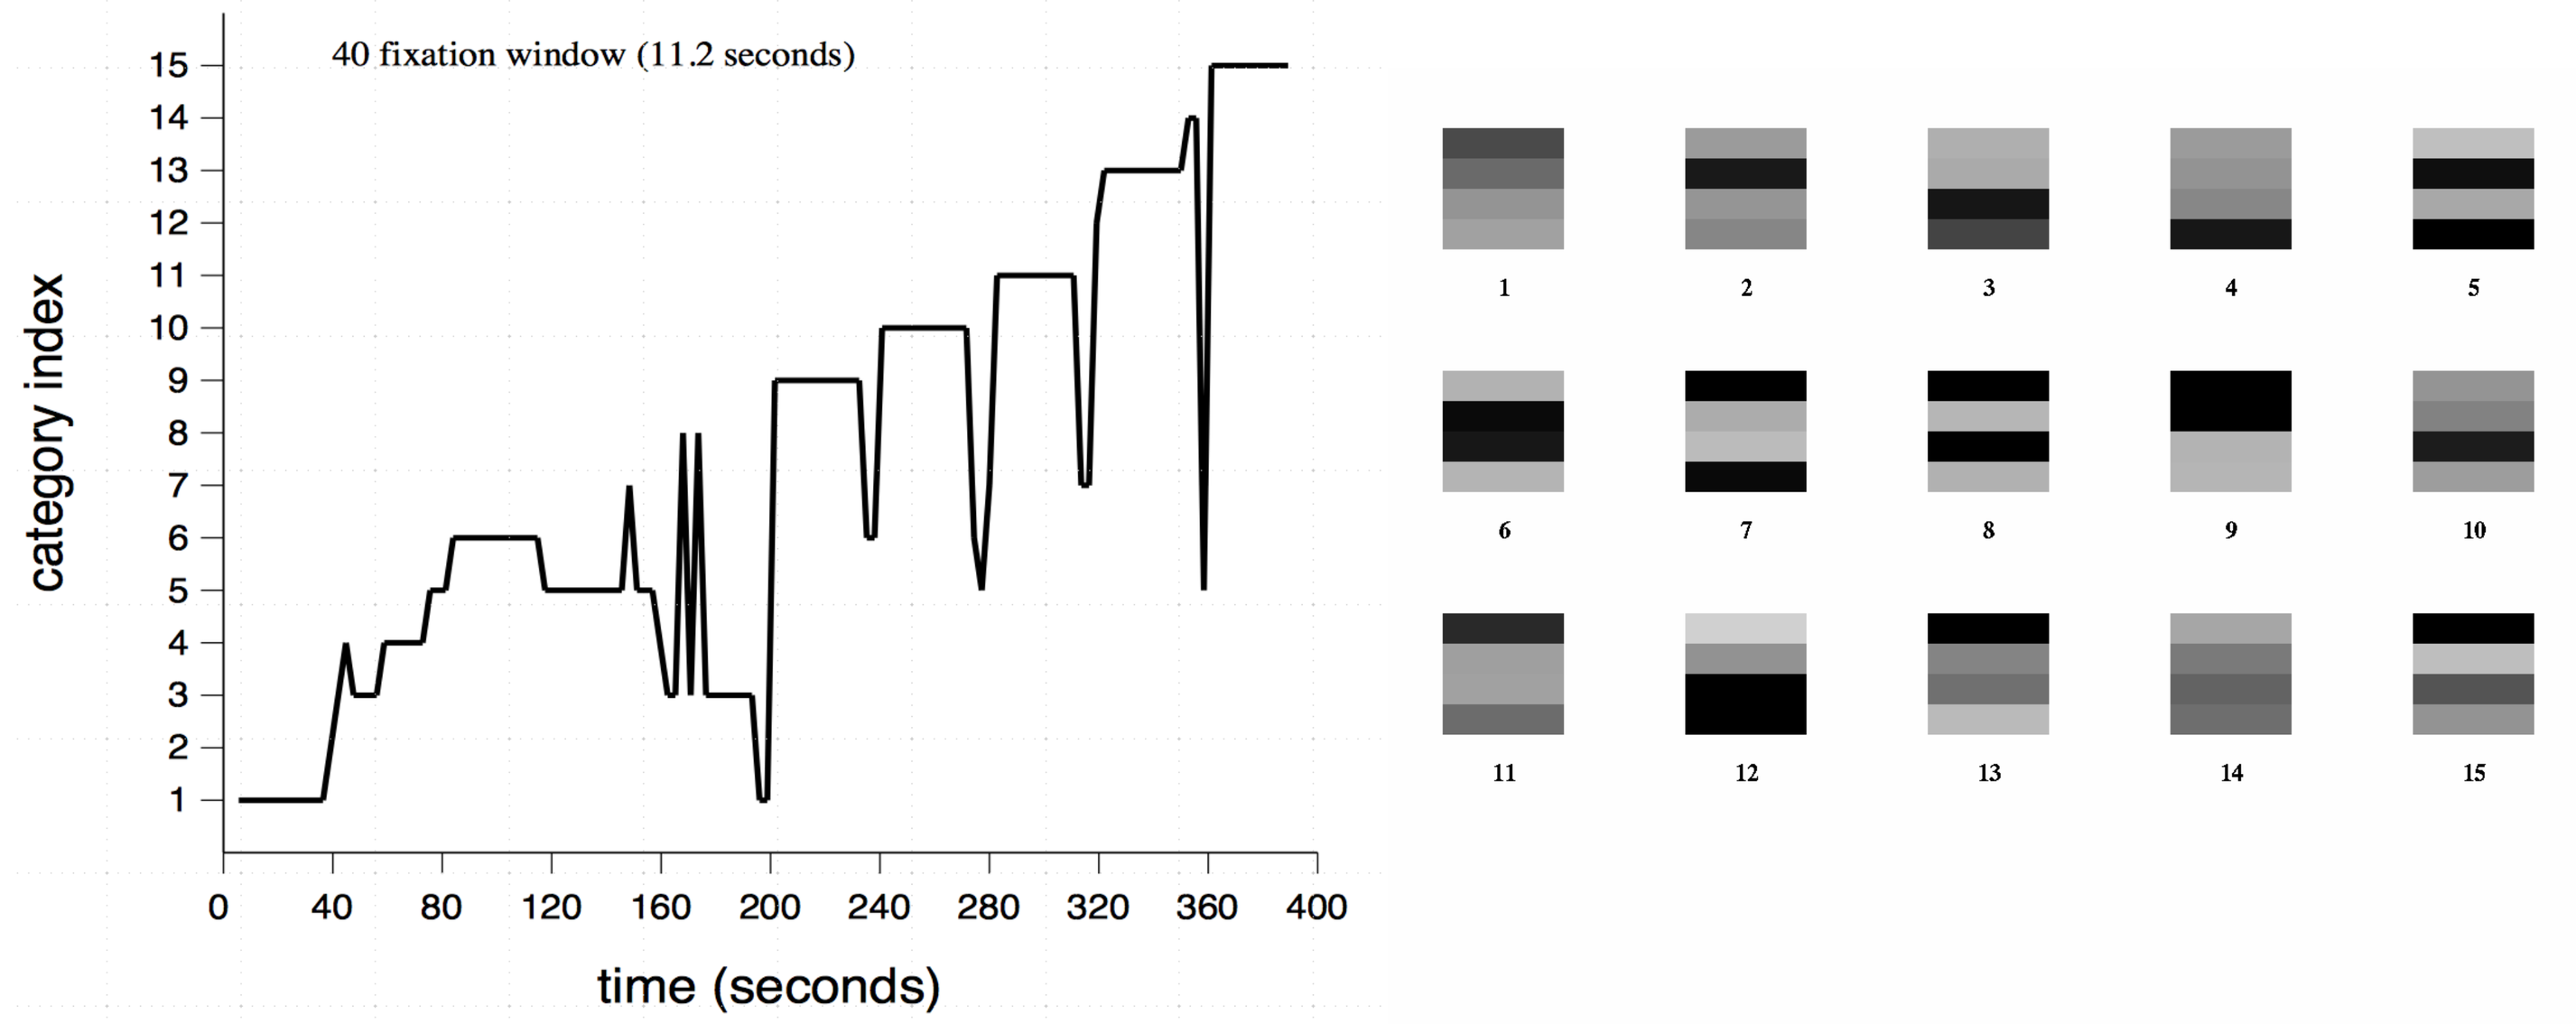
40 fixation window (11.2 seconds)
category index
time (seconds)

