Rapid Assessment of Contrast Sensitivity with Mobile Touch-screens

Jeffrey B. Mulligan, NASA Ames Research Center

Purpose

The need for periodic vision assessment during long-duration space flight has been exposed by the discovery of in- and post-flight vision problems experienced by some astronauts, possibly resulting from elevated cranial pressure, resulting in distention of the optic disc (papilladema), and shortening of the globe with a resulting hyperopic refractive shift. This project seeks to provide easy-to-use tools that allow rapid assessment of contrast sensitivity and other parameters of visual function, using readily-available low-mass equipment..

Approach

The images shown below depicts a “sweep grating,” in which spatial frequency is swept in the horizontal dimension, while contrast is swept in the vertical dimension. An observer's contrast sensitivity function can be traced out as the boundary of the region of visible pattern. One of the first appearances of such an image was in Cornsweet (1970), who attributed it to an unpublished photograph provided by F. Campbell and J. Robson.

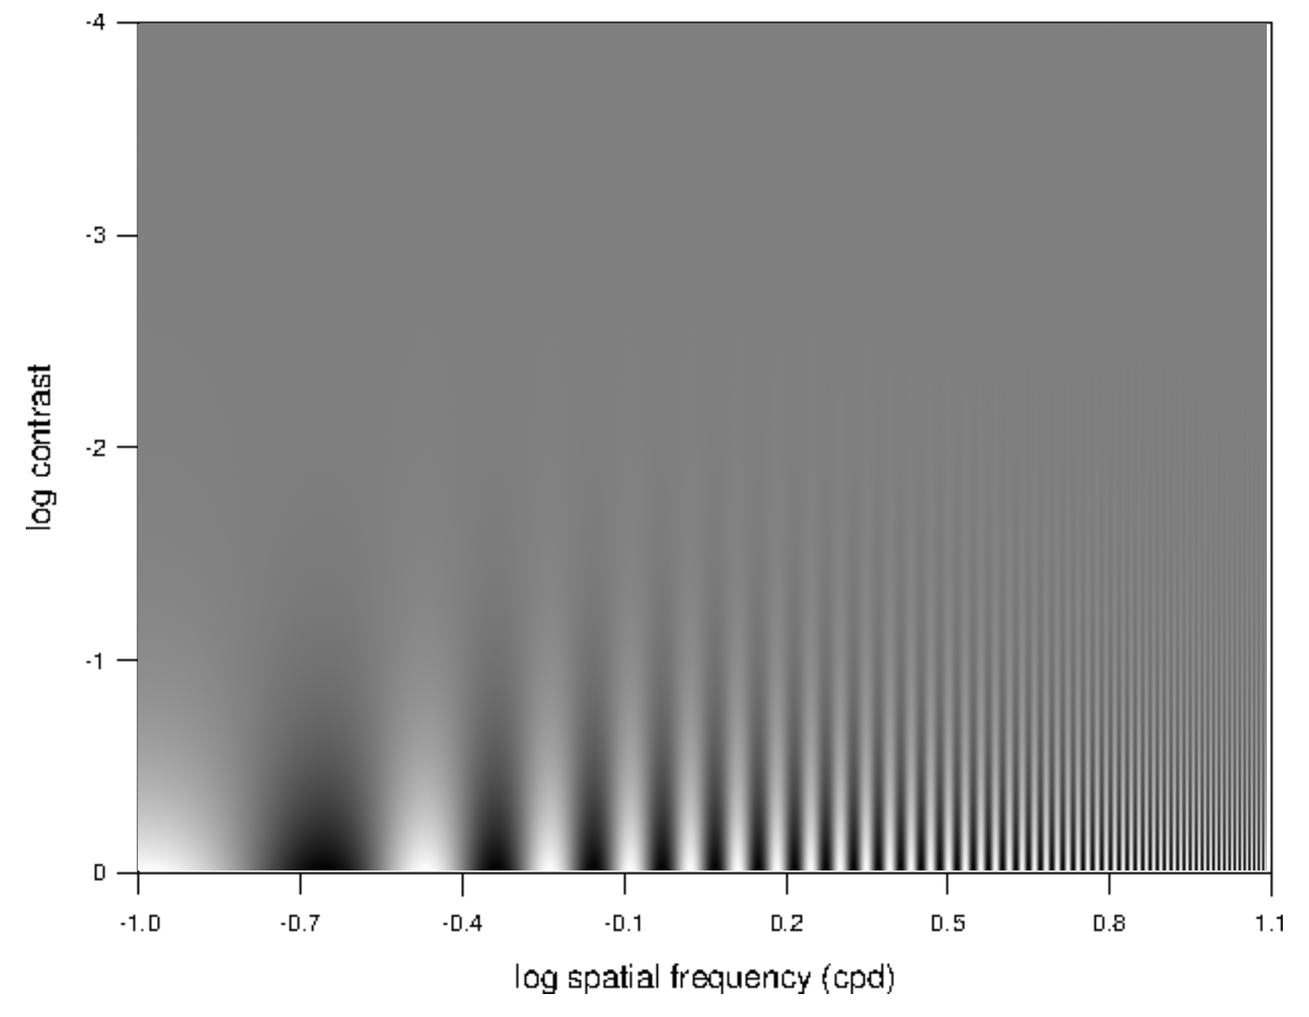

log contrast
log spatial frequency (cpd)

