Oculomotor assessment of diurnal arousal variations

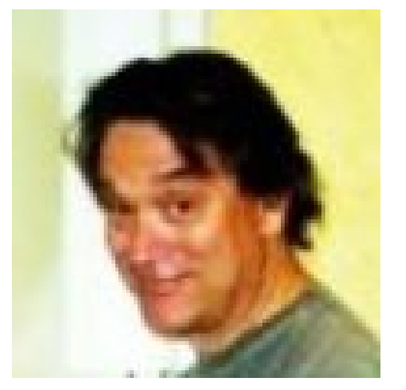

Jeffrey B. Mulligan[1], Carolina Diaz-Piedra[2] and Leandro L. Di Stasi[2,3]

[1]NASA Ames Research Center, [2]University of Granada, [3]University of Padova

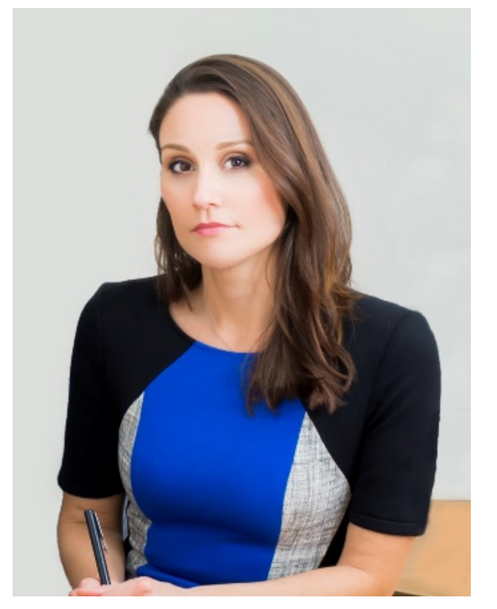

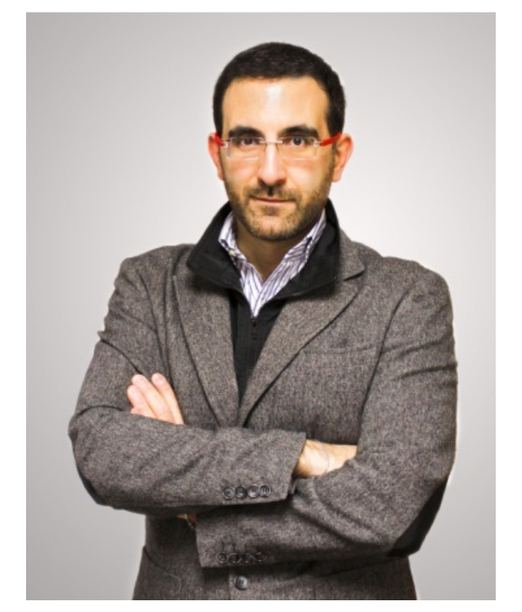

Abstract

Saccadic and pupil responses are reliable indices of arousal decrement (e.g. fatigue), that might be exploited to improve work schedule guidelines. In this study, we tested the sensitivity of a short 30-s oculomotor test to detect diurnal arousal variations. ~~Twelve participants (5 females, 7 males, 37.7±11.9 years)~~ Eleven participants volunteered to be assessed every hour (66±20 min) for three consecutive working days, during their regular office-hours. We used a fully automated testing system, the FIT 2000 Fitness Impairment Tester (Pulse Medical Instruments Inc., Rockville, MD, USA), to measure and record saccadic peak velocity, pupil diameter, and latency and amplitude of the pupillary light reflex. In addition, we collected subjective levels of arousal using the Stanford Sleepiness Scale, and body core temperature. We analyzed the data using a linear mixed model approach for longitudinal data. Both saccadic velocity and subjective alertness decreased over the course of a day, while body core temperature increased (all *p*-values < .05). The data also weakly suggested an increase of the pupil diameter (*p*=.07). The findings support the use of oculomotor indices in the assessment of arousal and fatigue in applied settings.

One of the original twelve participants was dropped from the analysis due to medication taken during the study.

Operator State Assessment

Human operators possess many advantages over fully-automated control systems, but they are not infallible. When humans are performing safety-critical tasks, it is desirable to employ monitoring technology to insure that operators remain alert and engaged.

Eye monitoring systems are likely to be an important component of future operator monitoring systems. Gaze direction can provide information about spatial allocation of attention, and slow eyelid closures are associated with imminent sleep. In this paper, we investigate *Peak Saccadic Velocity* (PSV) as a possible indicator of arousal decrements that occur long before the onset of sleep. Previous work (Diaz-Piedra et al., 2016) has demonstrated changes in PSV during helicopter missions lasting several hours.

