Smooth pursuit of flicker-defined motion

Jeffrey B. Mulligan
NASA Ames Research Center

Scott B. Stevenson
University of Houston College of Optometry

BACKGROUND

Delayed Visual Feedback

JOURNAL OF NEUROPHYSIOLOGY
Vol. 67, No. 3, March 1992. Printed in U.S.A.

Effect of Changing Feedback Delay on Spontaneous Oscillations in Smooth Pursuit Eye Movements of Monkeys

D. GOLDREICH, R. J. KRAUZLIS, AND S. G. LISBERGER
Department of Physiology, W. M. Keck Foundation Center for Integrative Neuroscience, and Neuroscience Graduate Program, University of California, San Francisco, California 94143

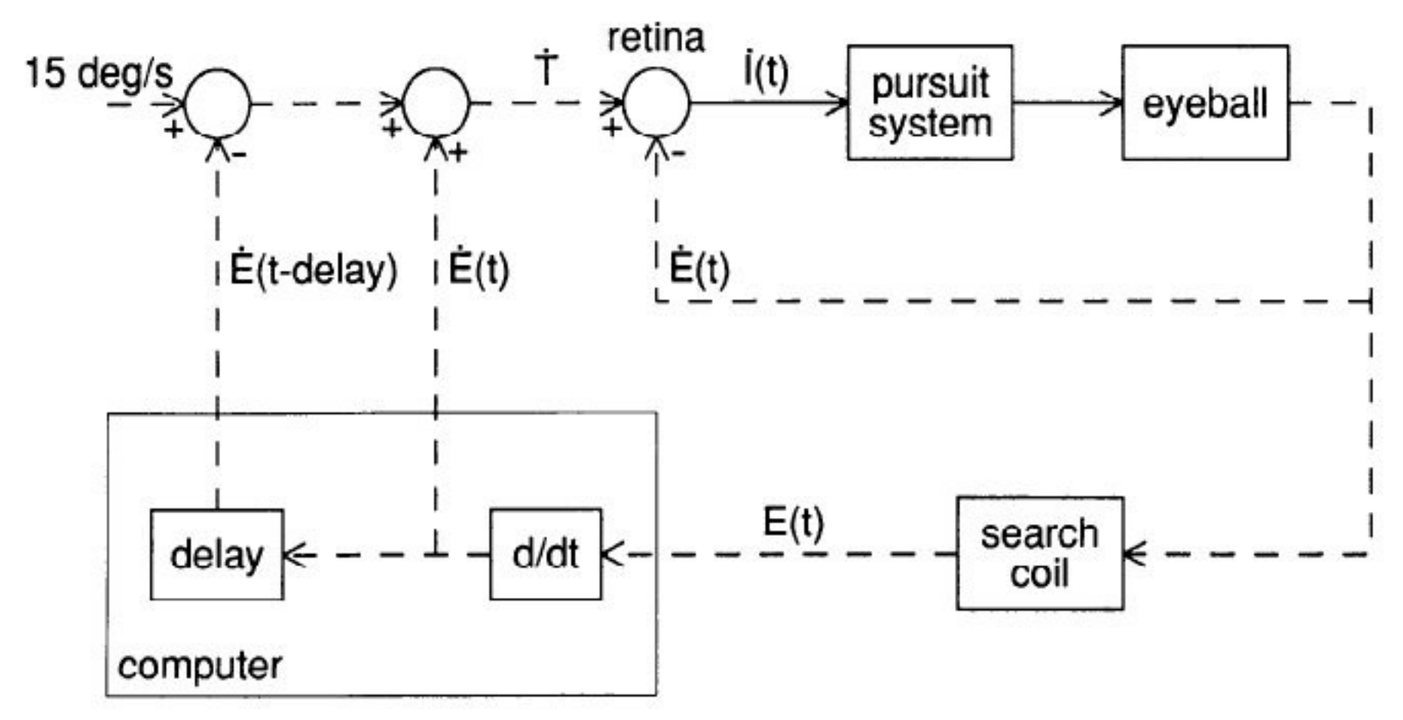

15 deg/s
retina
Ṫ
İ(t)
pursuit system
eyeball
Ė(t-delay)
Ė(t)
Ė(t)
delay
d/dt
E(t)
search coil
computer

Figure from Goldreich, Krauzlis & Lisberger (1992).

Theory

$e(t)$	eye position (EP)
$p(t)$	target position
$\dot{e}(t)$	eye velocity (EV)
$\dot{p}(t)$	target velocity
$\ddot{e}(t)$	eye acceleration (EA)
$\ddot{p}(t)$	target acceleration

$p(t) = k$	stationary target
$p(t) = e(t)$	ideal stabilization
$p(t) = e(t-\varepsilon)$	lab stabilization
$p(t) = e(t) + d(t)$	open-loop
$p(t) = e(t) - e(t-\delta_2)$	transient stabilization

$p(t) - e(t)$	retinal position error (RPE)
$\dot{p}(t) - \dot{e}(t)$	retinal velocity error (RVE)
$\ddot{p}(t) - \ddot{e}(t)$	retinal acceleration error (RAE)

$\ddot{e}(t) = k_1\left[p(t-\delta_1) - e(t-\delta_1)\right]$ RPE drives EA

$\ddot{e}(t) = k_2\left[\dot{p}(t-\delta_1) - \dot{e}(t-\delta_1)\right]$ RVE drives EA

$\ddot{e}(t) = k_3\left[\ddot{p}(t-\delta_1) - \ddot{e}(t-\delta_1)\right]$ RAE drives EA

Predictions

$p(t) = e(t) - e(t-\delta_2)$ transient stabilization

$\ddot{e}(t) = k_1\left[p(t-\delta_1) - e(t-\delta_1)\right]$ RPE drives EA

$\ddot{e}(t) = k_1\left[e(t-\delta_1) - e(t-\delta_1-\delta_2) - e(t-\delta_1)\right]$

$\ddot{e}(t) = -k_1 e(t-\delta_1-\delta_2)$

$\ddot{e}(t) = -k_1\, e(t-\delta)$

$e(t) = e^{i\omega t}$ trial solution

satisfied if $k_1 = \omega^2$ and $\lambda = \delta$,

where $\lambda \equiv \frac{2\pi}{\omega}$.

$\lambda = \delta$	RPE drives EA
$\lambda = 4\delta$	RVE drives EA
$\lambda = 2\delta$	RAE drives EA