Creating stimuli with balanced transition statistics

The bottom panel on the left illustrates that when a broad averaging window is used, samples near transition points can be erroneously classified as another activity (which is a mixture of the two activities on either side of the transition). In the simulations shown here, the activities were presented in a fixed sequence; thus, of all of the 45 possible (unordered) transitions, only 9 were represented. We ask the question, is it possible to devise a sequence so that every possible transition is sample once, and only once? While it may be a distracting tangential issue for the current work, it may be of value for experiments studying sequential dependencies of stimuli.

For this particular case, we can represent each of the 10 activities as a node in a fully-connected graph. We wish to find a traversal of the graph that visits each edge once and only once. As was shown by Euler, there is no solution to this problem, because there are more than two nodes with an odd number of edges. However, it *is* possible to construct a circuit that traverses each edge *twice,* even if it is required that each edge be traversed in each of the two possible directions! The solution can be extended to uniform sampling of all possible tri-grams, etc.

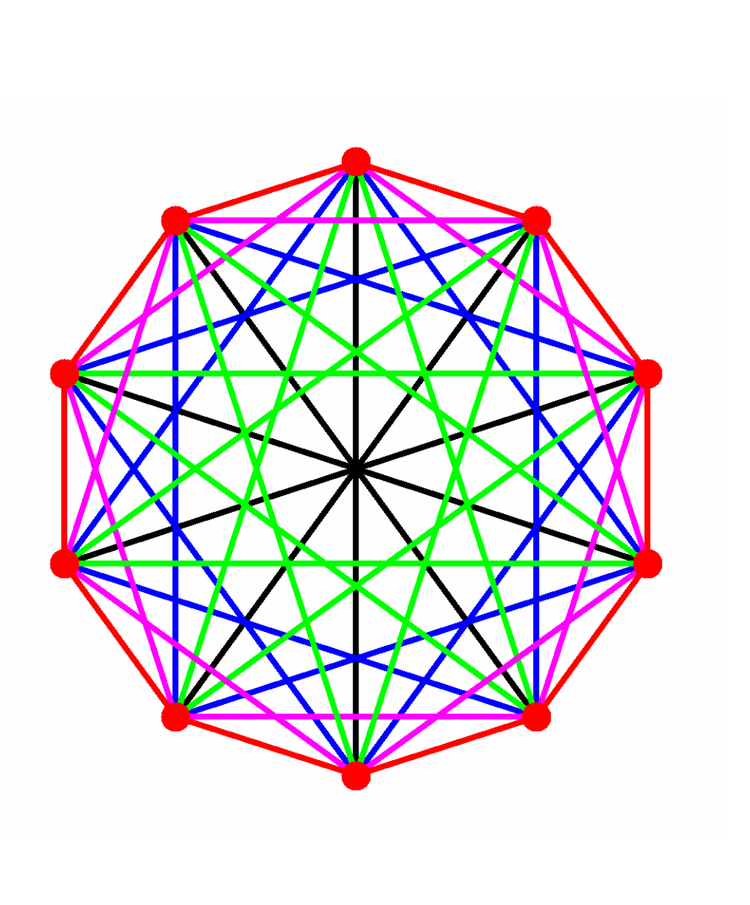

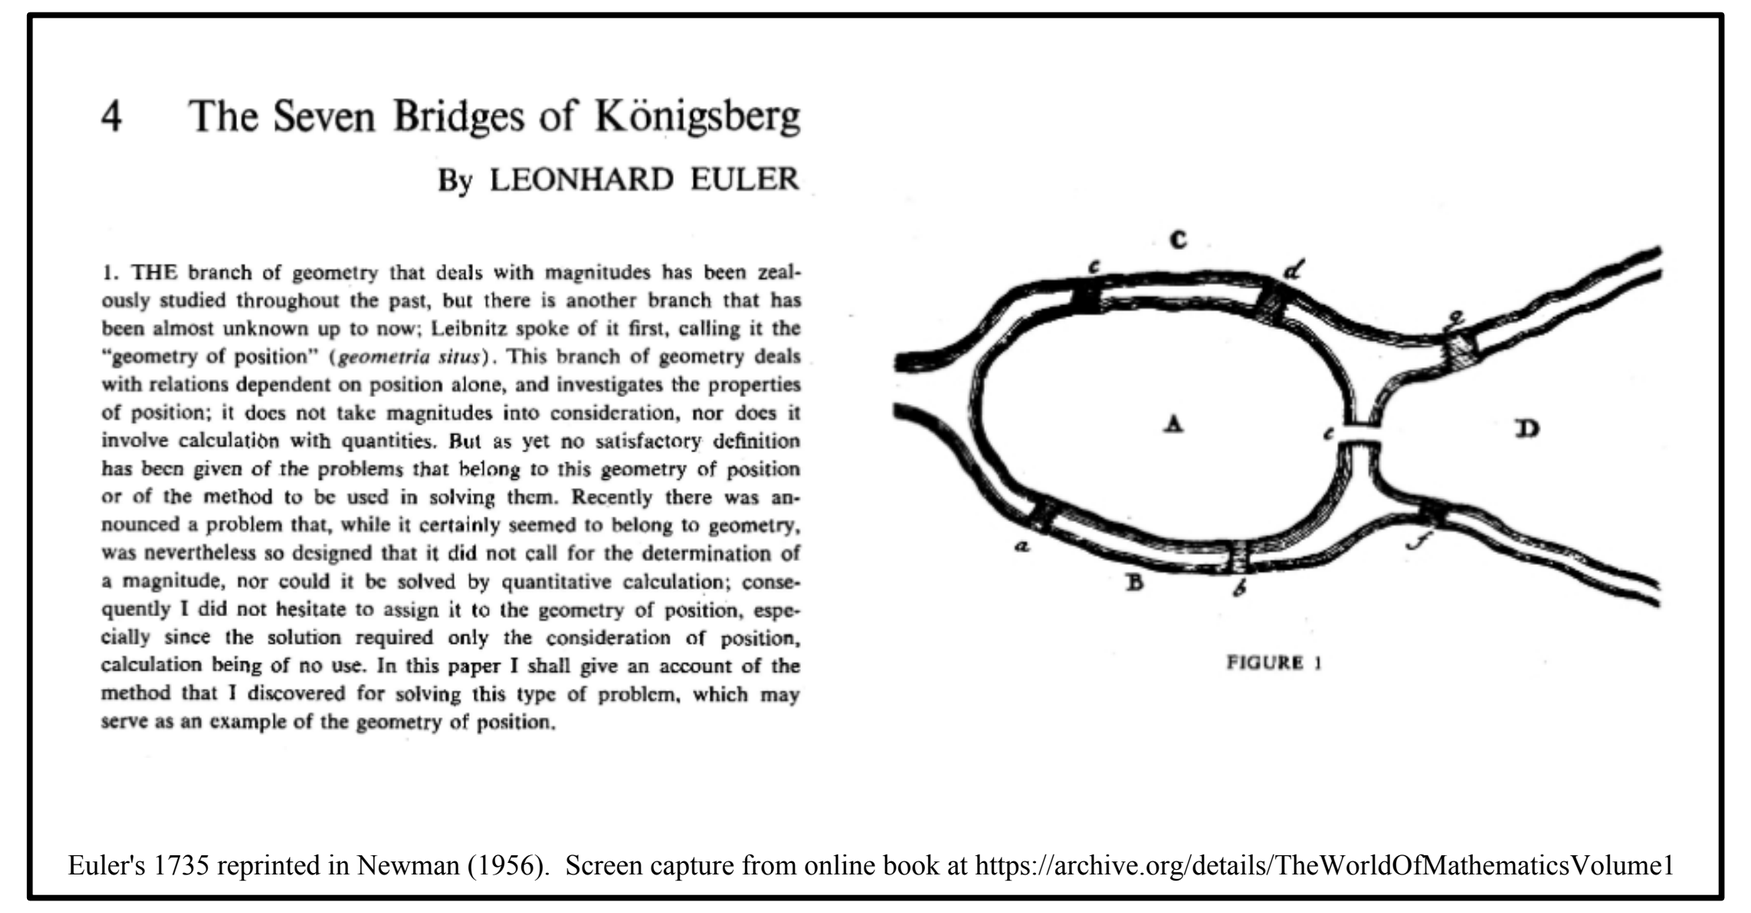

4 The Seven Bridges of Königsberg

By LEONHARD EULER

1. THE branch of geometry that deals with magnitudes has been zealously studied throughout the past, but there is another branch that has been almost unknown up to now; Leibnitz spoke of it first, calling it the "geometry of position" (*geometria situs*). This branch of geometry deals with relations dependent on position alone, and investigates the properties of position; it does not take magnitudes into consideration, nor does it involve calculation with quantities. But as yet no satisfactory definition has been given of the problems that belong to this geometry of position or of the method to be used in solving them. Recently there was announced a problem that, while it certainly seemed to belong to geometry, was nevertheless so designed that it did not call for the determination of a magnitude, nor could it be solved by quantitative calculation; consequently I did not hesitate to assign it to the geometry of position, especially since the solution required only the consideration of position, calculation being of no use. In this paper I shall give an account of the method that I discovered for solving this type of problem, which may serve as an example of the geometry of position.

FIGURE 1

Euler's 1735 reprinted in Newman (1956). Screen capture from online book at https://archive.org/details/TheWorldOfMathematicsVolume1

Eye movement patterns reflect task demands!