Unfortunately,due to the brief nature of saccades, accurate measurement of PSV requires specialized equipment: the system used in this study (PMI FIT-2000, see lower left panel) employs an analog optical servo system with high bandwidth; measurement of PSV with a video-based eye-tracker generally requires a high frame rate (500 fps or above). Mulligan (2008) has demonstrated a method employing multiple intra-frame flashes enabling measurement of PSV using a standard camera with 60 fps.

Weak correlations

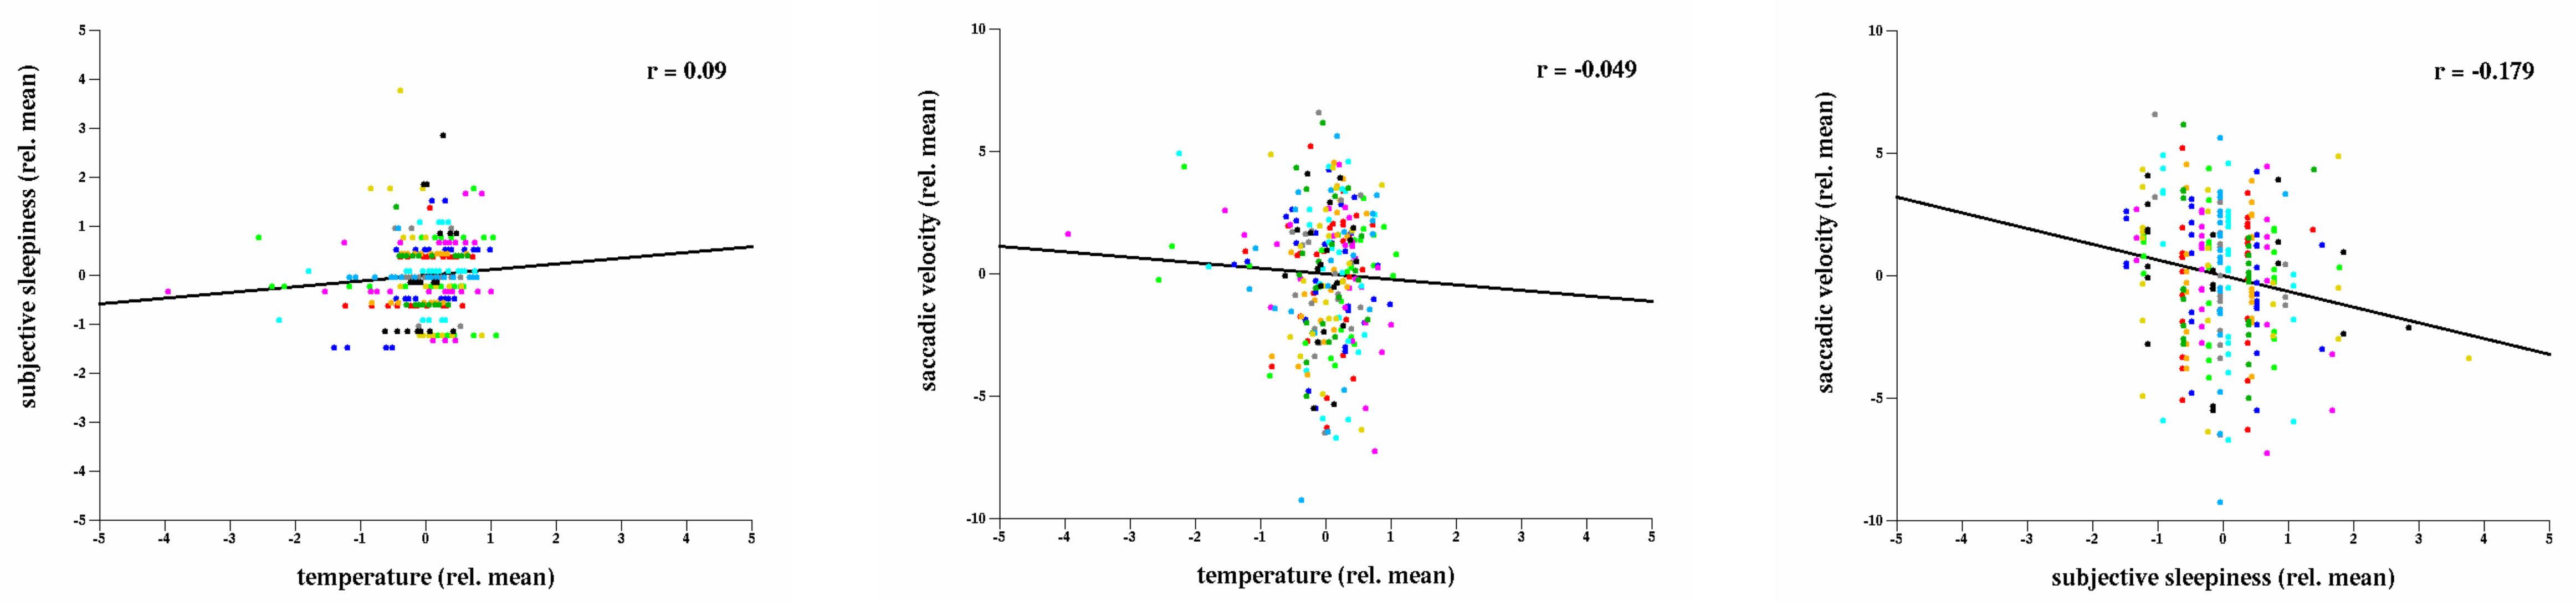

r = 0.09
subjective sleepiness (rel. mean)
temperature (rel. mean)
r = -0.049
saccadic velocity (rel. mean)
temperature (rel. mean)
r = -0.179
saccadic velocity (rel. mean)
subjective sleepiness (rel. mean)

Three variables (saccadic velocity, temperature, and subjective sleepiness) showed significant trends throughout the work-day (see data below). But these variables are only weakly correlated with each other. This may be due in part to measurement noise (see panels to the right). We have also, somewhat inappropriately, treated the sleepiness ratings as metric quantities.