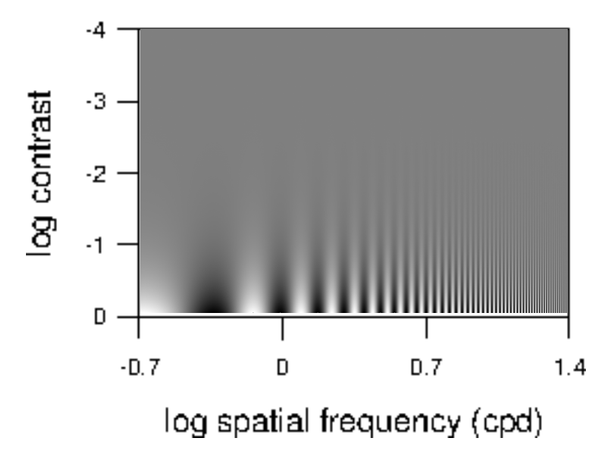

log contrast
log spatial frequency (cpd)

This image has been used to allow an observer to visualize their own contrast sensitivity function (CSF). The touch-screen interface which has become ubiquitous on tablet computers and smart phones allows a subject to quickly indicate this locus with a swipe of the finger over the screen (a “sweep swipe”?), permitting accurate estimation of the CSF with a very small number of trials. The sweep grating on the left approximated the appearance on an iPad, while the grating on the right simulates an iPod touch.

Software implementation

The implementation has been developed using an in-house scripting language known as QuIP (QUick Image Processing). QuIP provides an interpreted environment similar to Matlab, includes an extensive image processing library, and provides linkage to a number of other external libraries. QuIP is publicly available under the NASA Open Source Agreement (NOSA), and may be downloaded from http://scanpath.arc.nasa.gov/quip/. QuIP has been developed primarily for Unix/X11 platforms, and was ported to Apple Computer's iOS for this project.

Devices

The initial implementation demonstrated here has been done for Apple Computer’s iOS operating system, which runs on the popular iPad and iPod devices. The table below shows the display parameters of the models used. Calculations of max. and min. spatial frequencies were performed assuming a viewing distance of 20 inches, a minimum period of four pixels and a maximum period of half the largest dimension of the screen.

Device	iPad2	iPad4	iPad mini	iPod4	iPod5
Screen size (inches)	9.7	9.7	7.9	3.5	4.0
Screen resolution (pixels)	1024 x 768	2048 x 1536	1024 x 768	960 x 640	1136 x 640
Pixel pitch (pixels per inch)	132	264	163	326	326
Min spatial freq. (cpd)	0.09	0.09	0.11	0.24	0.20
Max. spatial freq. (cpd)	11.5	23	14.2	28.4	28.4

Calibration

To facilitate field checks of calibration (and recalibration if needed), a suite of psychophysically-based calibration procedures have been developed. Linearization (gamma correction) is performed by making luminance matches between uniform patches and dithered patches (under the assumptions of spatial independence). This can be done either by matching static patches or using a motion-nulling technique (Mulligan, 2009), illustrated in the figure above, which is based on on a method introduced by Anstis and Cavanagh (1983). Once linearization has been performed, the relative luminosities of the color primaries and be determined using hetero-chromatic flicker photometry (HFP) or the Anstis and Cavanagh motion-nulling procedure.