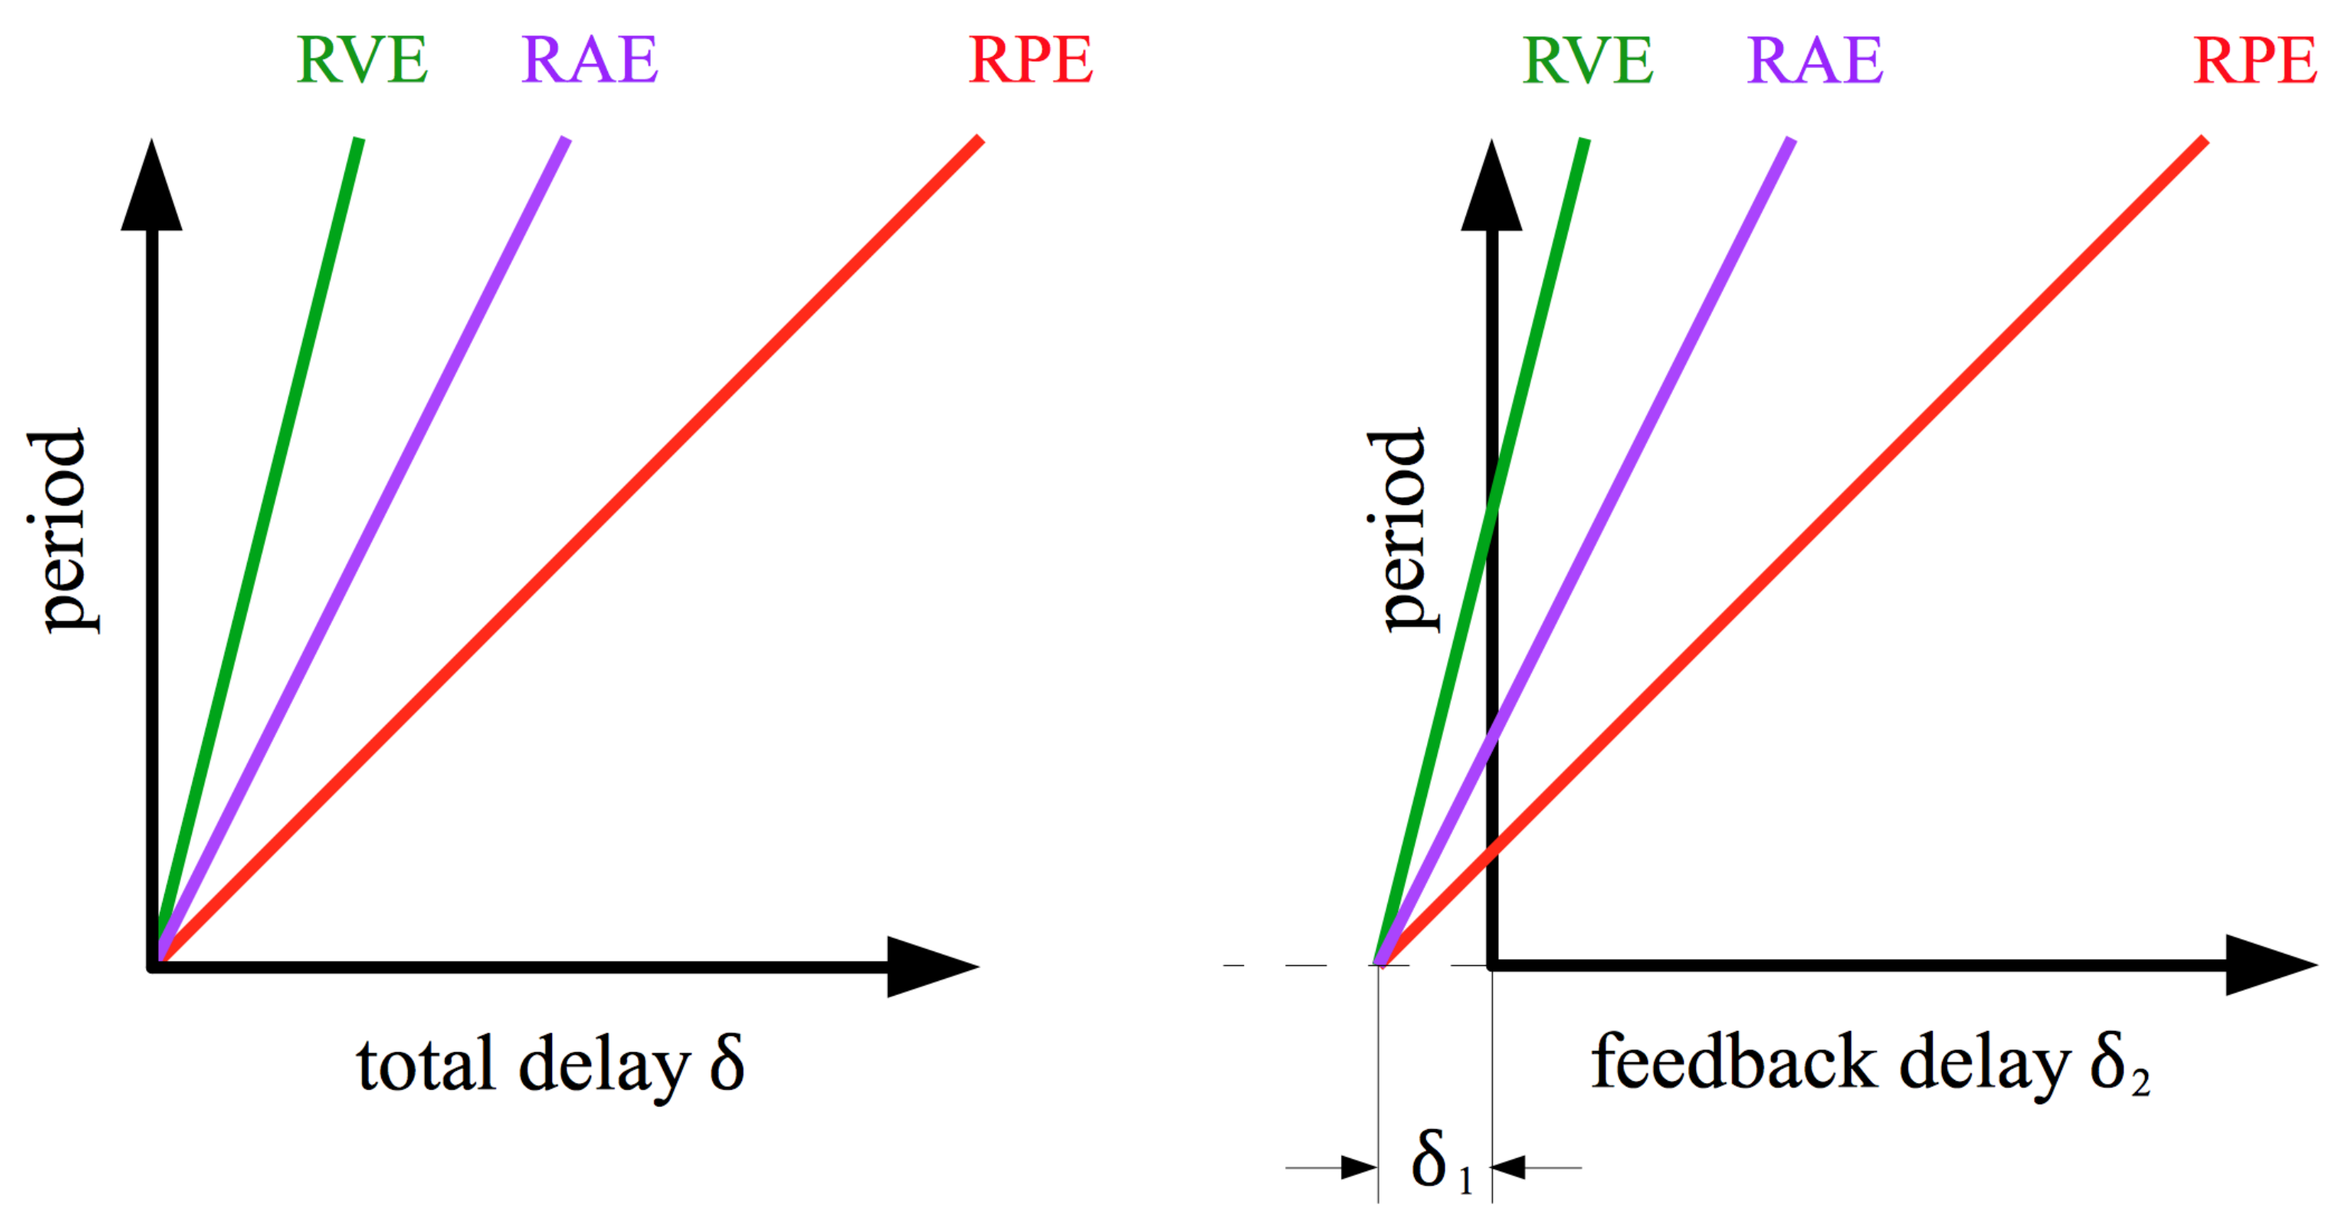

RVE
RAE
RPE
period
total delay δ
RVE
RAE
RPE
period
feedback delay δ_2
δ_1

Results

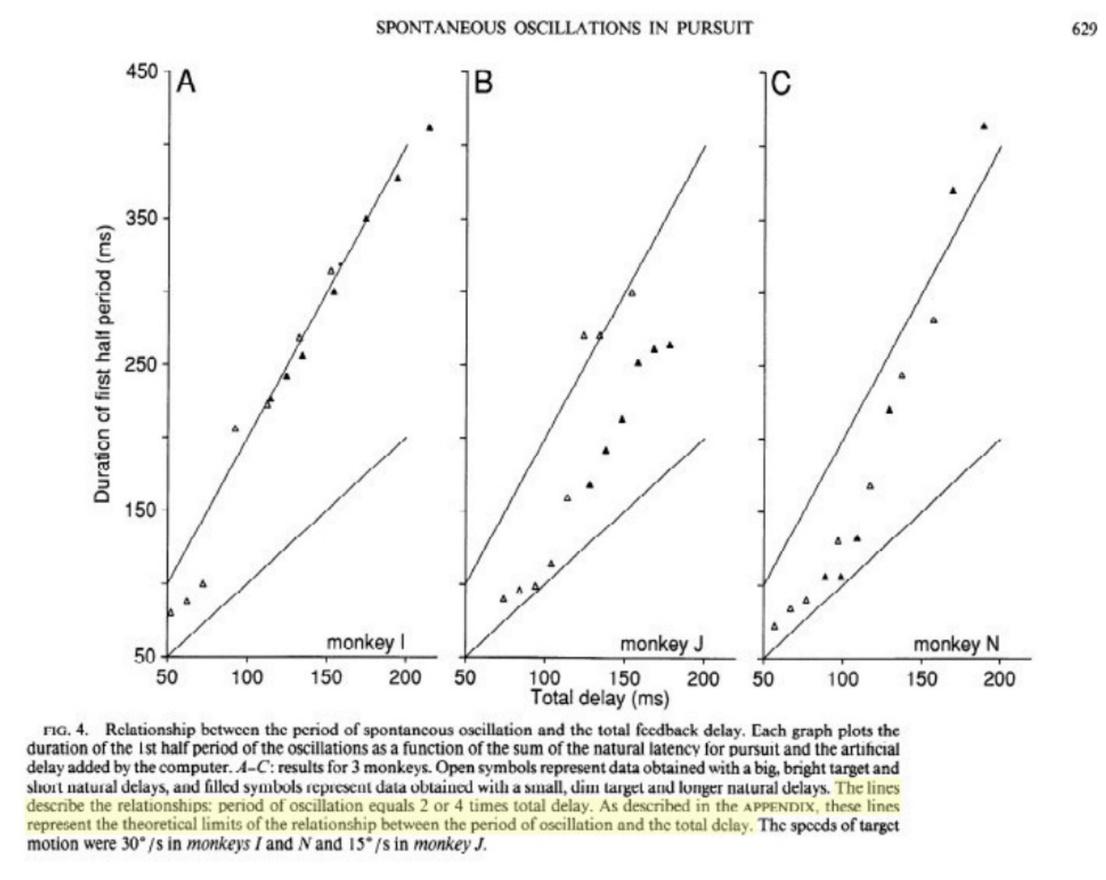

SPONTANEOUS OSCILLATIONS IN PURSUIT
A
B
C
Duration of first half period (ms)
monkey I
monkey J
monkey N
Total delay (ms)

Figure from Goldreich, Krauzlis & Lisberger (1992).