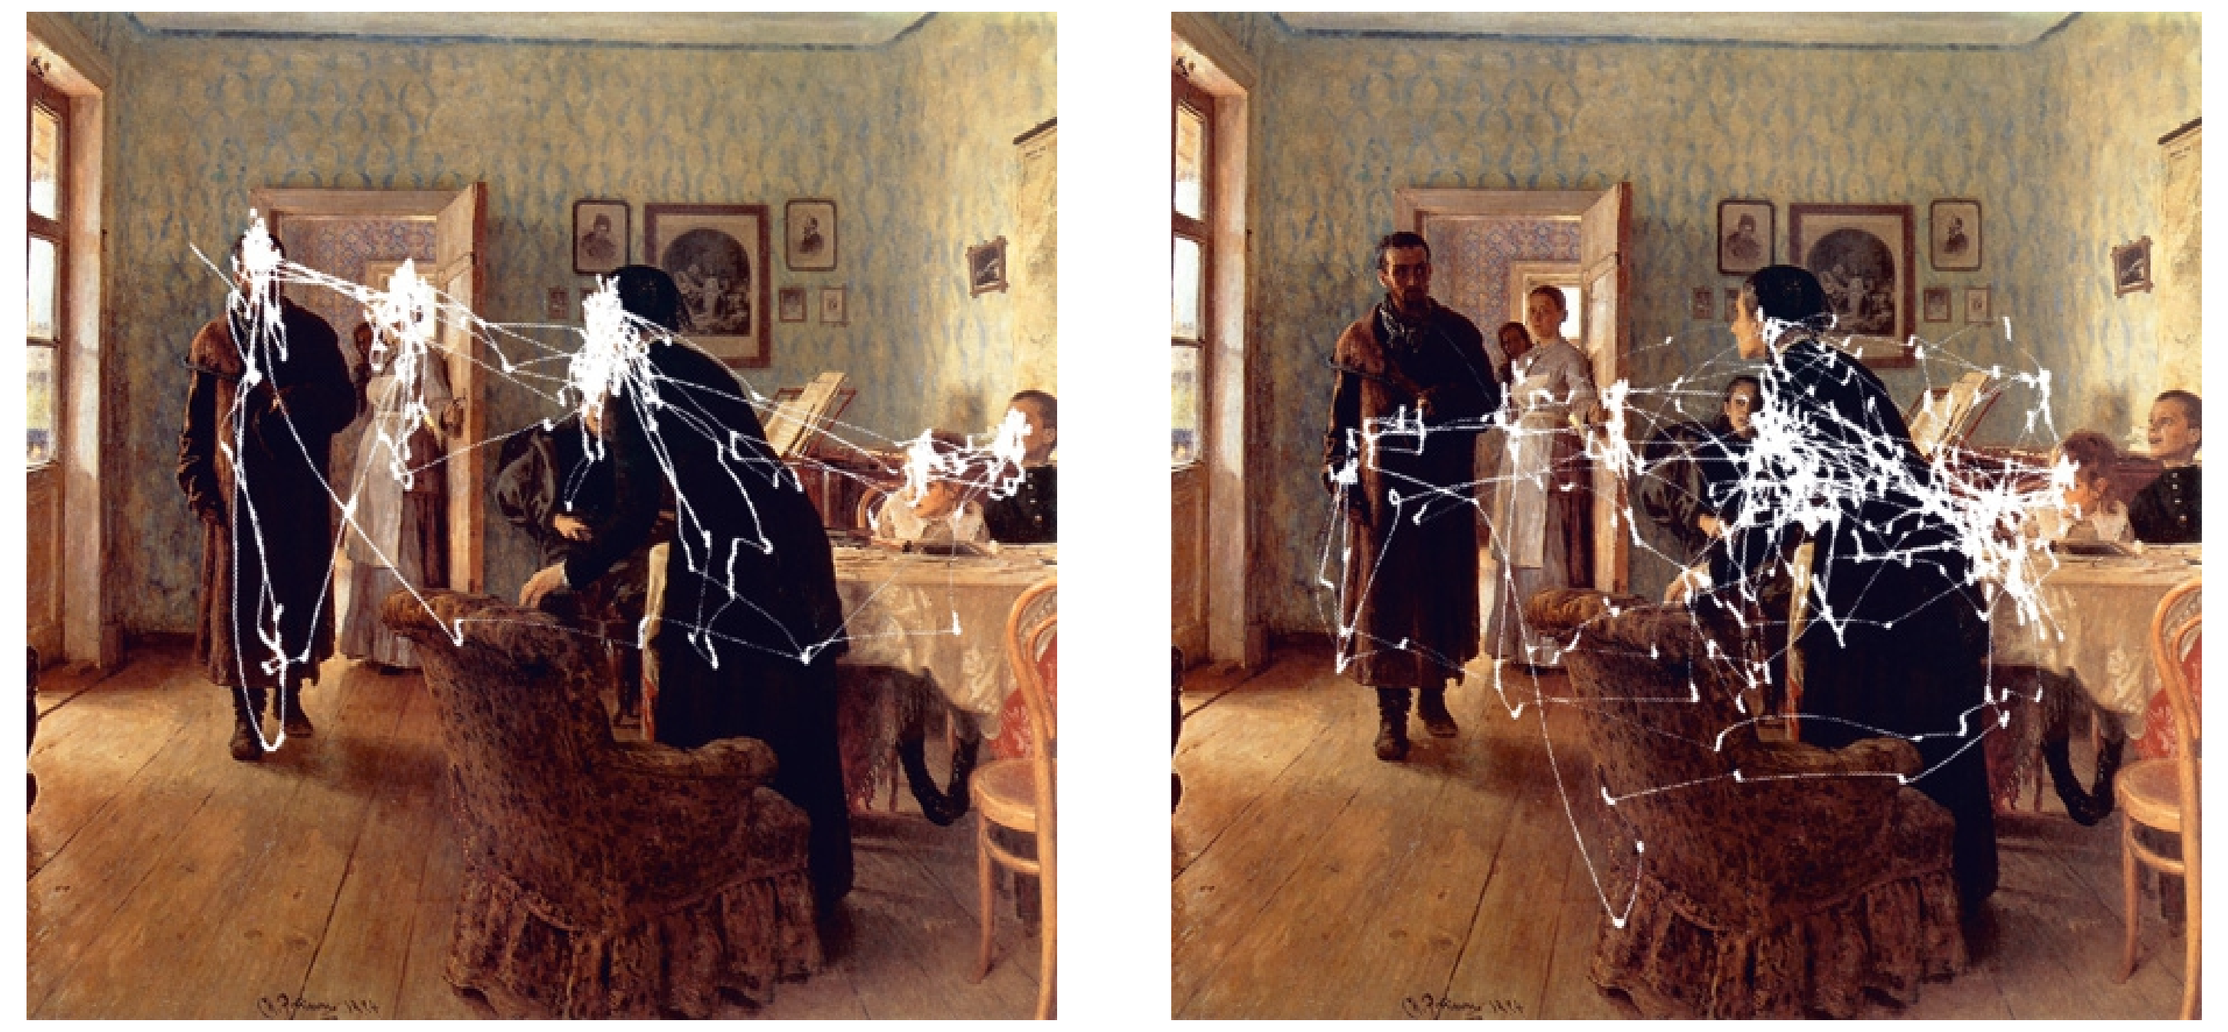

Images depicting data from Yarbus (1967), downloaded from http://www.cabinetmagazine.org/issues/30/archibald.php

Classic data from Yarbus showing different scan patterns resulting from observer instructions.

The image on the left shows data resulting when the observer was instructed to estimate the ages of the subjects, while on the right the instructions were to estimate the material circumstances.

Previous approaches

Measure	Resampling	Quantization	Simplified?	Truncated?	Preserves temporal ordering?	Target scanpath variable
String edit	No	Grid	No	No	Yes	Position, Sequence
ScanMatch	Yes	Grid	No	No	Yes	Position, Duration, Sequence
Overlap	Yes	Radius	No	Yes	No	Sequence, Position
Correlate	Yes	Direct	No	Yes	Yes	Position, Sequence
Gaze shift	Yes	Direct	No	Yes	Yes	Amplitude, Sequence
Linear distance	No	Direct	No	No	No	Position
MM vector	No	Direct	Yes	No	Yes	Shape
MM direction	No	Direct	Yes	No	Yes	Saccade Direction
MM length	No	Direct	Yes	No	Yes	Saccade Length
MM position	No	Direct	Yes	No	Yes	Position
MM duration	No	Direct	Yes	No	Yes	Duration
Recurrence	No	Radius	No	Yes	No	Position
Determinism	No	Radius	No	Yes	No	Fixation Trajectories
Laminarity	No	Radius	No	Yes	No	Fixation Persistence