Repeatability

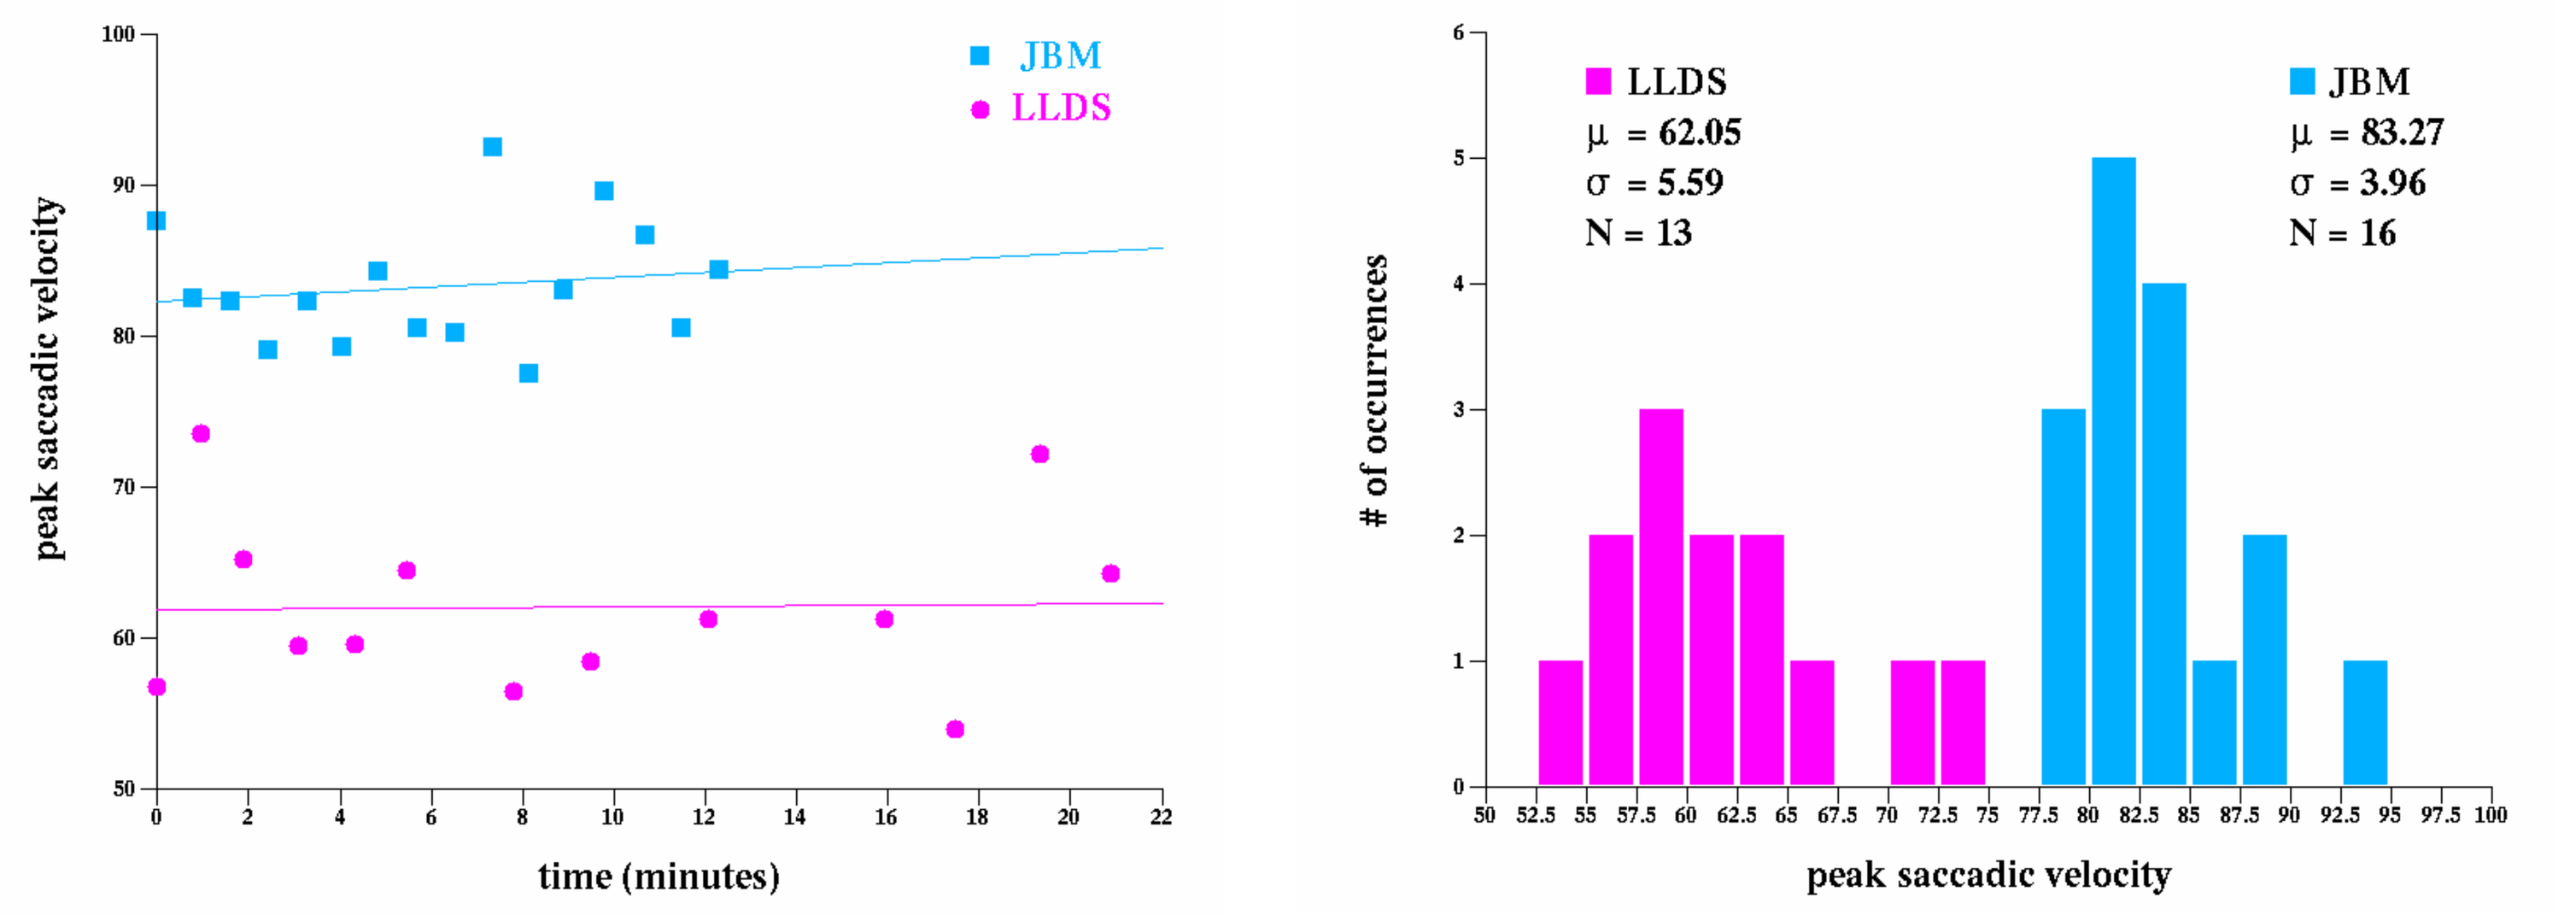

JBM
LLDS
peak saccadic velocity
time (minutes)
LLDS
µ = 62.05
σ = 5.59
N = 13
JBM
µ = 83.27
σ = 3.96
N = 16
of occurrences
peak saccadic velocity

Two of the authors made a number of measurements in rapid succession to assess precision. Measurement variability is large – the standard deviation is approximately 10 times the average hourly decrease in saccadic velocity. Ongoing work seeks to determine how much of this is due to behavioral variability, versus measurement noise in the instrument. It is not clear whether the instrument's velocity measurements are normalized with respect to the amplitude of the executed saccade.

The PMI FIT 2000

The PMI FIT-2000 fitness-for-duty tester (Pulse Medical Instruments, Rockville MD) performs a brief (30 second) oculomotor test battery designed to detect impairment produced by fatigue or intoxication.