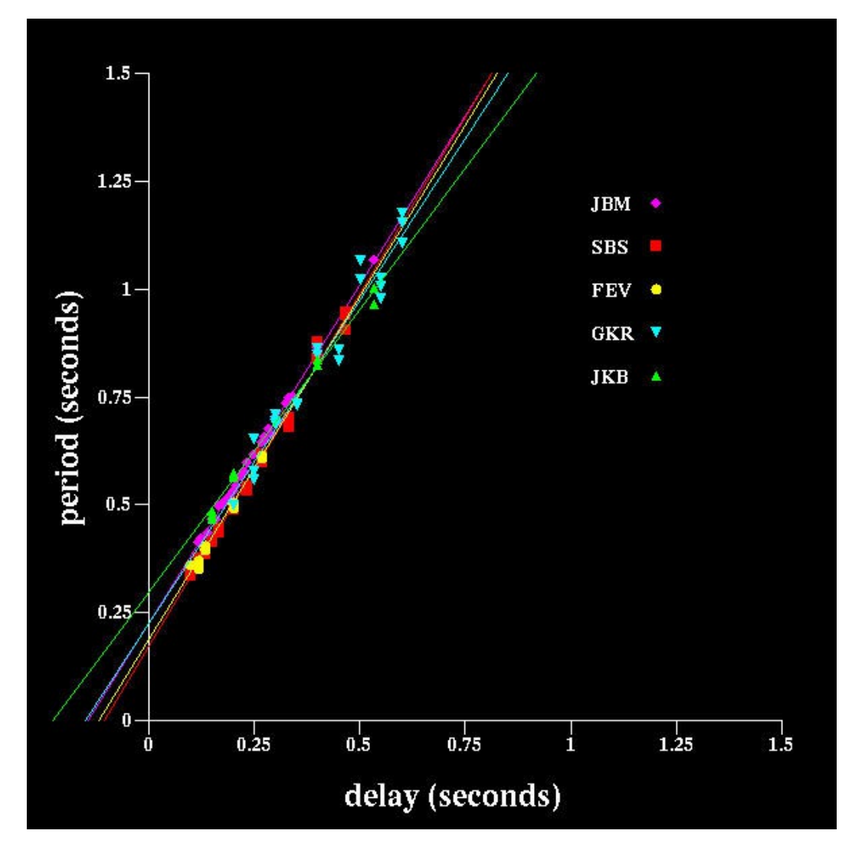

period (seconds)
delay (seconds)
JBM
SBS
FEV
GKR
JKB

Motivation:

Previous work suggests pursuit responses combine signals from voluntary and reflexive pathways with different properties, which respond to different stimuli. The reflexive system appears to respond only to first-order (luminance-defined) motion; here we investigate whether the voluntary system has a different weighting of position and velocity inputs.

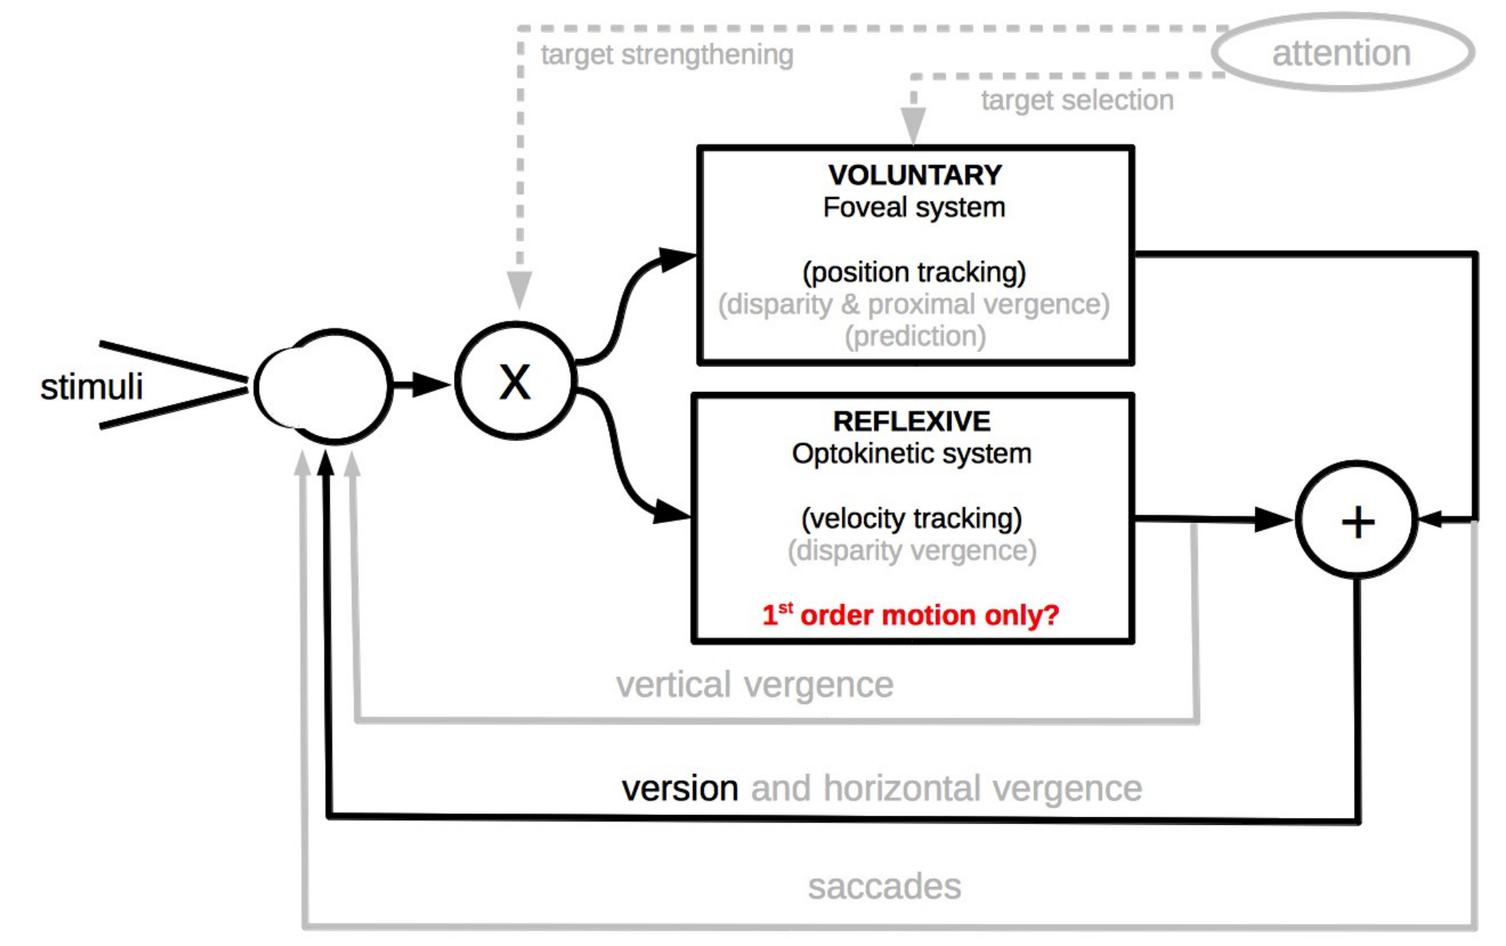

target strengthening
attention
target selection
VOLUNTARY
Foveal system
(position tracking)
(disparity & proximal vergence)
(prediction)
stimuli
X
REFLEXIVE
Optokinetic system
(velocity tracking)
(disparity vergence)
1st order motion only?
+
vertical vergence
version and horizontal vergence
saccades

Figure from Mulligan, Stevenson & Cormack (2013), showing proposed architectural model of pursuit control system, with portions relevent to this poster highlighted.

BIG QUESTION:

Can we demonstrate a position-driven pursuit response for certain classes of stimuli?