Corm	No	Radius	No	Yes	Yes	Leading/Following

Table from Anderson et al. (1967)

A method is desired that can be applied to area-of-interest (AOI) labels, targeting position and duration, that is insenstive to temporal order, and is grounded in statistics.

Comparison using the chi-square statistic

	2F57	2C57	total	frac.	expected	
PFD	12	13	25	0.338	8.108	16.892
ATT	1	2	3	0.041	0.973	2.027
ALT	3	6	9	0.122	2.919	6.081
HDG	3	1	4	0.054	1.297	2.703
FMA	4	2	6	0.081	1.946	4.054
ND	0	22	22	0.297	7.135	14.865
SP	1	0	1	0.014	0.324	0.676
Wind	0	4	4	0.054	1.297	2.703
total	24	50	74			

$$s = \sum_i \frac{(o_i - e_i)^2}{e_i} = 23.85 \qquad p = 0.0012$$

The chi-square test assesses the null hypothesis that two sets of counts were produced by a single process characterized by a set of probabilities for each event (AOI fixation). For each AOI, the fraction of the total fixations that are made to that AOI is computed. These fractions are used to compute the expected number of fixations to each AOI within each set, by multiplying the fraction times the total number of fixations in the set. The statistic is large when the observed values have large deviations from the values expected under the null hypothesis.