The subject presses a button to initiate the measurements; saccadic peak velocity (SPV) is measured first, by having the subject track the apparent motion between a pair of alternately lit LEDs. After a brief pause, the a series of flashes are delivered to measure the pupillary light reflex (PLR). Three pupil parameters are measured, the resting pupil diameter (PD), and the amplitude and latency of the PLR.

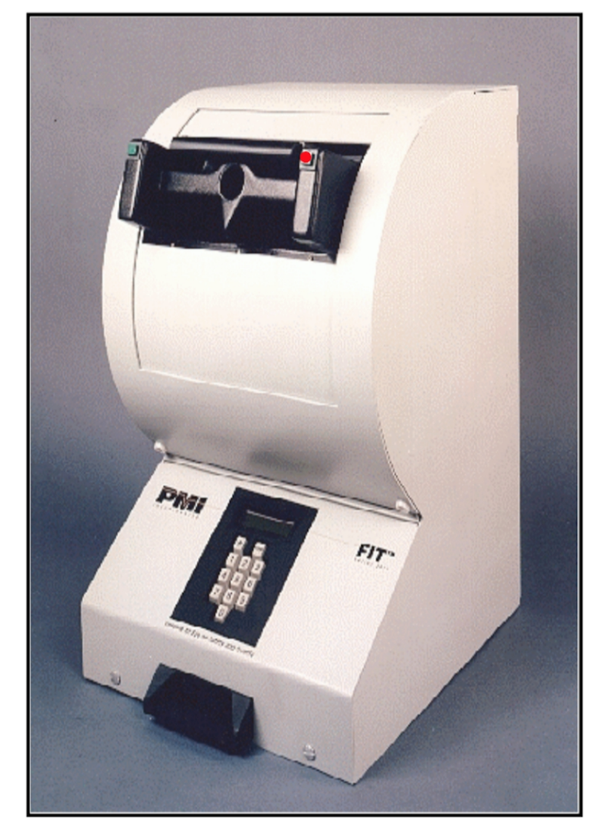

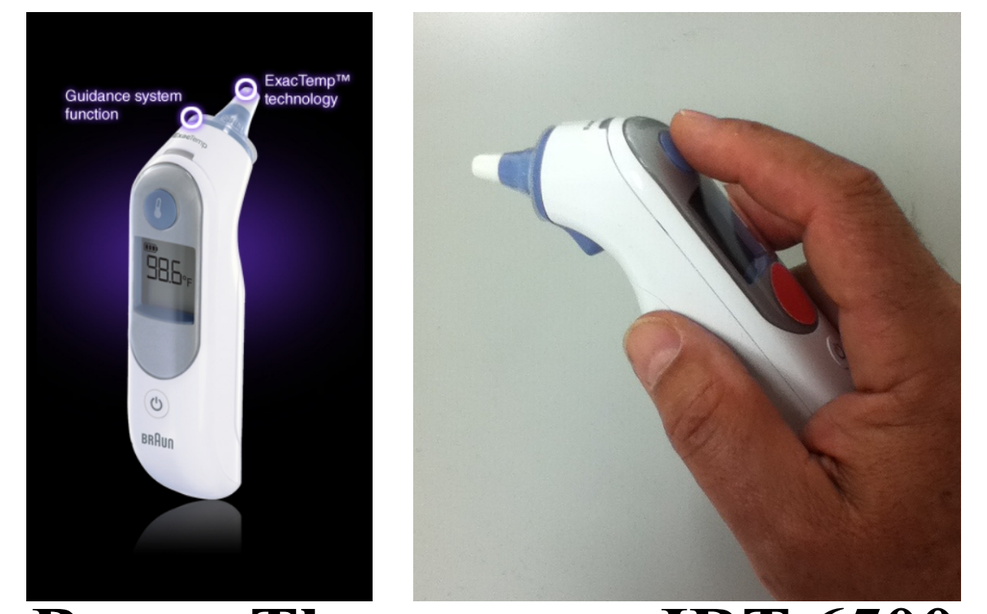

Braun Thermoscan IRT 6500

Subjects used this ear thermometer to record body temperature before each FIT measurement.