Approach:

Delayed Visual Feedback
(see Background panel at left)

First-order stimuli

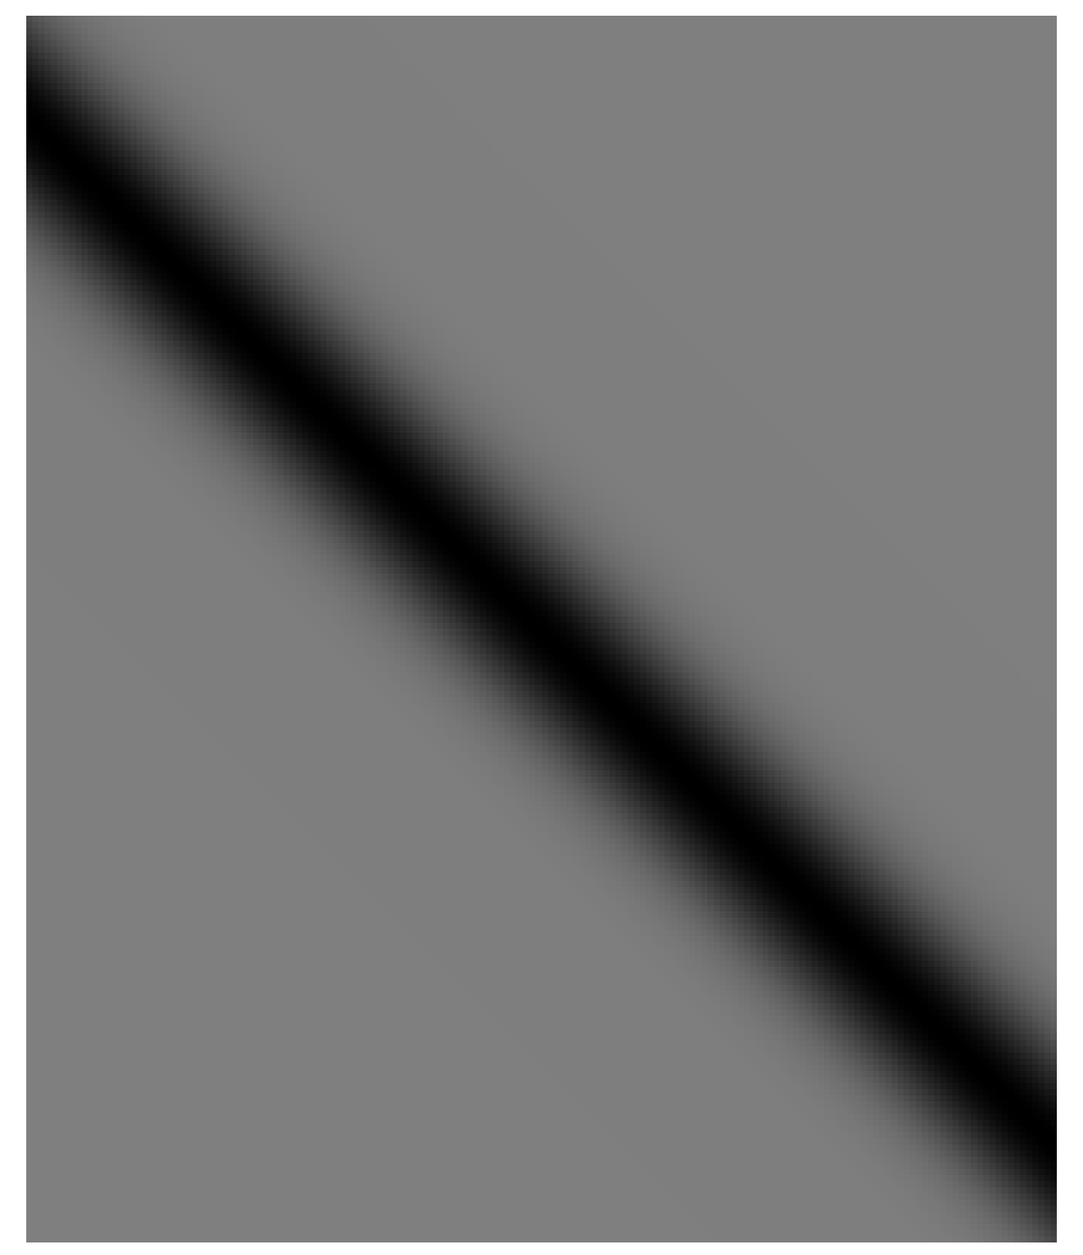

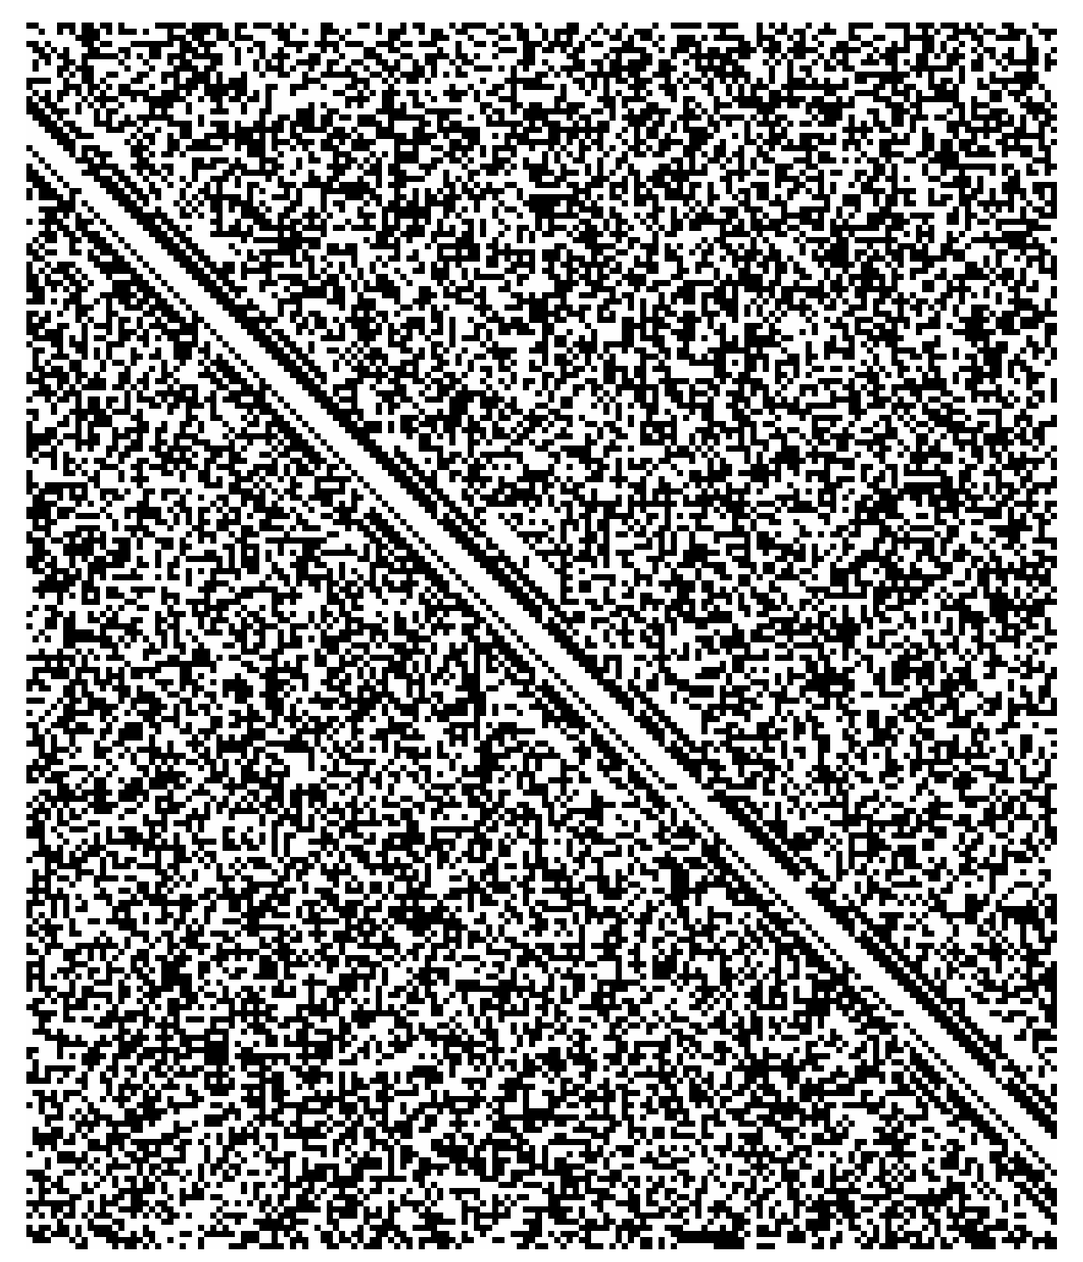

"black" "rigid"