A simple model of human behavior

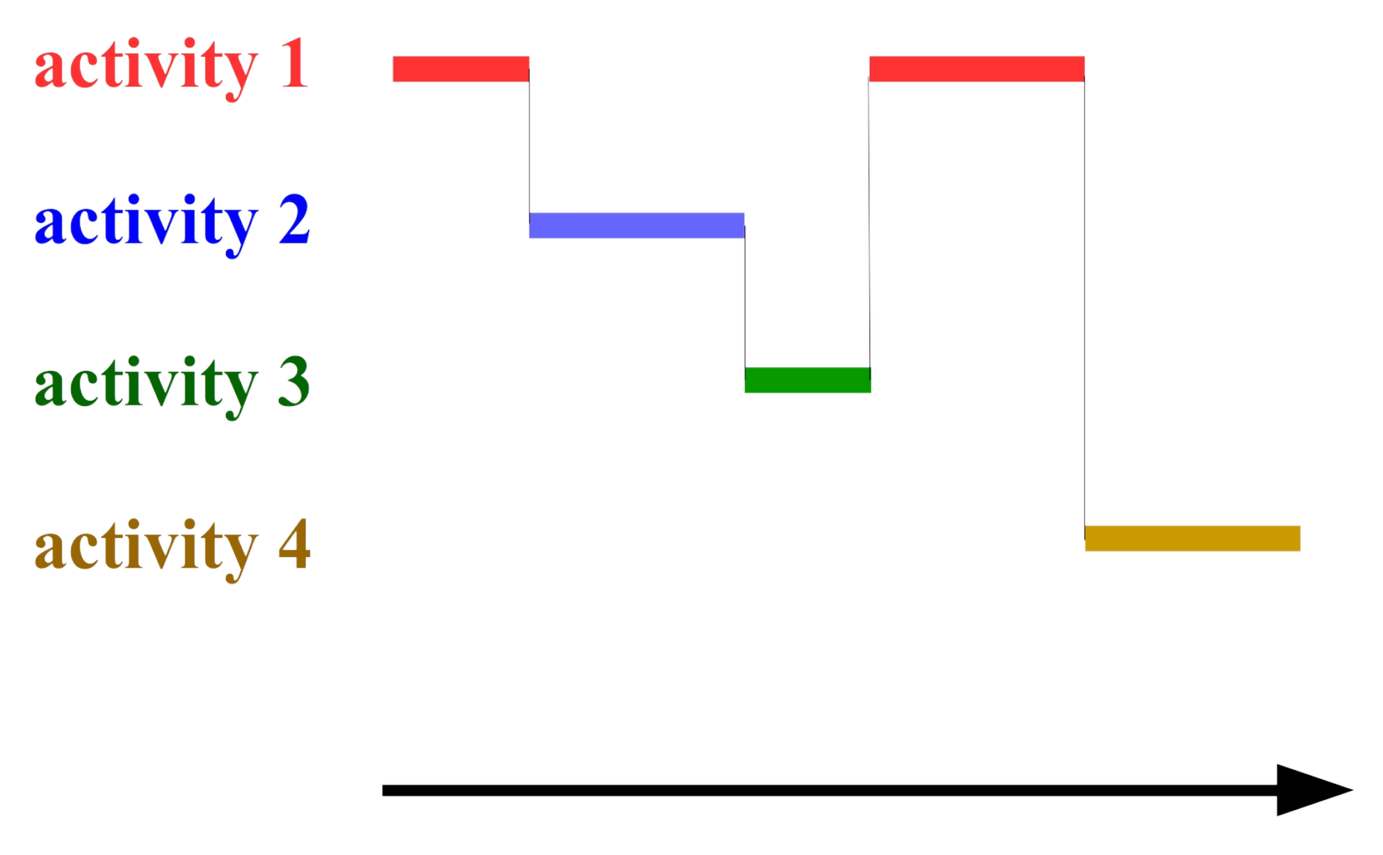
activity 1
activity 2
activity 3
activity 4
time

Test case: 10 activities over 4 AOIs

The random activities illustrated above need long observation intervals in order to be discriminated. We investigated simpler, more easily discriminable activity models consisting of uniform probabilities of visiting 2 or 3 of a total of 4 AOIs.