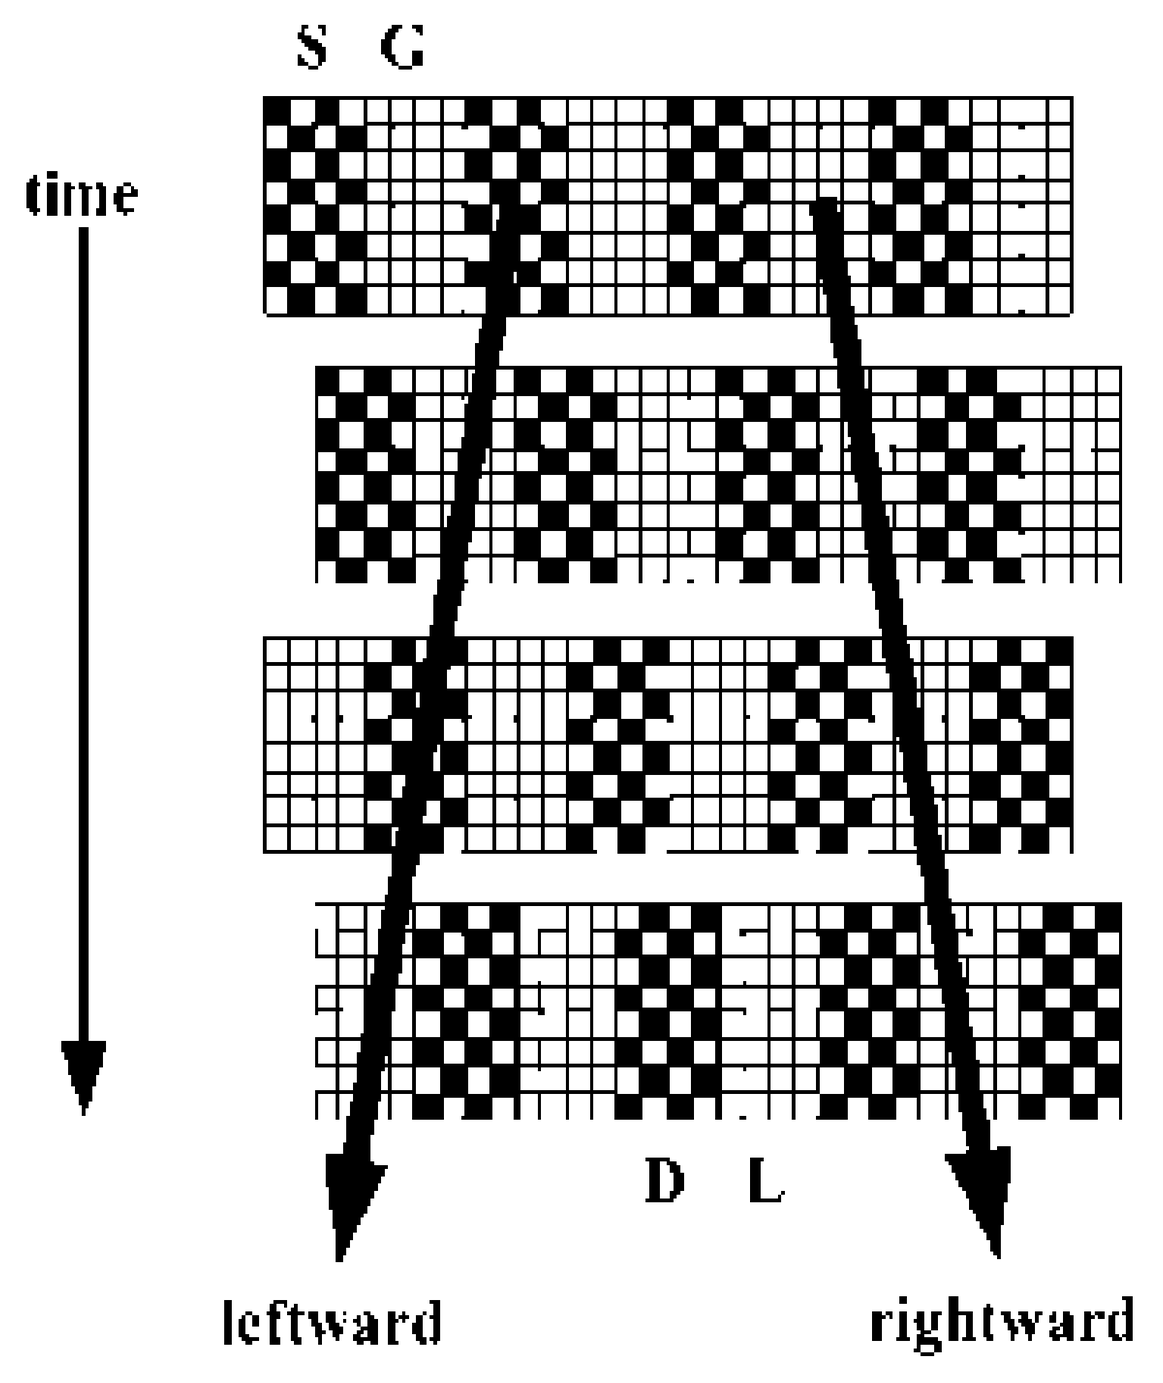

S G
time
D L
leftward
rightward

Schematic diagram showing the four frame sequence used for gray level bisection. Stippled (S) and gray (G) stripes alternate with light (L) and dark (D) stripes in spatial quadrature; the perceived direction of motion depends on the luminance of the variable gray pixels. (figure from Mulligan, 2009)