Third-order stimuli

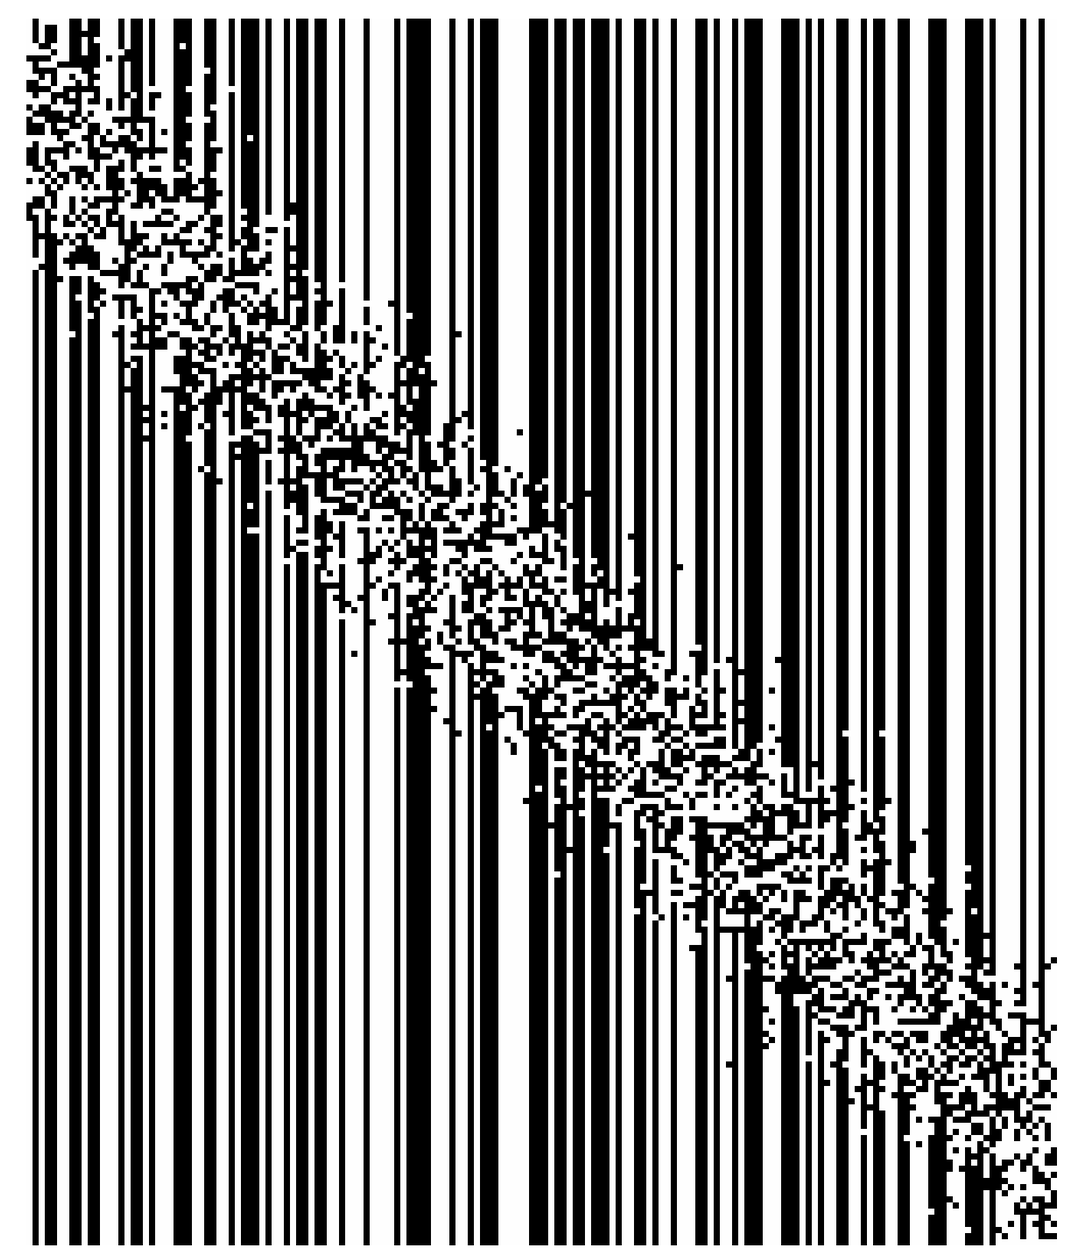

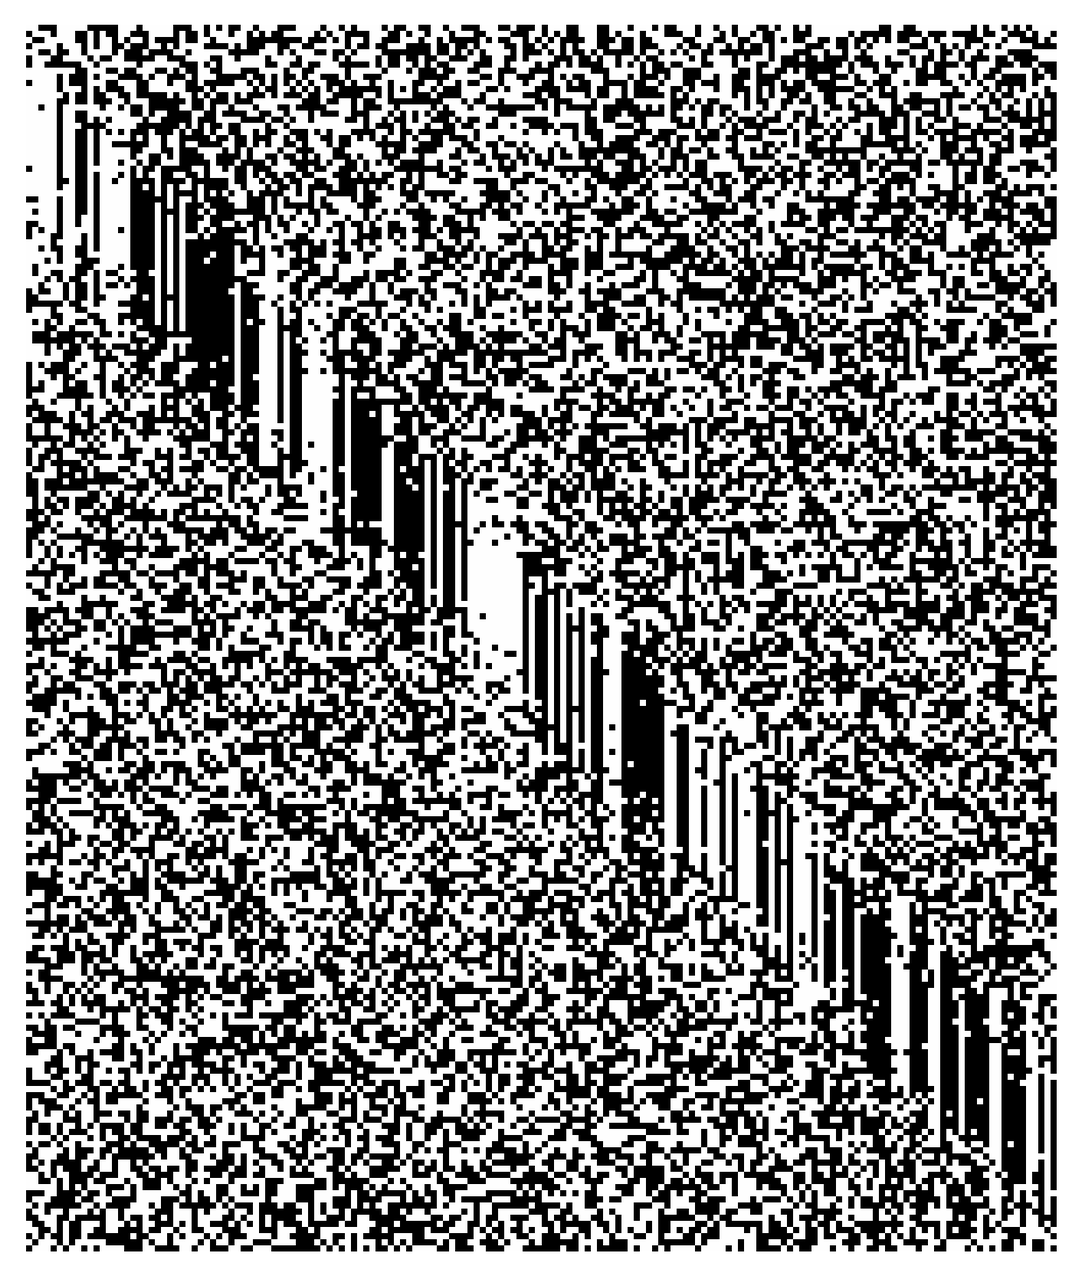

"twinkle" "static"

Space-time plots showing time (top-to-bottom), and one spatial dimension for the four classes of stimuli studied.