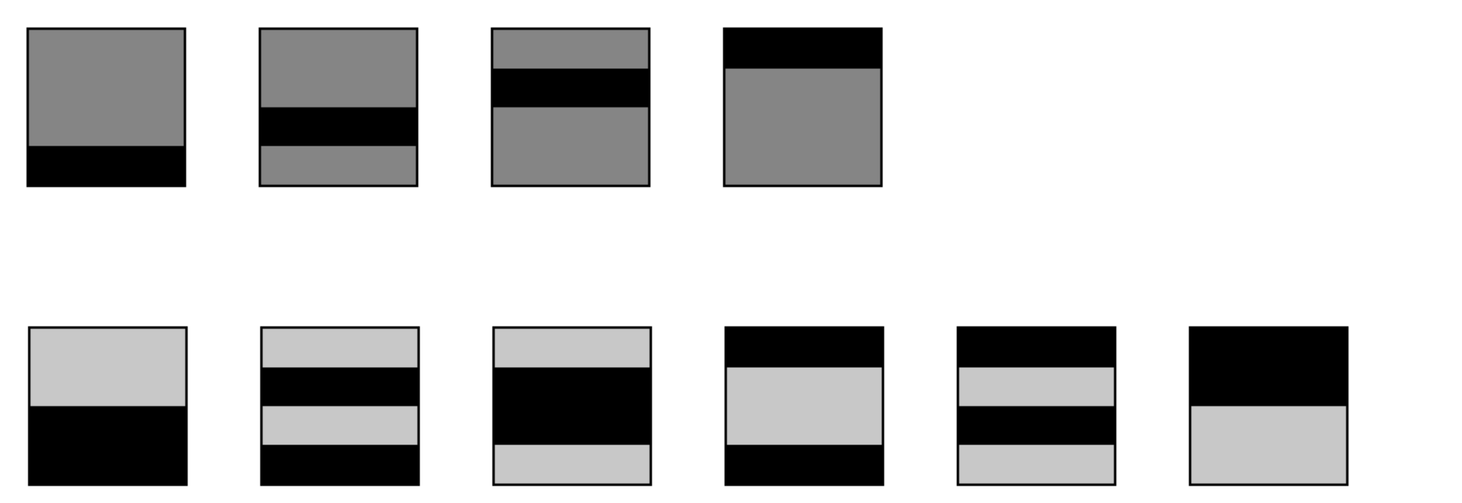
leave-one-out
leave-two-out

400 seconds of behavior were generated by running each of the activities for 40 seconds. The chi-square test was then performed for various sizes of the temporal averaging window. Shown below are the raw chi-square statistic (left), and a mask displaying a black pixel where the corresponding statistic has a p-value greater than 0.1.

