Assessing visual delays using pupil oscillations

Jeffrey B. Mulligan, NASA Ames Research Center

BACKGROUND

The pupil system

The pupil: more than just light adaptation

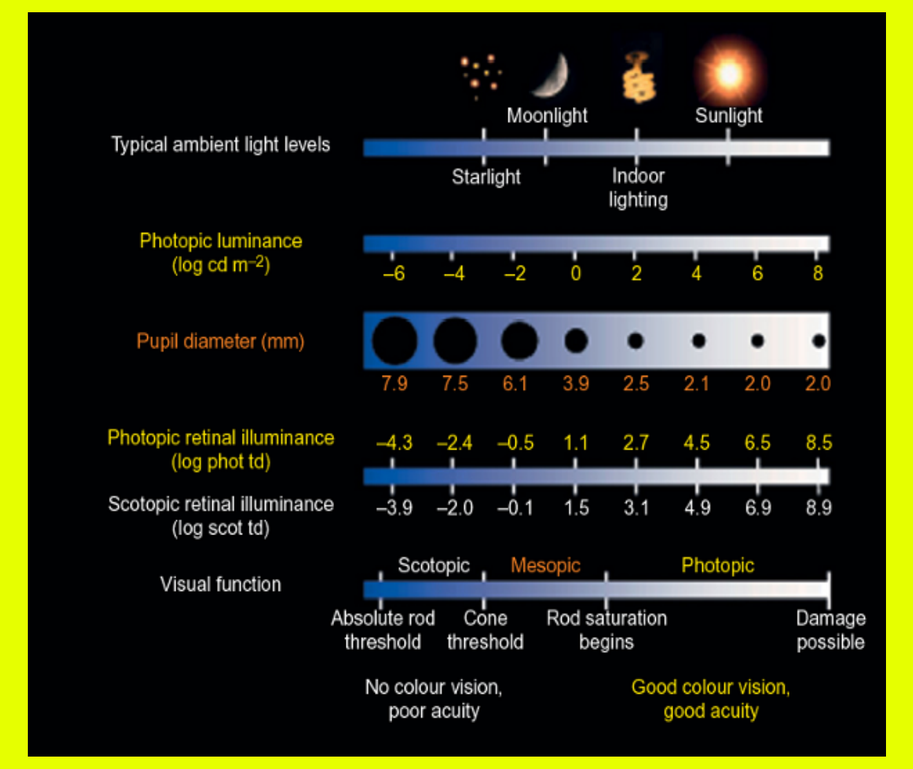

Moonlight
Sunlight
Typical ambient light levels
Starlight
Indoor lighting
Photopic luminance (log cd m−2)
−6 −4 −2 0 2 4 6 8
Pupil diameter (mm)
7.9 7.5 6.1 3.9 2.5 2.1 2.0 2.0
Photopic retinal illuminance (log phot td)
−4.3 −2.4 −0.5 1.1 2.7 4.5 6.5 8.5
Scotopic retinal illuminance (log scot td)
−3.9 −2.0 −0.1 1.5 3.1 4.9 6.9 8.9
Visual function
Scotopic
Mesopic
Photopic
Absolute rod threshold
Cone threshold
Rod saturation begins
Damage possible
No colour vision, poor acuity
Good colour vision, good acuity

Figure from Barbur & Stockman chapter in Encyclopedia of the Eye (2010)

Dilation: sympathetic nervous system (fight or flight)

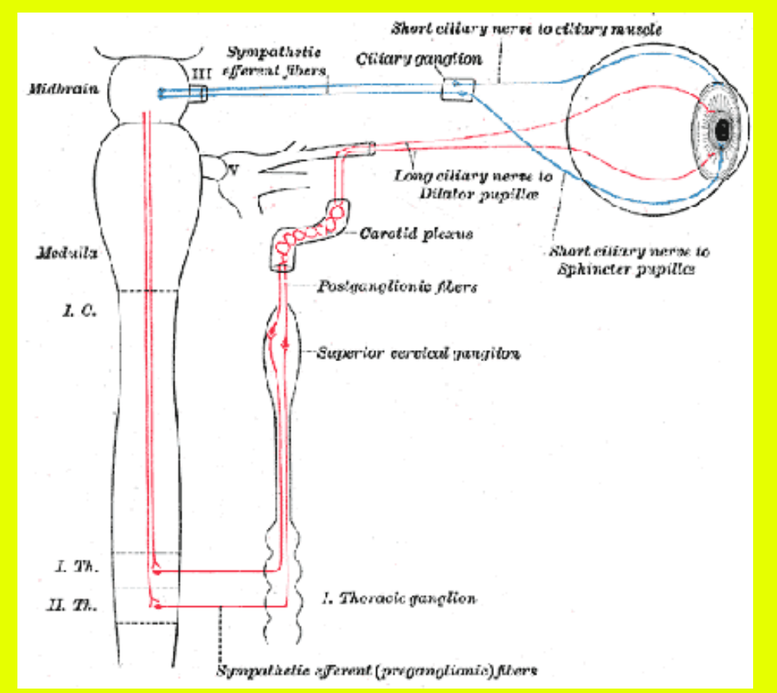

Figure from Gray's Anatomy (1918)