Trials

Vigorous oscillation seen with conventional target

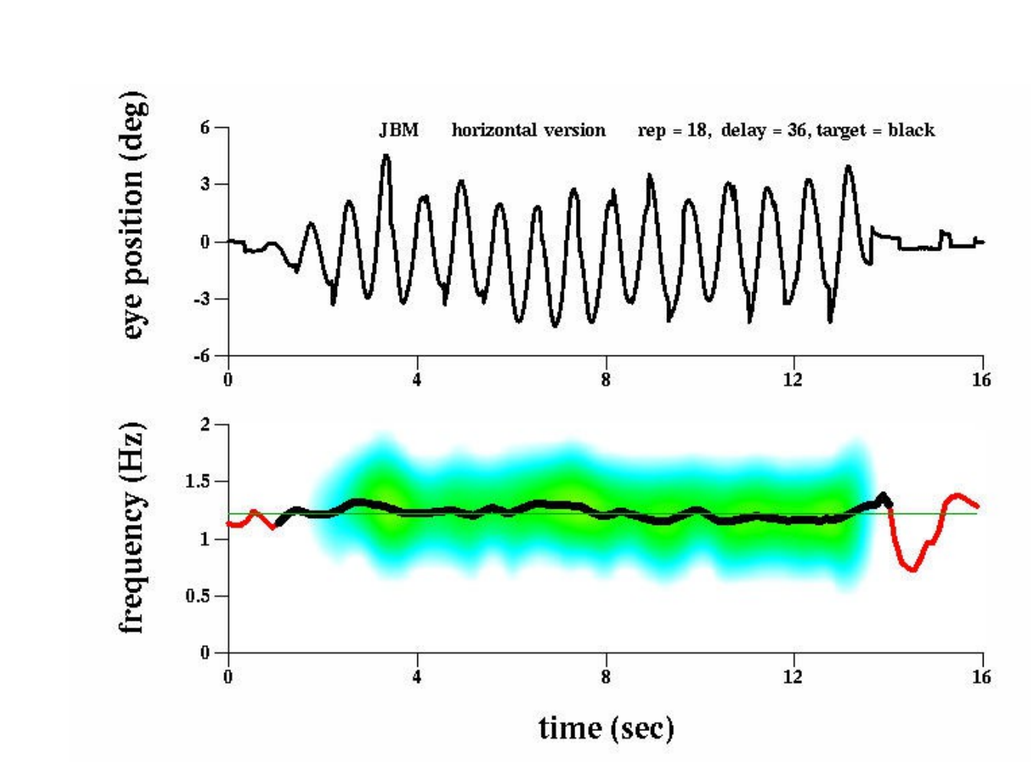

JBM horizontal version rep = 18, delay = 36, target = black
eye position (deg)
frequency (Hz)
time (sec)

Twinkle target oscillates only after learning

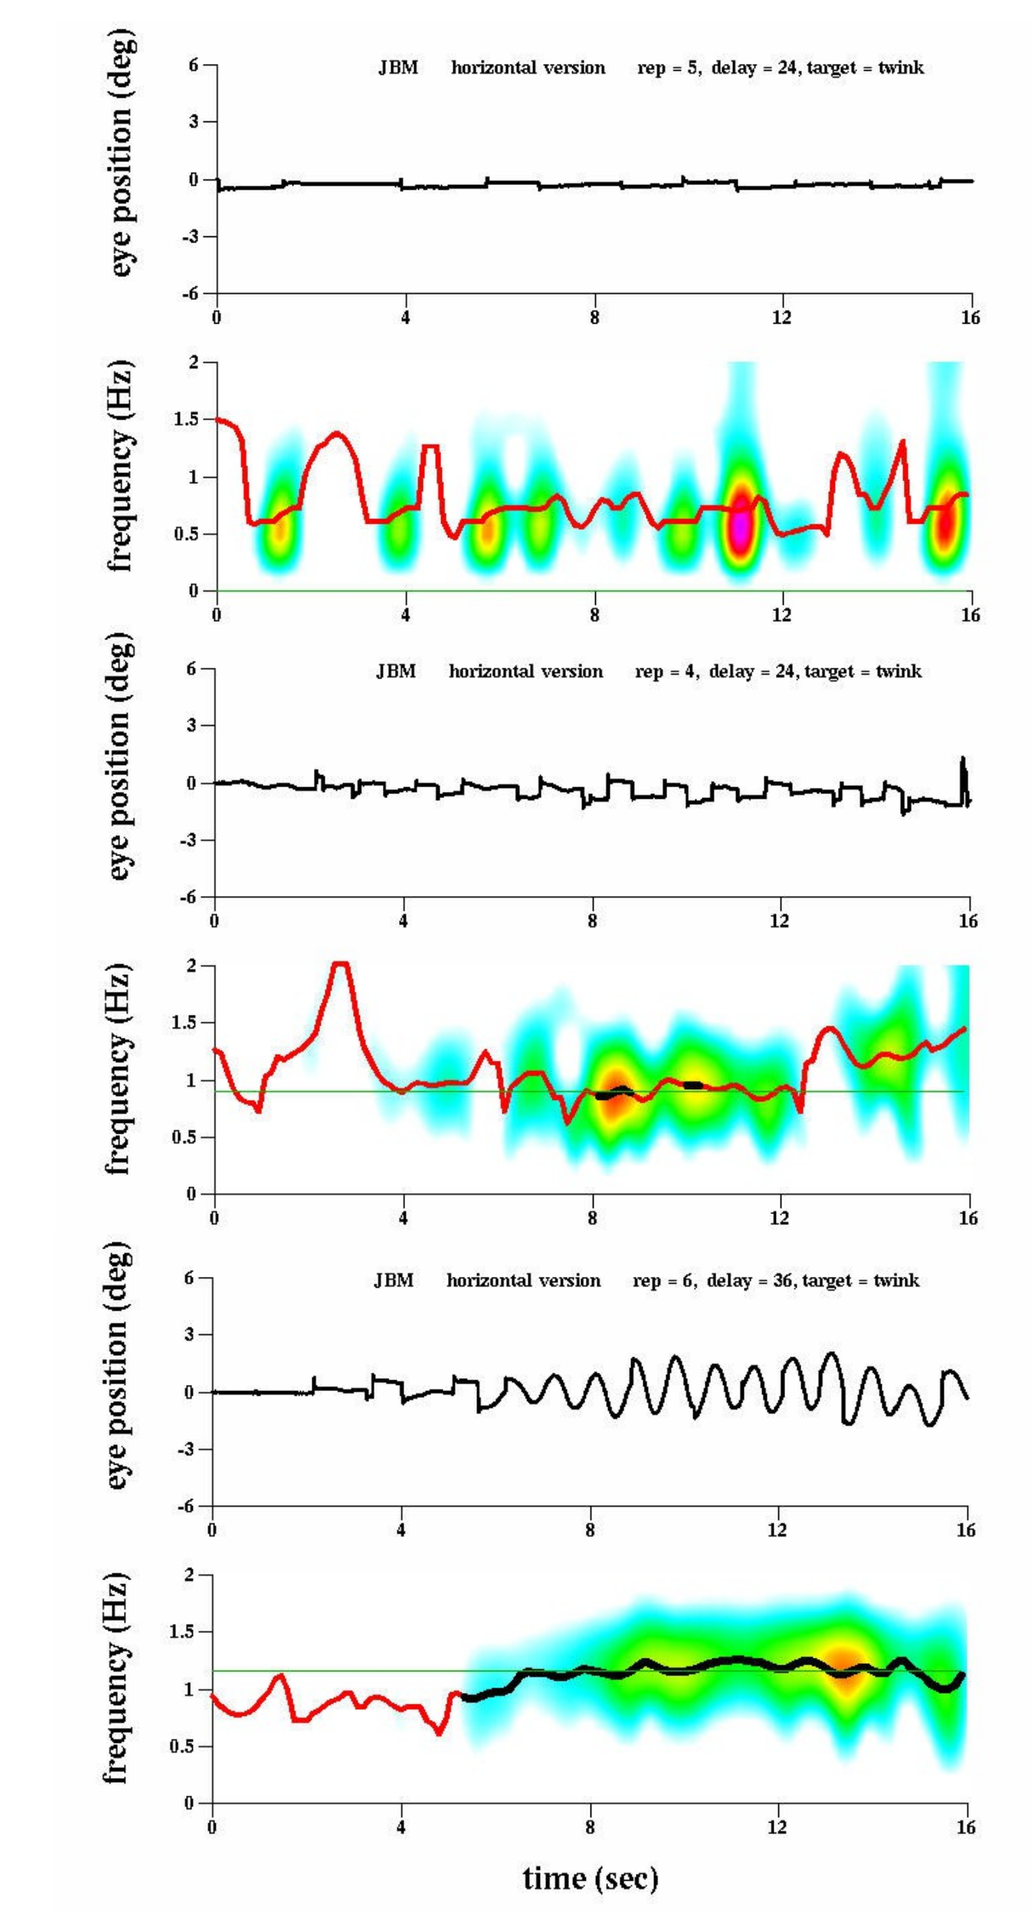

JBM horizontal version rep = 5, delay = 24, target = twink
JBM horizontal version rep = 4, delay = 24, target = twink
JBM horizontal version rep = 6, delay = 36, target = twink
eye position (deg)
frequency (Hz)
time (sec)