Stimulus Rendering

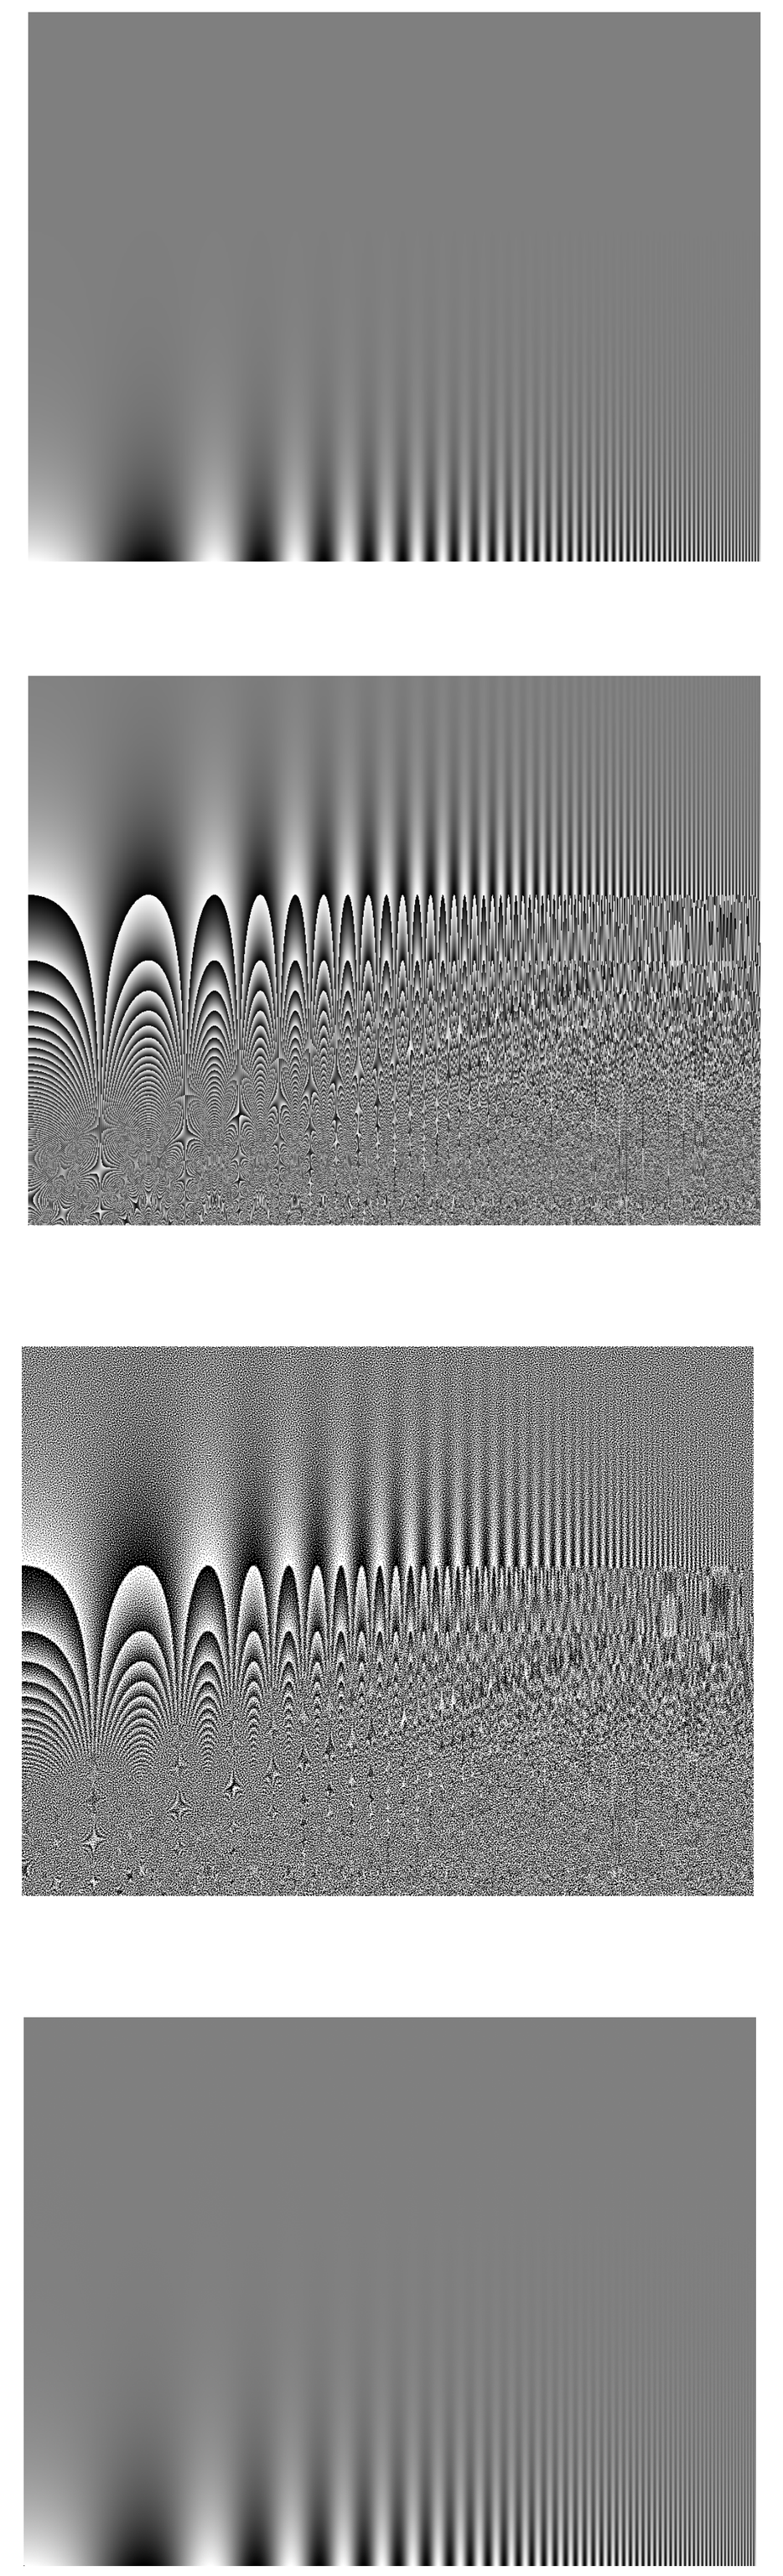

The limited (8-bit) gray level resolution of the devices poses challenges for the rendering of threshold level stimuli. On the right we see a sweep grating where each pixel has been quantized to 8 bits. In the next panel below, the quantization error is displayed (normalized to the full display range for clarity).

The quantization error is then converted to a 1-bit image using an iterative optimization algorithm which seeks to minimize the filtered error (Mulligan & Ahumada, 1992). This image is added to the quantized image shown at the top to produce the final image.

Additional improvements can be obtained by extending the dithering principle to the time domain, and by shifting error from luminance to chrominance (Mulligan, 1990; Tyler, 1997).

CSF Estimation

Although the entire CSF may in principle be obtained with a single swipe, we may wish to obtain an estimate of the precision of the measurement, by analyzing the variability of repeated measures. Additionally, we have no way of knowing whether the subject is responding honestly, or trying to conceal a vision loss by deliberately swiping in the invisible area. Both of these concerns may be addressed by presentations of sweep gratings rendered with different parameters. Although it is common for the variations of contrast and spatial frequency to occur in orthogonal directions, we can add variety by tilting the contrast dimension. Several examples are shown below.