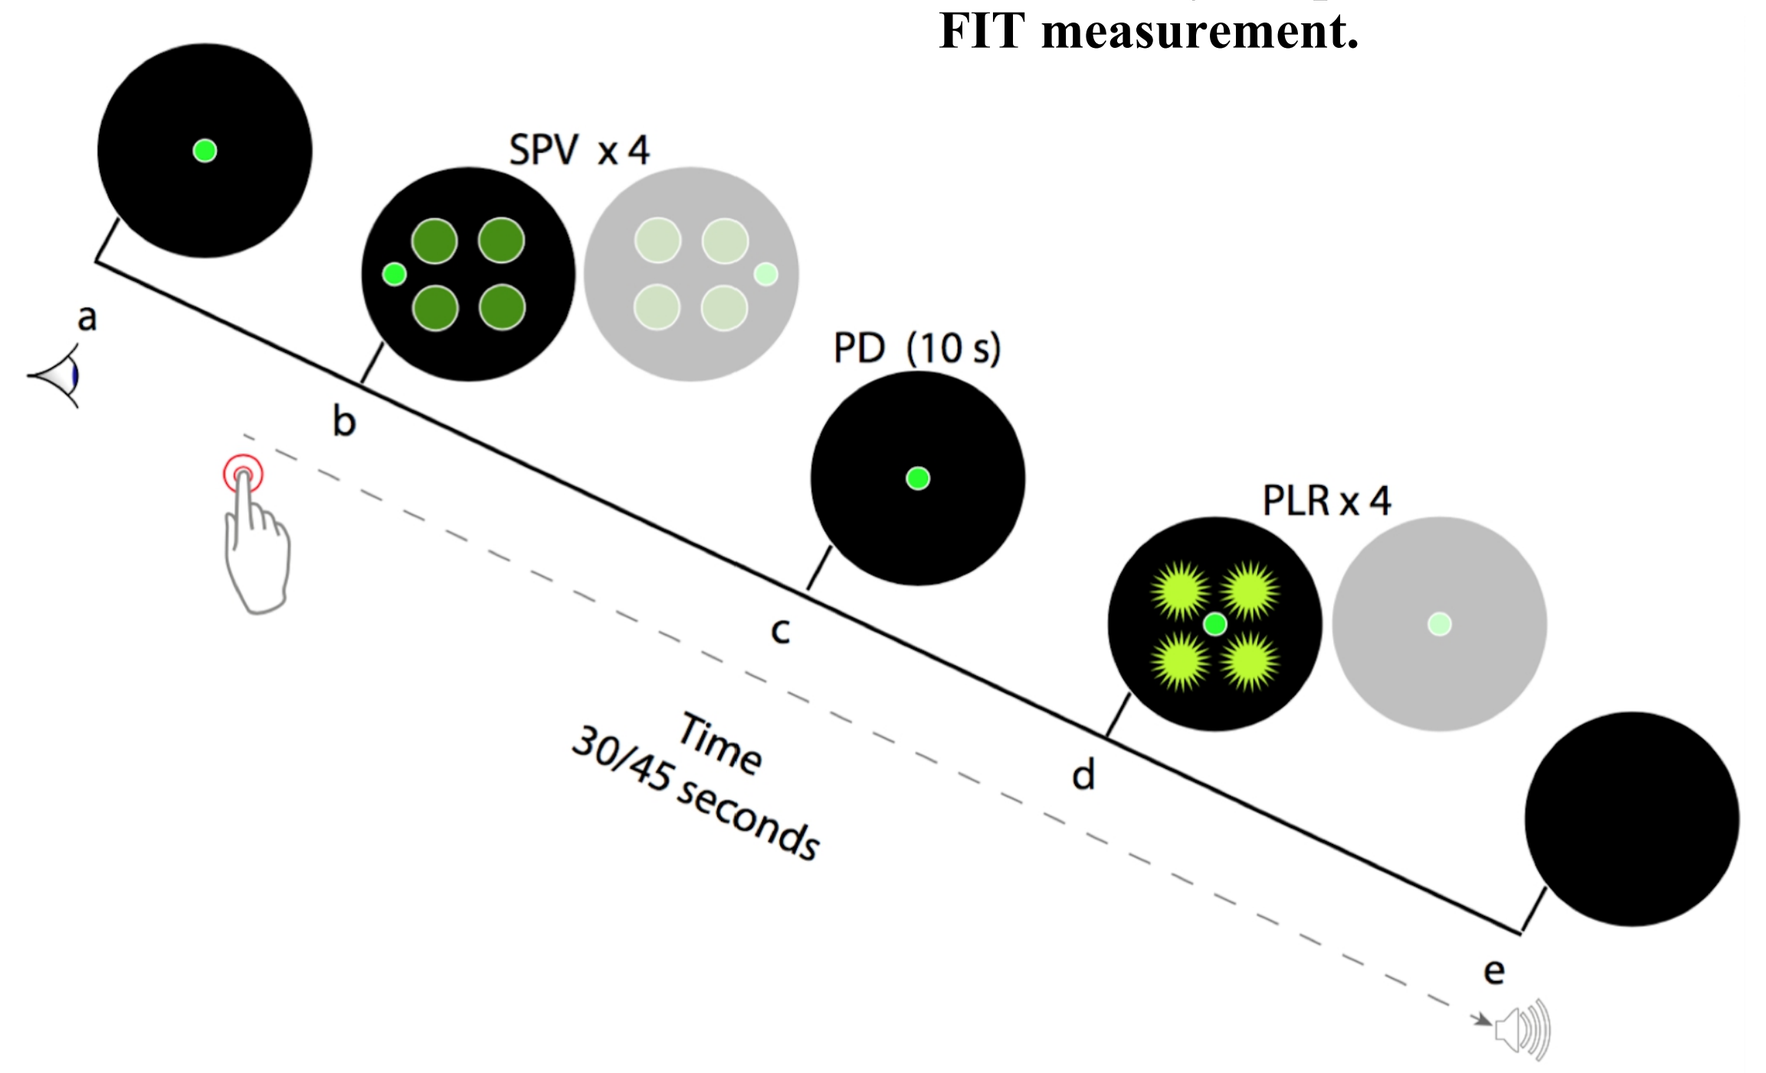

SPV x 4
PD (10 s)
PLR x 4
a
b
c
d
e
Time
30/45 seconds

JBM LLDS CDP S4 S5 S6 S7 S8 S9 S10 S11

Peak Saccadic Velocity

Pupil Diameter

Pupil Constriction

Pupil Latency

Temperature

Subjective Sleepiness

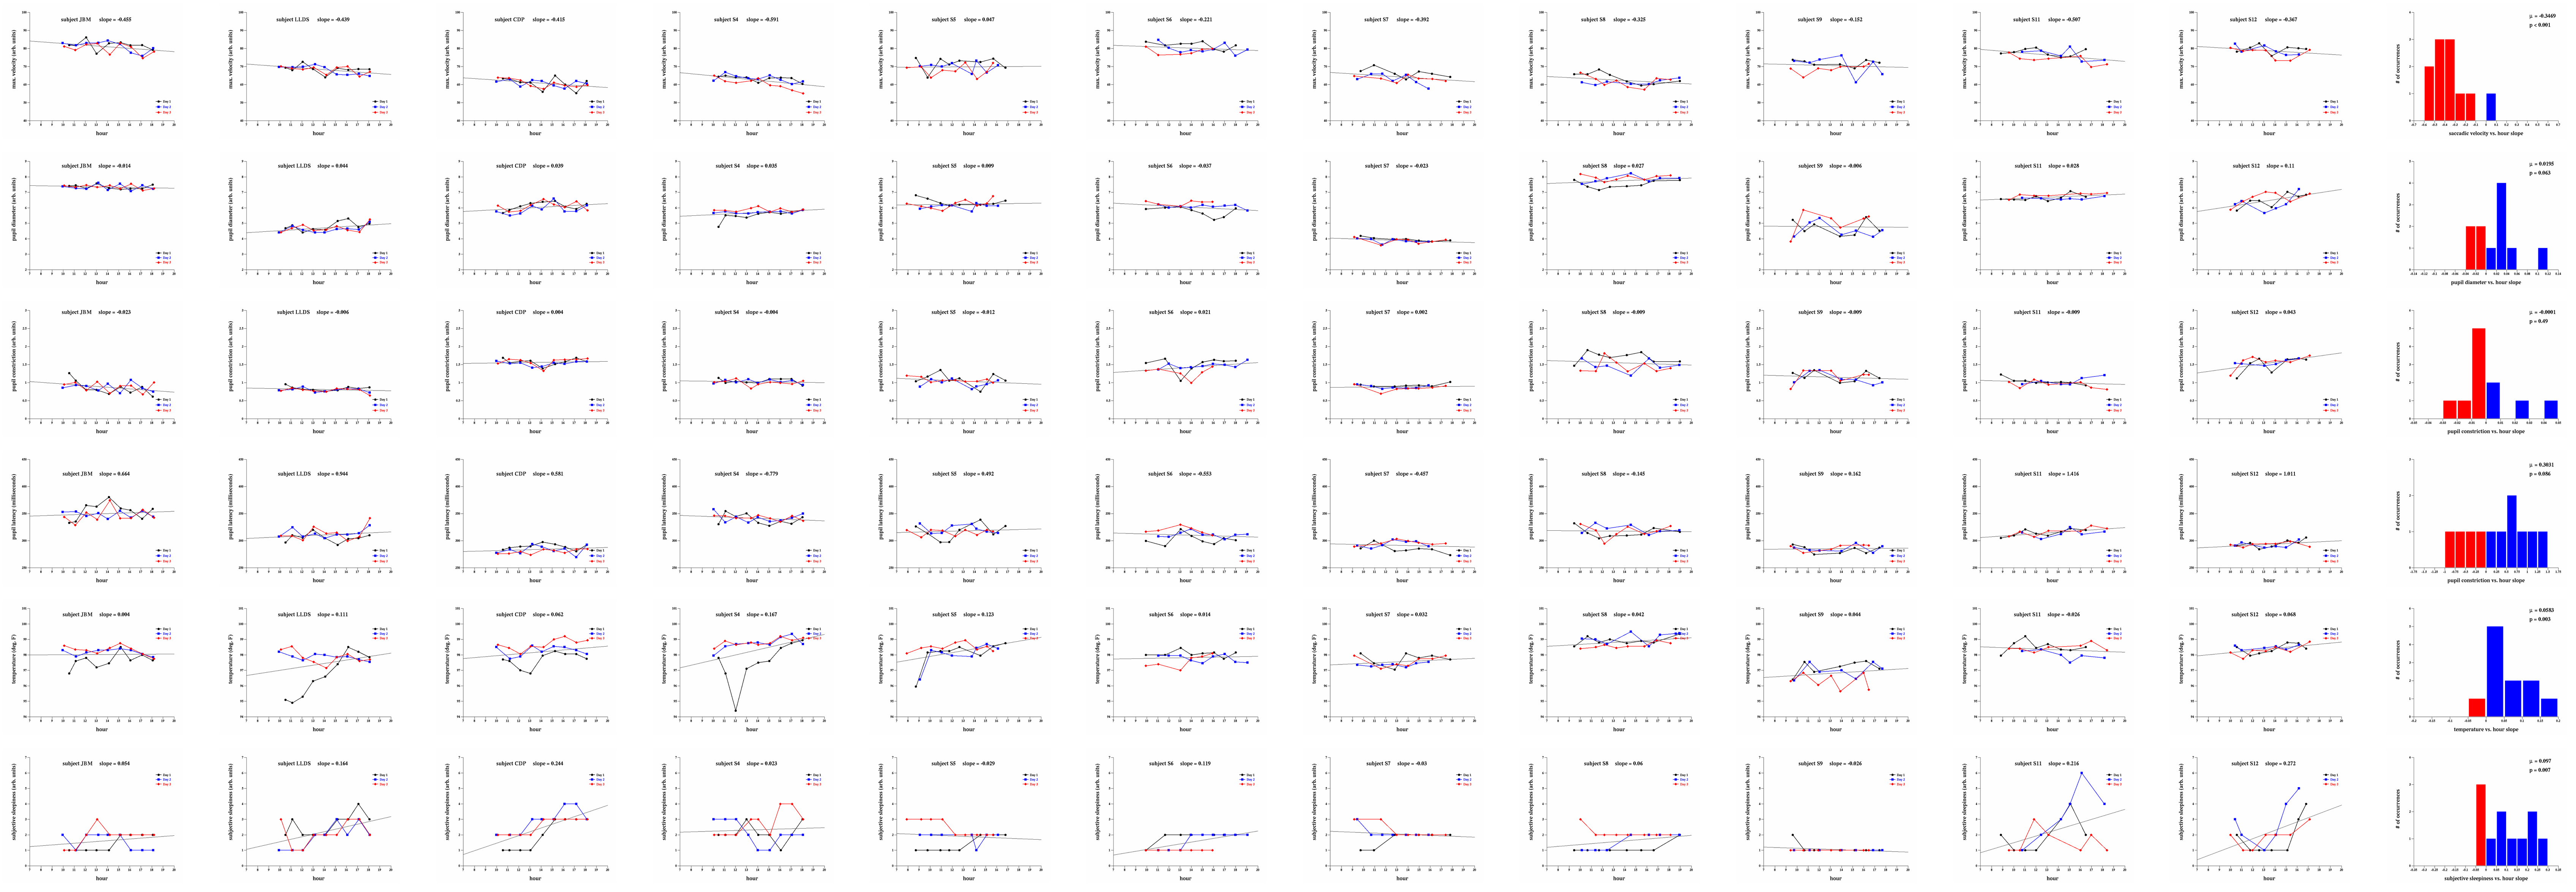

**

**

Summary

Of the four oculomotor parameters measured by the PMI FIT-2000, only peak saccadic velocity appears to vary systematically during the work day. Although the measurements are noisy, saccadic velocity could be a valuable component of an operator monitoring system, because so many saccades are made in the course of normal behavior.

References

Diaz-Piedra, C., Rieiro, H., Suárez, J., Rios-Tejada, F., Catena, A., and Di Stasi, L. L. (2016). Fatigue in the military: towards a fatigue detection test based on the saccadic velocity. *Physiological Measurement*, **37**(9), N62-75.

Mulligan, J. B. (2008). Measurement of eye velocity using active illumination. *Proc. 2008 ACM Symposium on Eye Tracking Research and Applications (ETRA)*, 35-38.

Acknowledgements

JBM was supported by the Technologies for Airplane State Awareness (TASA) project of NASA's Airspace Operations and Safety Program (AOSP), and the NASA Engineering and Safety Center (NESC). CDP was supported by a UGR Postdoctoral Fellowship (2013 University of Granada Research Plan). LLDS was supported by the Talentia Postdoc Program (COFUND—Grant Agreement no. 267226) and is currently supported by the the Ramón y Cajal fellowship program (RYC-2015-17483).

The authors also express their gratitude to all of the volunteer subjects that participated in this study.