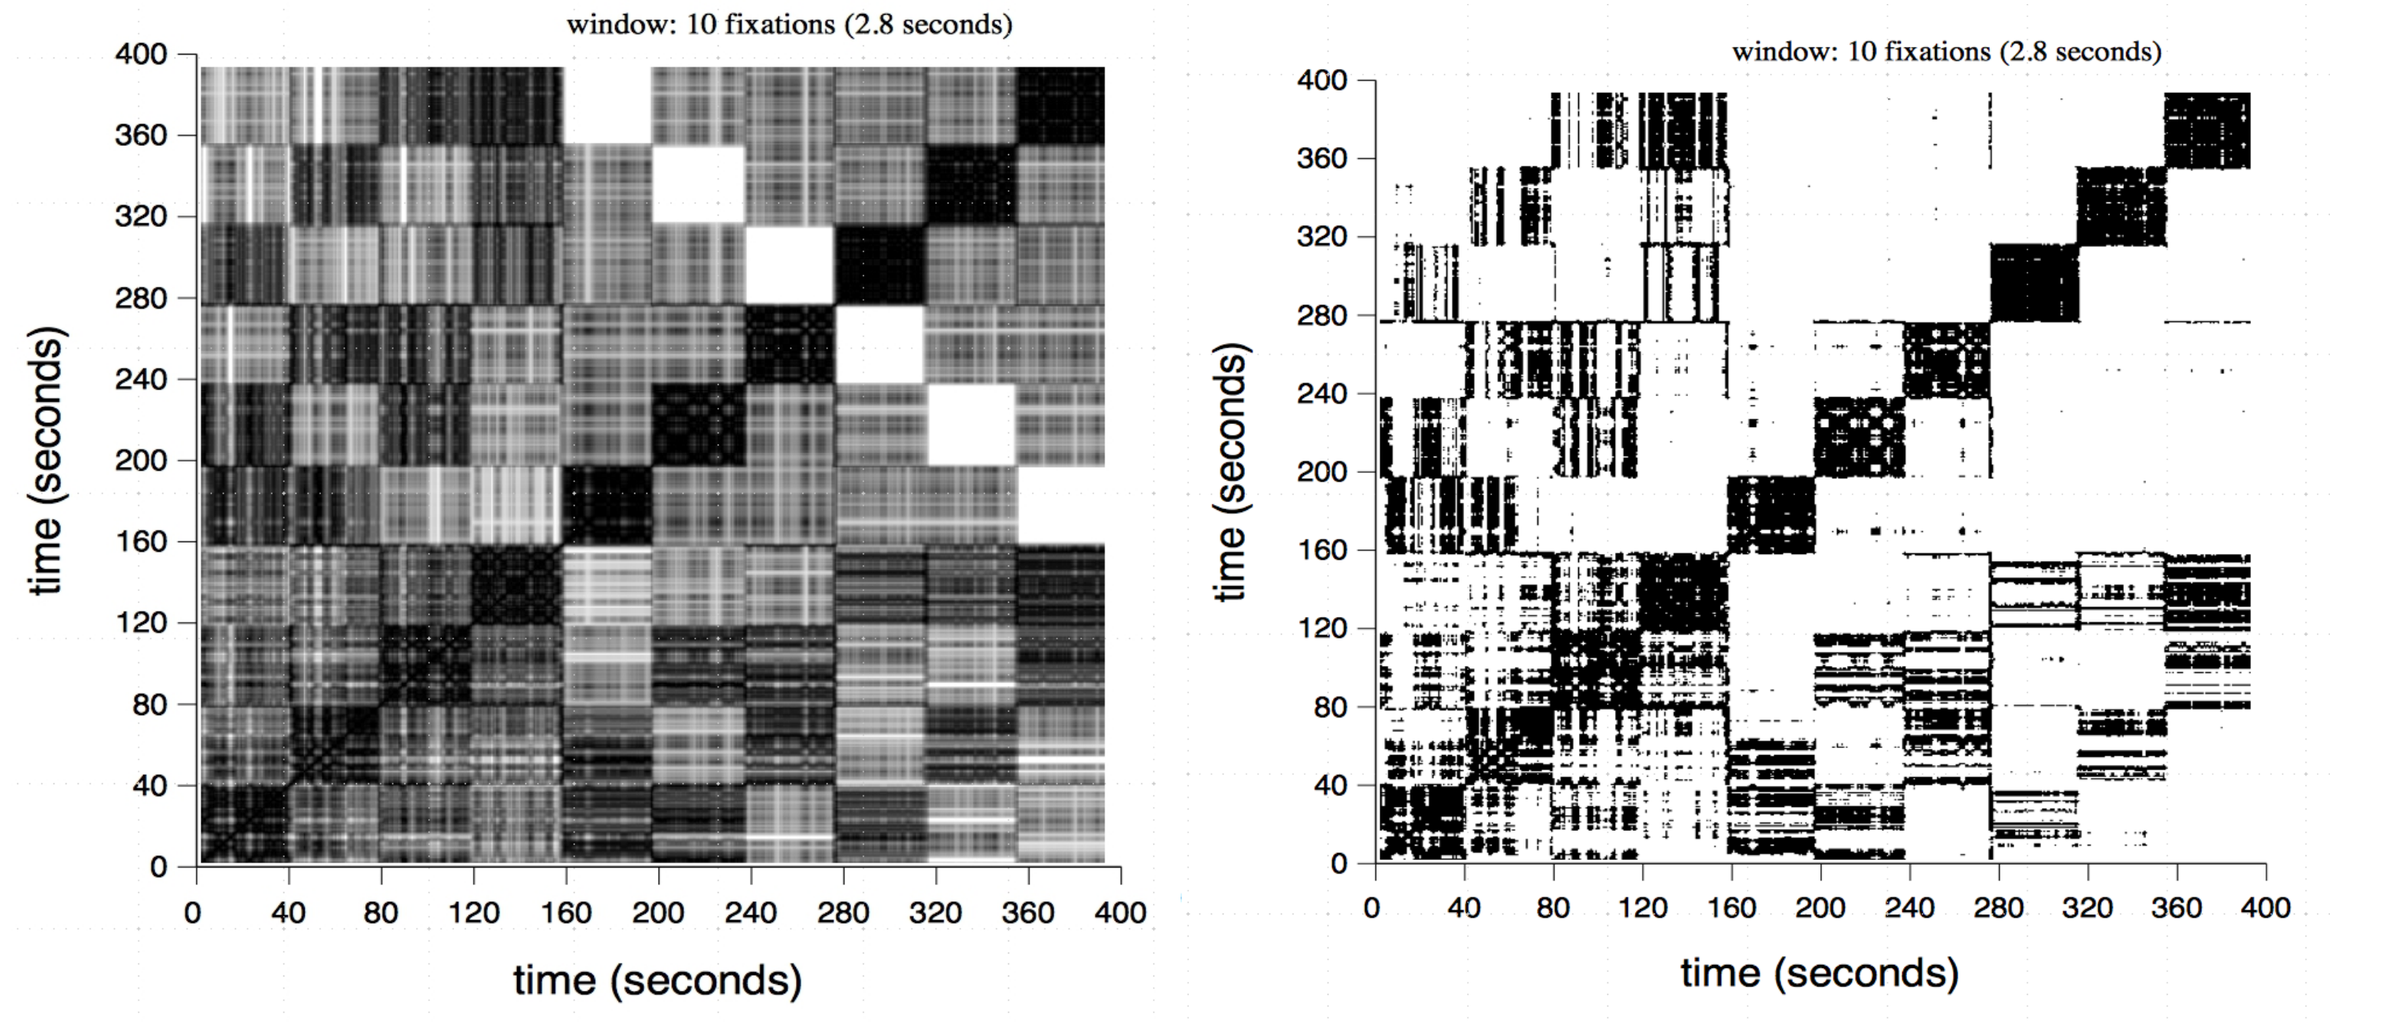
window: 10 fixations (2.8 seconds)
time (seconds)
time (seconds)
window: 10 fixations (2.8 seconds)
time (seconds)
time (seconds)

Clustering

Clustering was performed using the p-value mask images as shown on the right above. Clusters correspond to groups of black pixels on the same row or column. A method was developed to "grow" clusters from black clumps along the main diagonal.

Summary

Activities with distinct eye movement signatures can be automatically identified from scan-path data with reasonable accuracy. As activities become more similar, longer observation windows are needed in order to discriminate them. Further work is needed to determine the nature of real-life activities.

References

Anderson, N. C., Anderson, F., Kingstone, A., and Bischof, W F. (2015). A comparison of scanpath comparison methods. *Behav. Res.*, **47**:1377-1392.

Euler, L. (1735). The seven bridges of Königsberg. In Newman, J. R. (1956). *The World of Mathematics, Vol. 1.* , Simon & Schuster, New York.

Yarbus, A. L. (1967). *Eye Movements and Vision*, trans. B. Haigh, Plenum Press, New York.

Acknowledgements

JBM was supported by the Technologies for Airplane State Awareness (TASA) project of NASA's Airspace Operations and Safety Program (AOSP), and the NASA Engineering and Safety Center (NESC). Thanks to Al Ahumada for suggesting the use of the chi-square test to compare scan-path segments, and to Mike Feary, Dorrit Billman and the members of the ACE lab for useful discussions.