Constriction: parasympathetic nervous system (rest and digest)

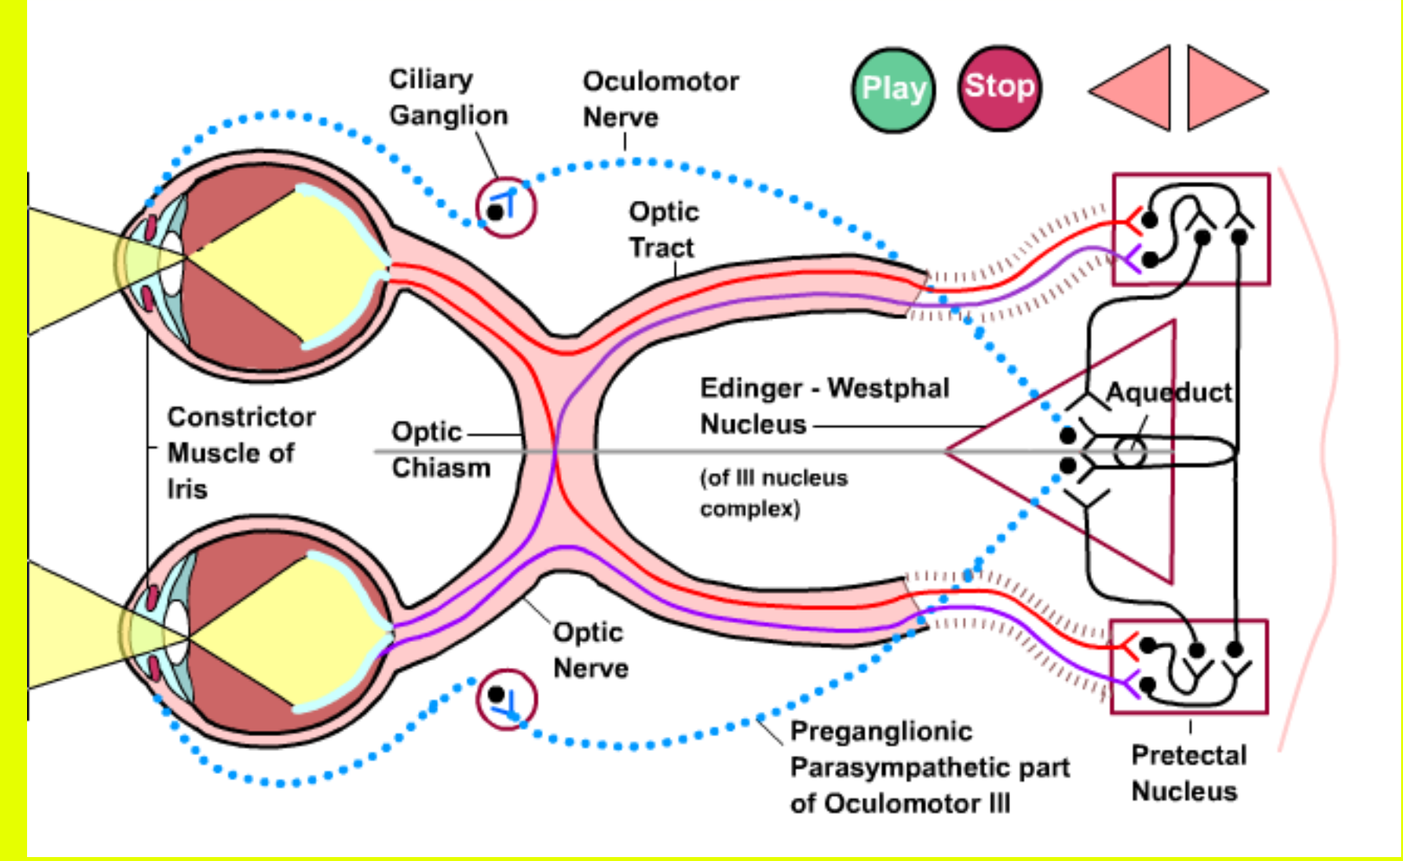

Ciliary Ganglion
Oculomotor Nerve
Play
Stop
Optic Tract
Edinger - Westphal Nucleus
(of III nucleus complex)
Aqueduct
Constrictor Muscle of Iris
Optic Chiasm
Optic Nerve
Preganglionic Parasympathetic part of Oculomotor III
Pretectal Nucleus