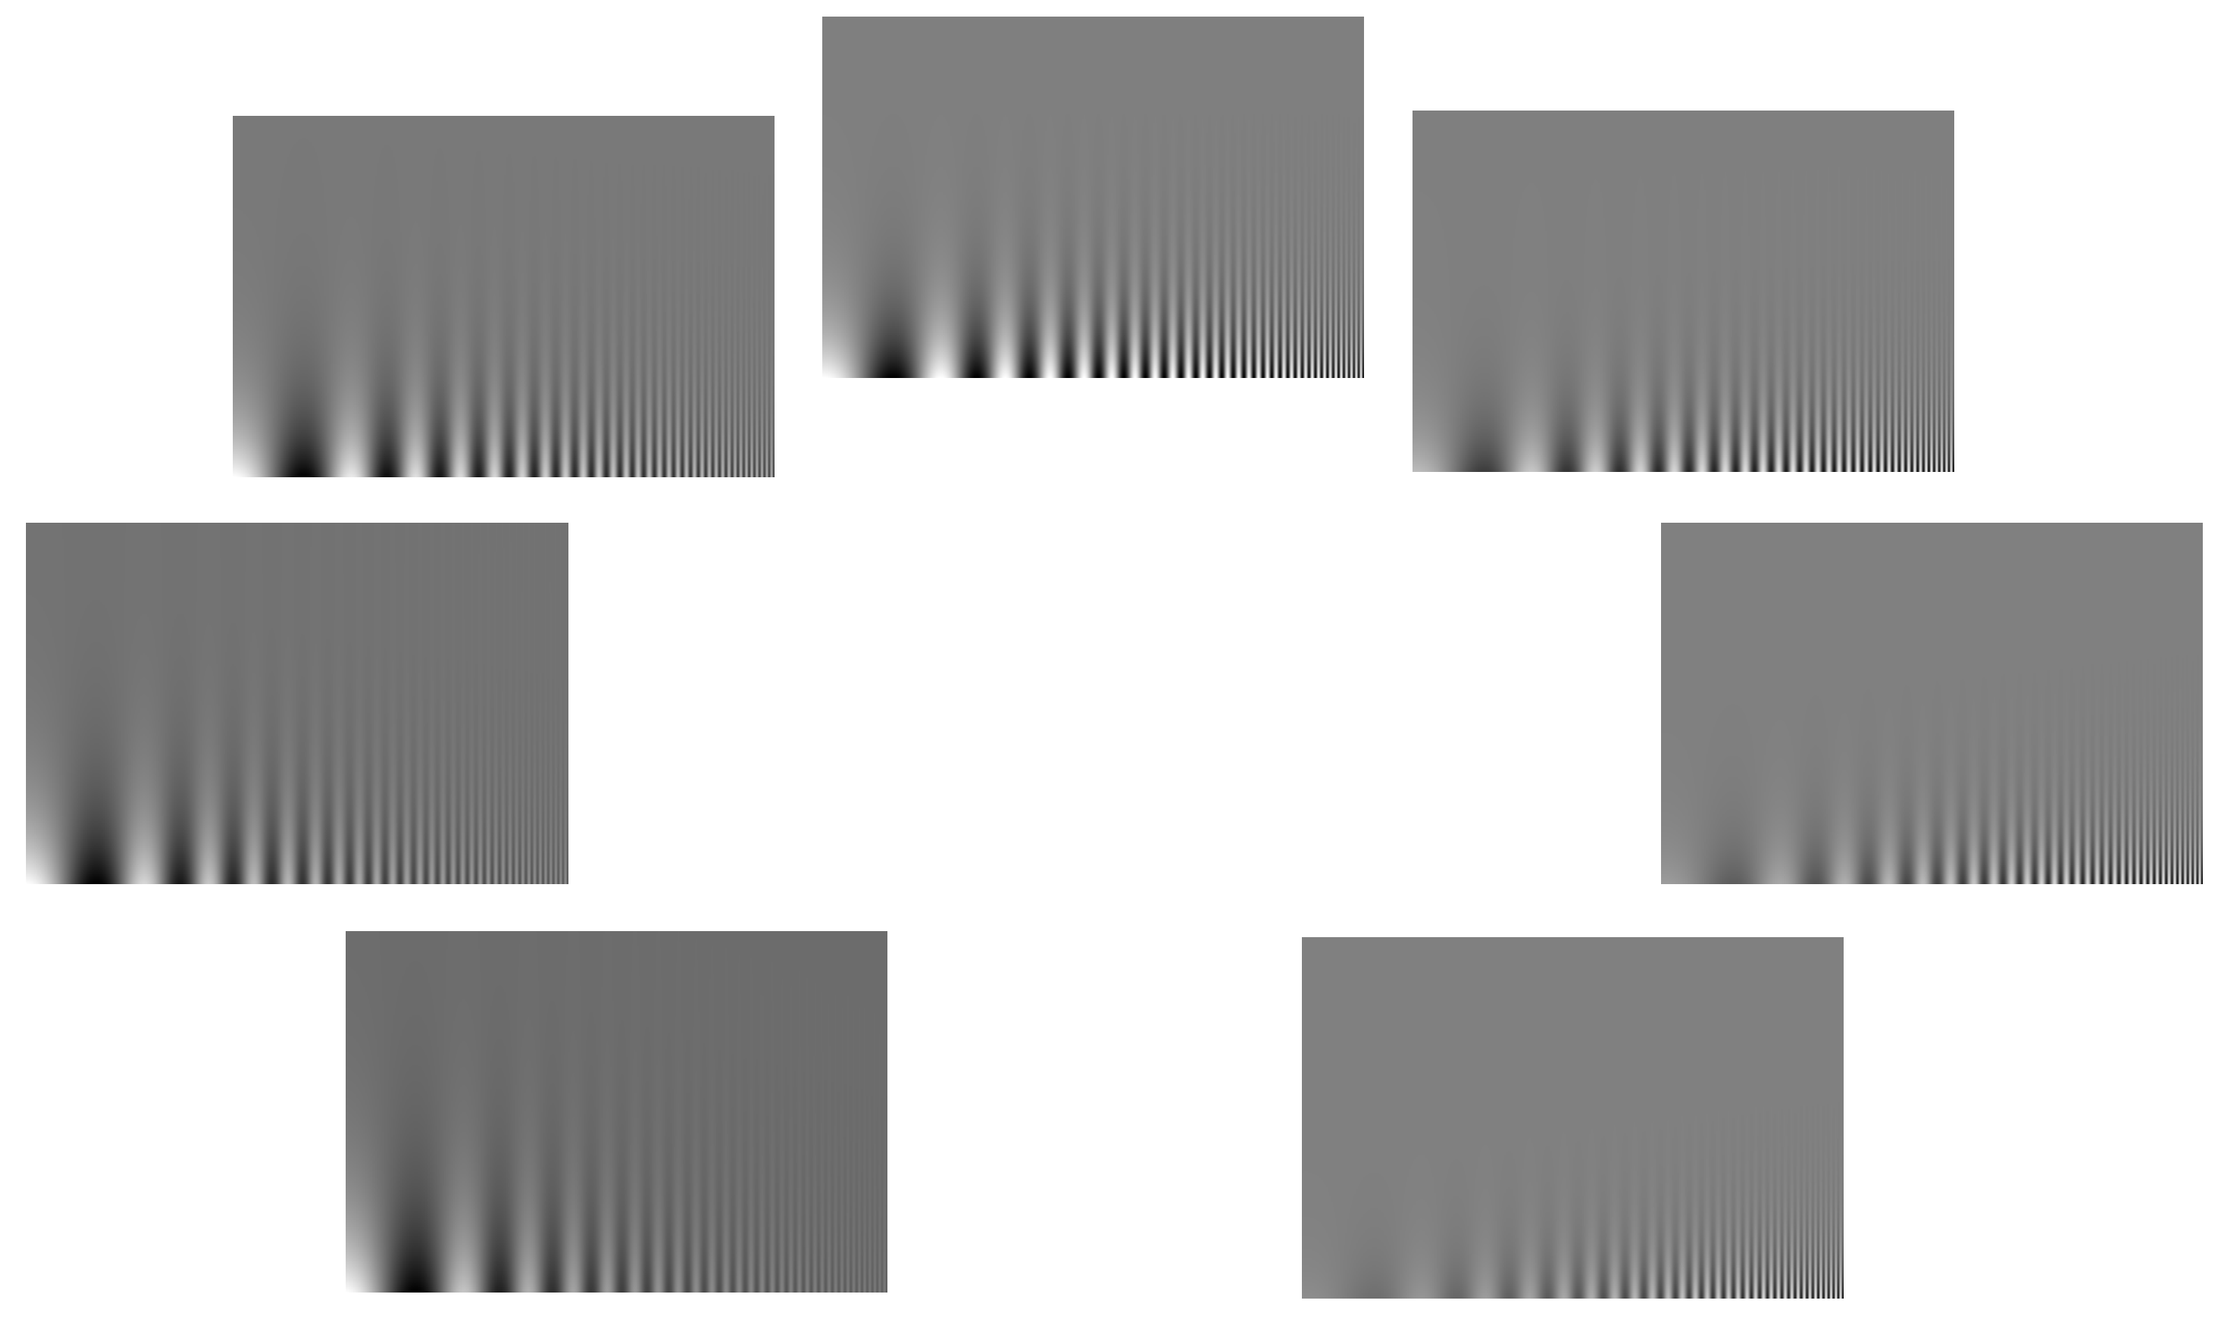

Traditionally, the CSF has been measured by first estimating threshold at a set of distinct frequencies, and then fitting a curve to the threshold estimates. Lesmes and colleagues have made significant improvements in the efficiency of this process by estimating the form of the CSF directly from a small number of discrete trials, without the intermediate step of estimating particular thresholds (Lesmes *et al*., 2010; Lesmes & Lu, 2011; Dorr *et al*., 2012). We employ a similar method, fitting parametric curve (a parabola) to data from one or more swipes.