"Static" target show mixed response

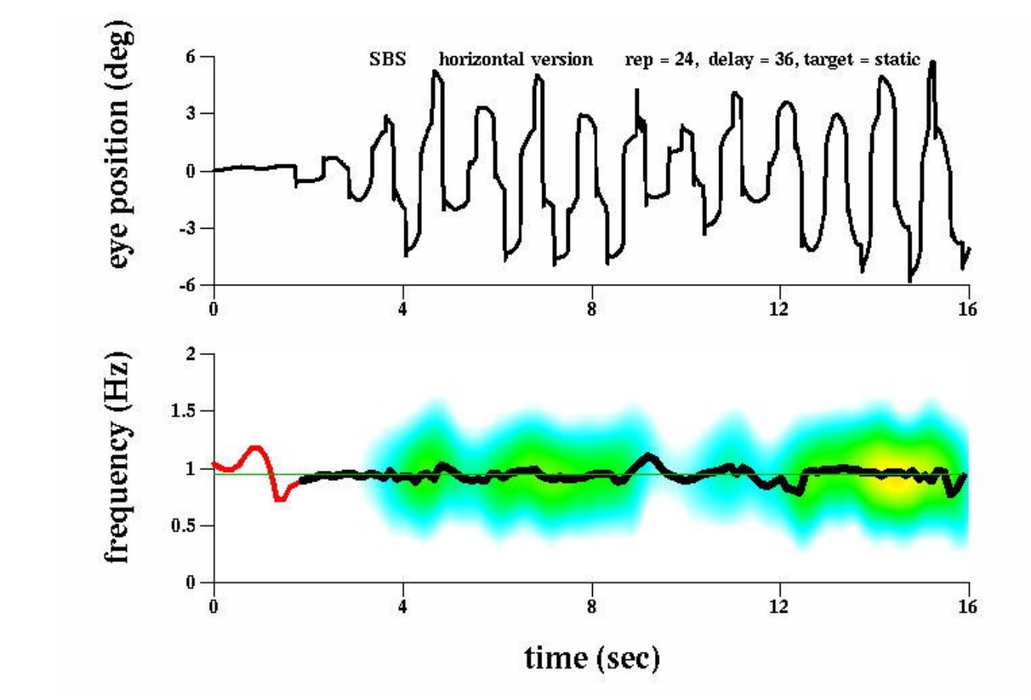

SBS horizontal version rep = 24, delay = 36, target = static
eye position (deg)
frequency (Hz)
time (sec)

Results

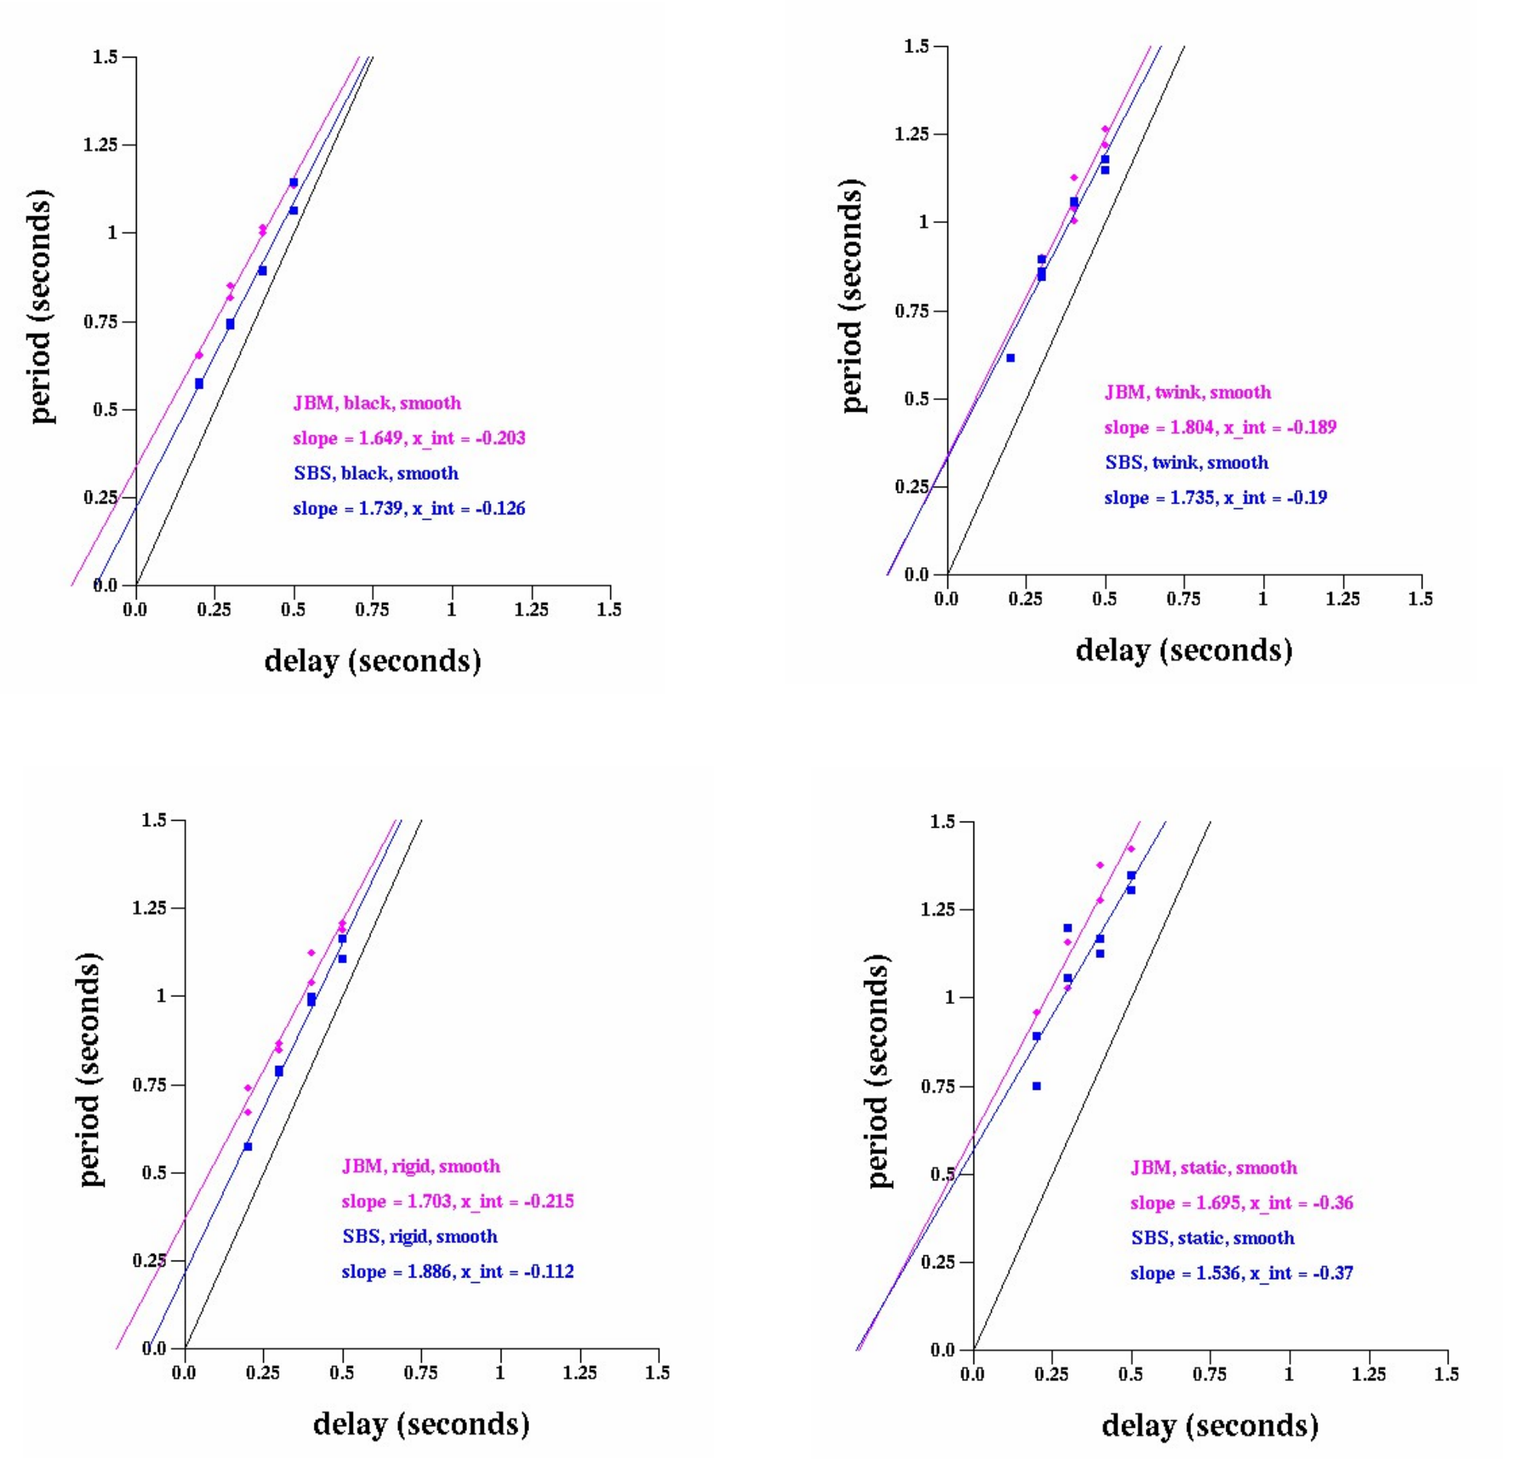

JBM, black, smooth
slope = 1.649, x_int = -0.203
SBS, black, smooth
slope = 1.739, x_int = -0.126
JBM, twink, smooth
slope = 1.804, x_int = -0.189
SBS, twink, smooth
slope = 1.735, x_int = -0.19
JBM, rigid, smooth
slope = 1.703, x_int = -0.215
SBS, rigid, smooth
slope = 1.886, x_int = -0.112
JBM, static, smooth
slope = 1.695, x_int = -0.36
SBS, static, smooth
slope = 1.536, x_int = -0.37
period (seconds)
delay (seconds)