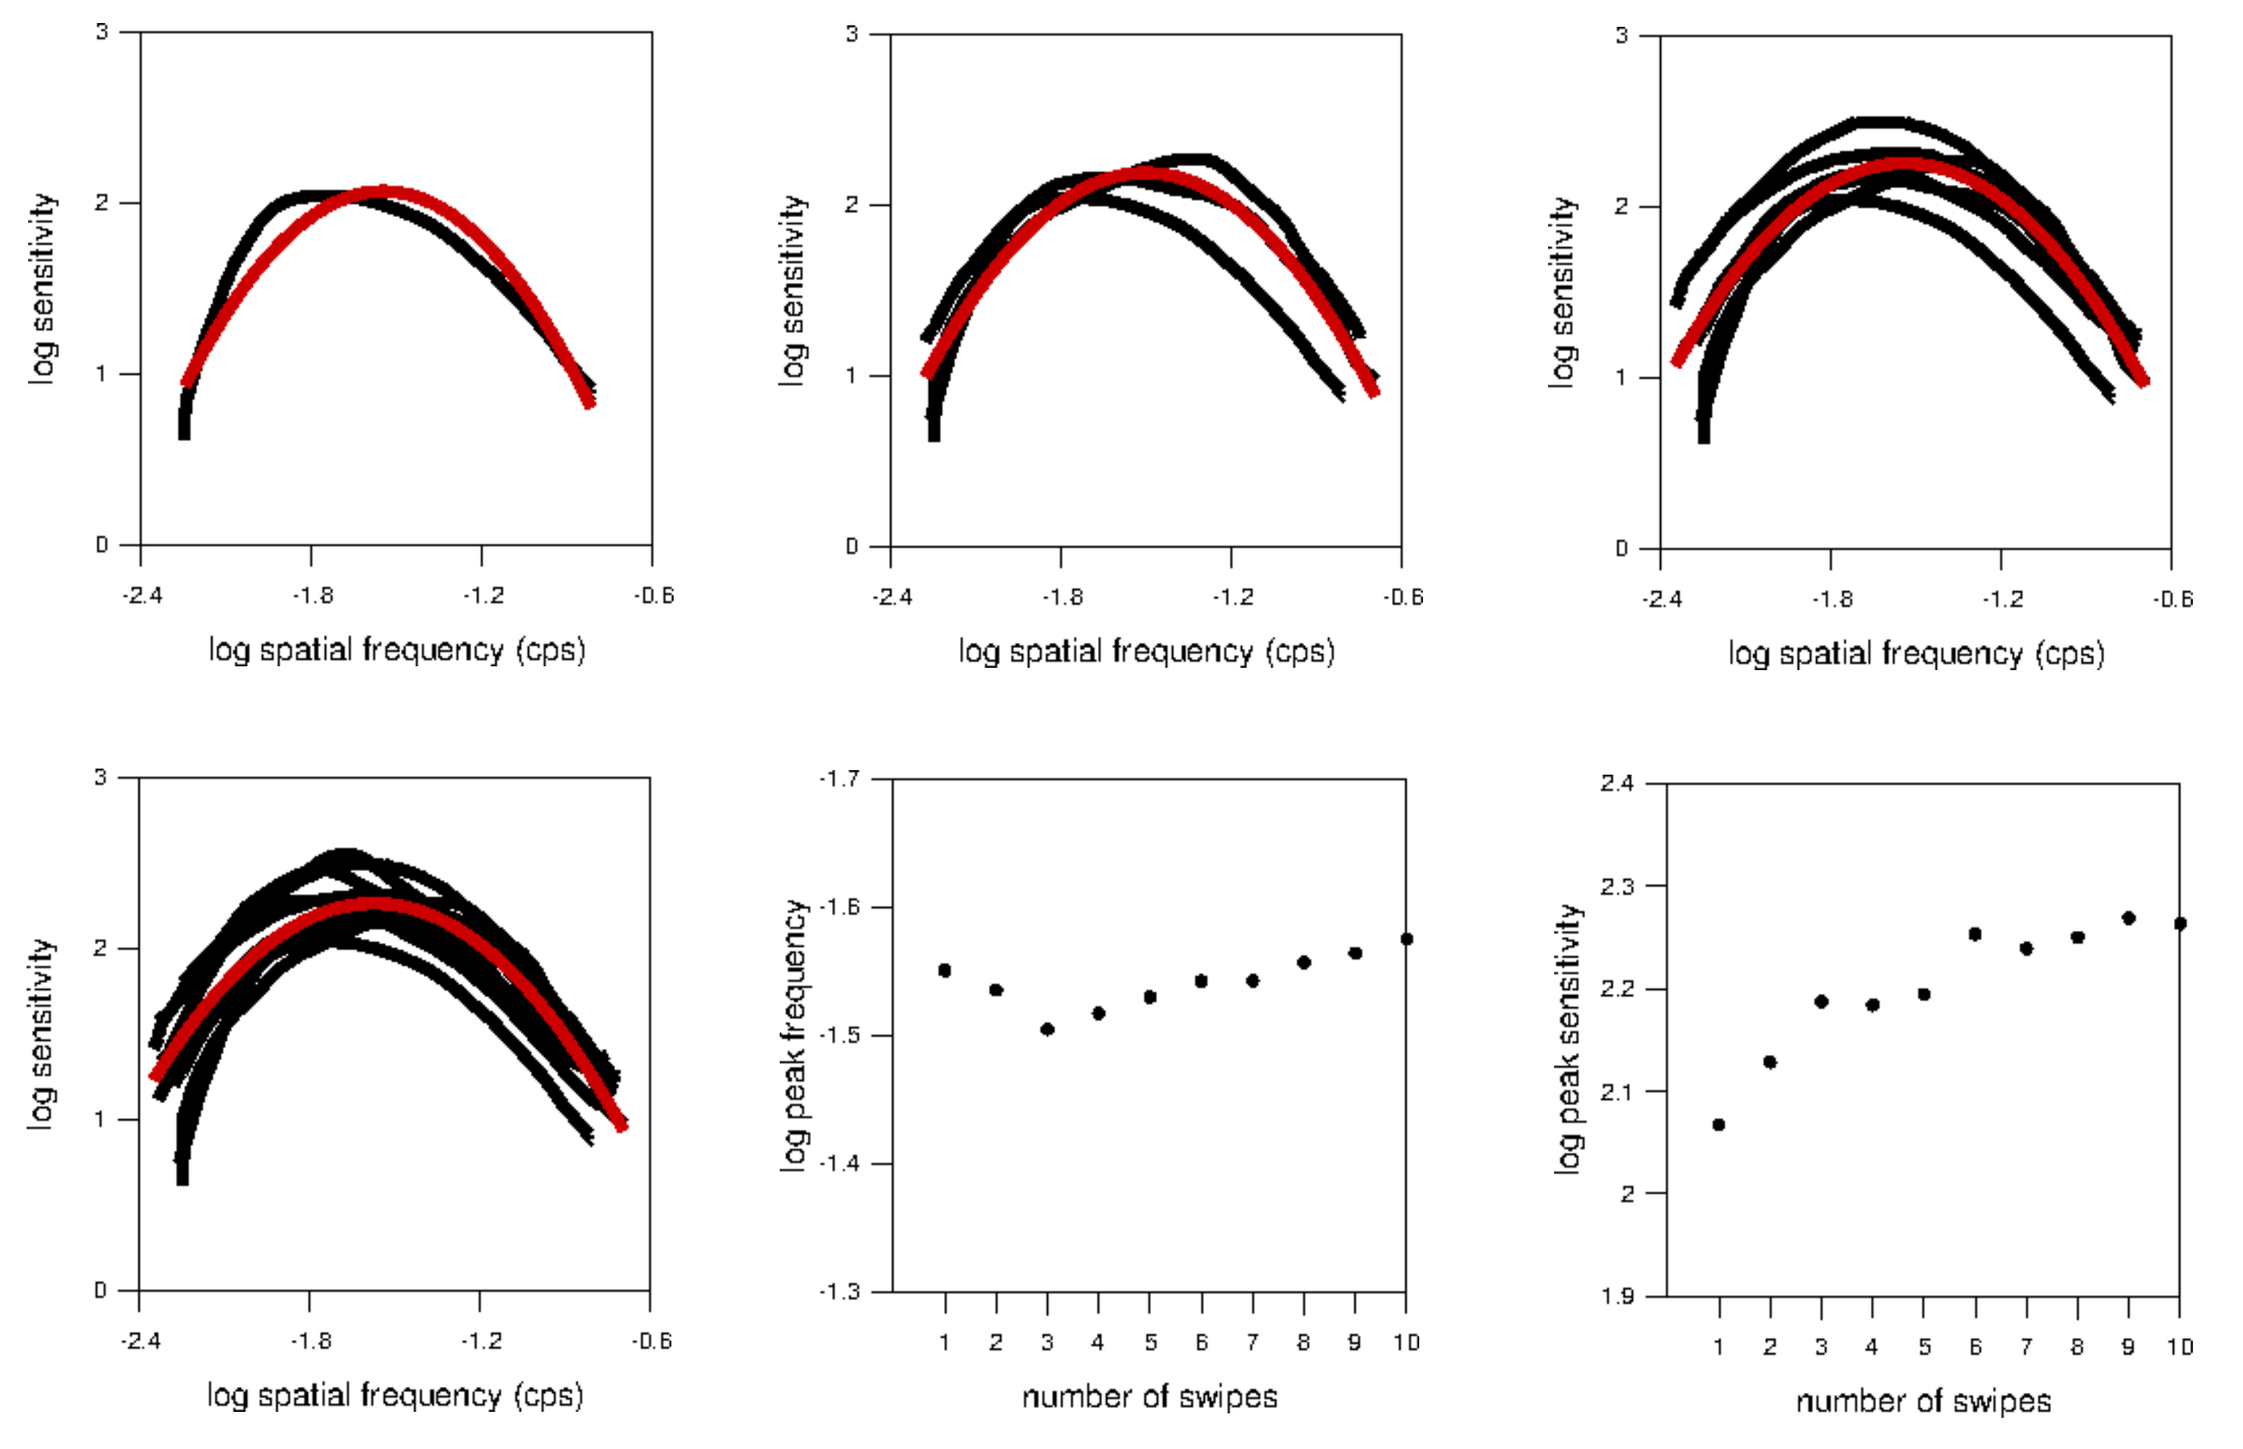

log sensitivity
log spatial frequency (cps)
log peak frequency
log peak sensitivity
number of swipes

Conclusion

Much work remains to be done to develop a robust system – collaborators and beta-testers welcome! The results presented here demonstrate proof-of-concept that the touch-screen interface can enable new efficient psychophysical methods.

[this space reserved for special late-breaking results]

References

Anstis, S. and Cavanagh, P. (1983). “A minimum motion technique for judging equiluminance,” in *Colour vision: psychophysics and physiology*, Mollon, J. B. and Sharpe, L. T. (eds.), pp. 155-166, Academic Press.

Cornsweet, T. N. (1970). *Visual Perception*. Academic Press, New York, p. 343.

Dorr, M., Lesmes, L., To, L., Lu, Z. L. and Bex, P. J. (2012). “High-precision psychophysics on the iPad,” *Perception*, **41** (*ECVP Abstract supplement*), p. 98.

Lesmes, L. A. and Lu, Z.-L. (2011). “Methods and devices for rapid measurement of visual sensitivity,” U.S. Patent #7938538.

Lesmes, L. A., Lu, Z.-L., Baek, J. and Albright, T. D. (2010). “Bayesian adaptive estimation of the contrast sensitivity function: The quick CSF method,” J. Vis., 10(3), pp. 17.1-17.21.

Mulligan, J. B. (2009). “Presentation of calibrated images over the web,” *Proc. SPIE 7240, Human Vision and Electronic Imaging XIV.*

Mulligan, J. B. and Ahumada, A. J., Jr. (1992). “Principled halftoning based on human vision models,” *Proc. SPIE 1666, Human Vision, Visual Processing, and Digital Display III.*

Tyler, C. W. (1997). “Colour bit-stealing to enhance the luminance resolution of digital displays on a single pixel basis,” *Spatial Vision*, **10**(4), pp. 369-377.

Supported by the NASA Engineering & Safety Center (NESC), and the System-wide Safety Assurance Technologies (SSAT) project of NASA's Aviation Safety Program.