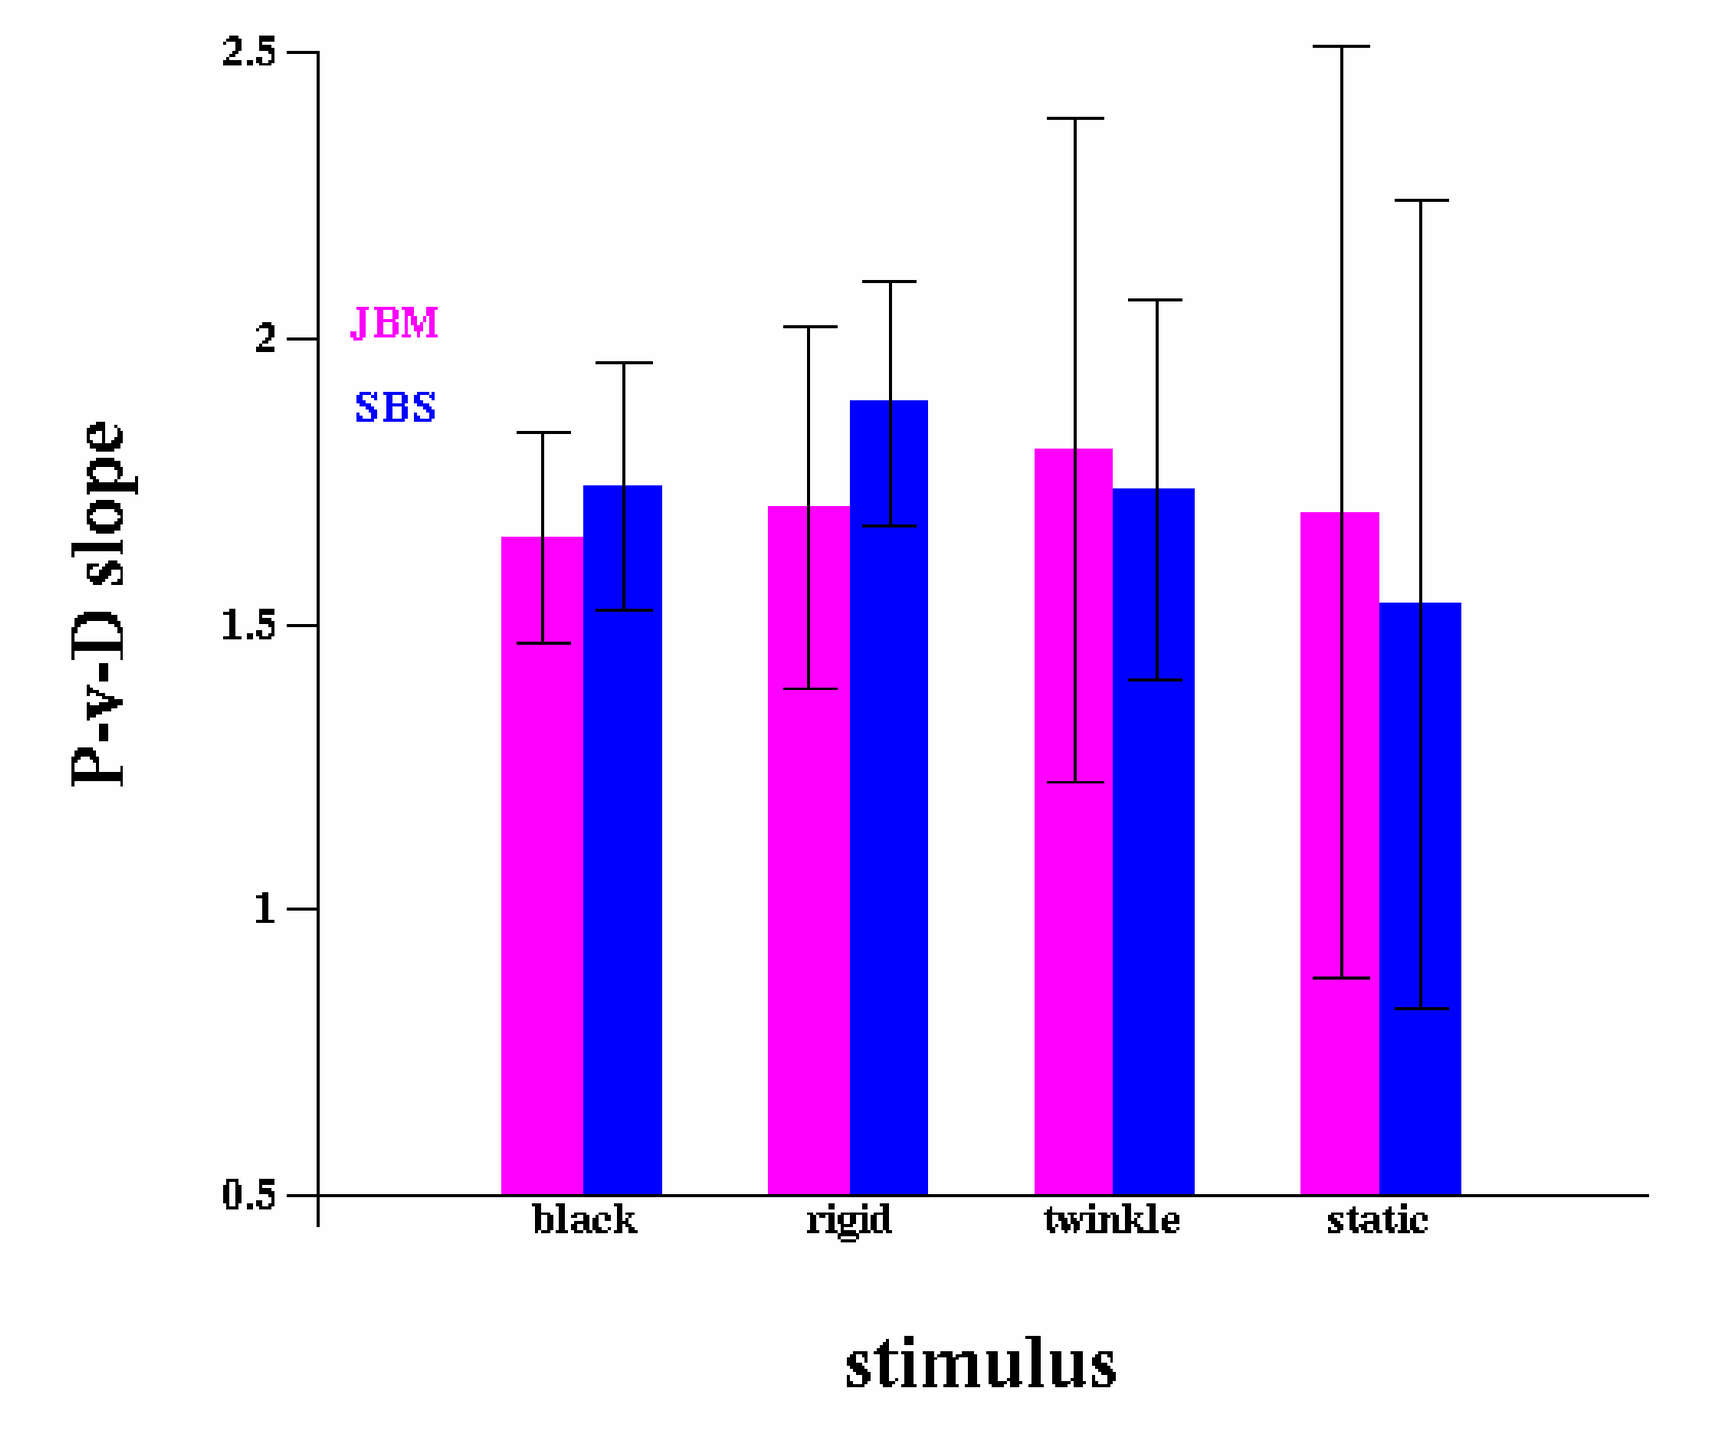

JBM
SBS
P-v-D slope
black
rigid
twinkle
static
stimulus

Conclusion

Can we demonstrate a position-Driven pursuit response for certain Classes of stimuli?

TODAY'S ANSWER: Not yet...

References

Goldreich, D., Krauzlis, R. J., and Lisberger, S. G. (1992). "Effect of changing feedback delay on spontaneous oscillations in smooth pursuit eye movements in monkeys." J. Neurophysiol., **67**(3), 625-638.

Mulligan, J. B., Stevenson, S. B., and Cormack, L. K. (2013). "Reflexive and voluntary control of smooth eye movements." in Rogowitz, B.E. Pappas, T.N., and de Ridder, H. (eds.), *Human Vision and Electronic Imaging XVIII*, Proc. SPIE, v. 8651.