Figure captured from animated online demo at tedmontogomery.com

Stark's experiment (1962)

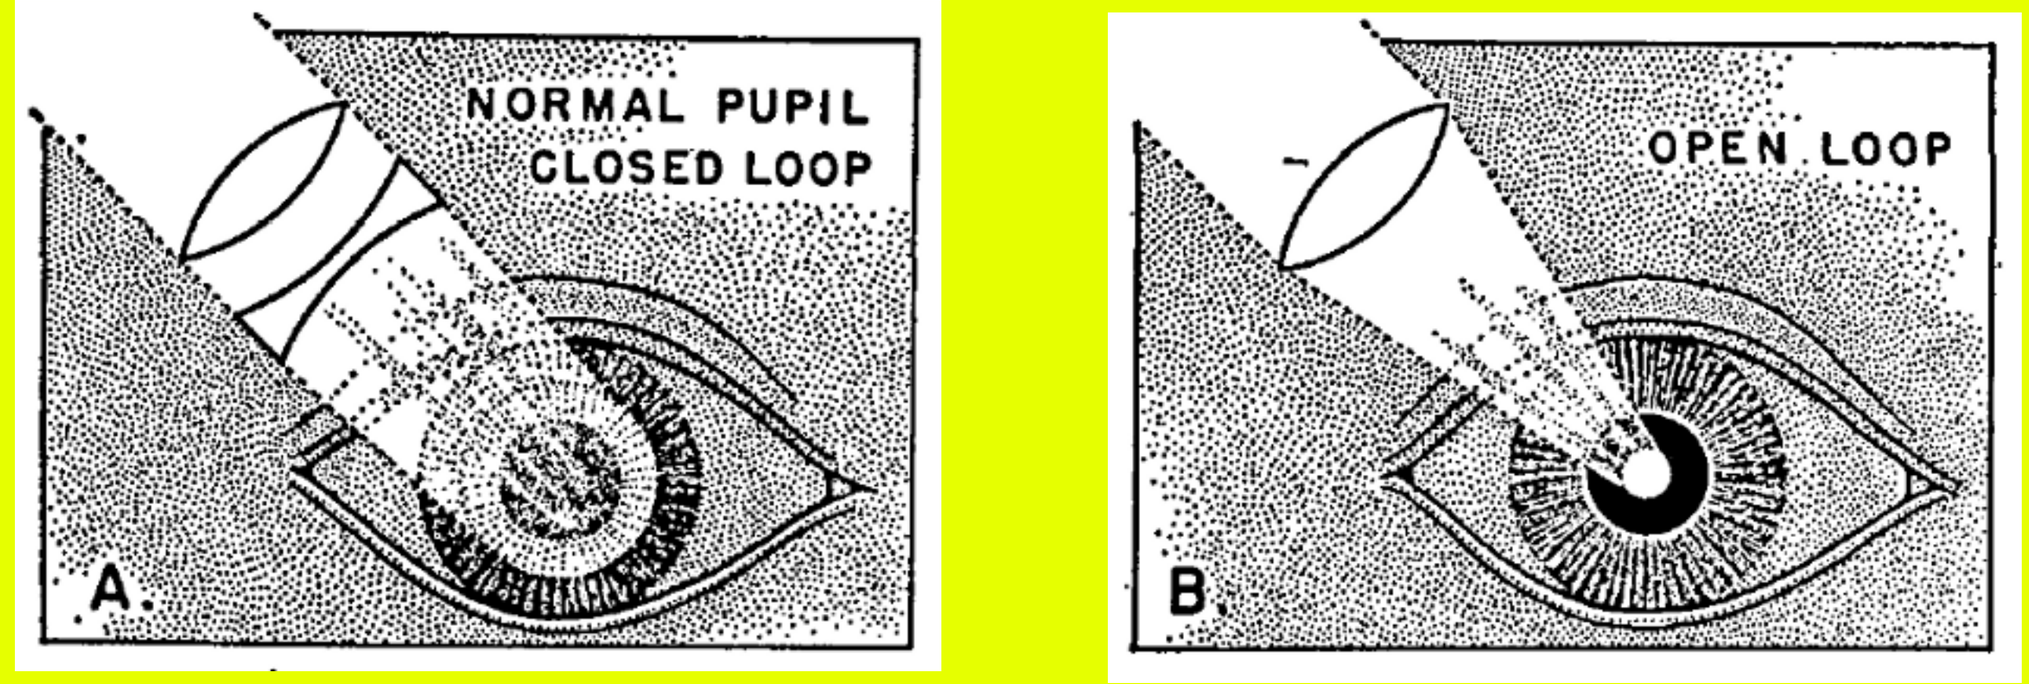

NORMAL PUPIL
CLOSED LOOP
A.
OPEN LOOP
B.

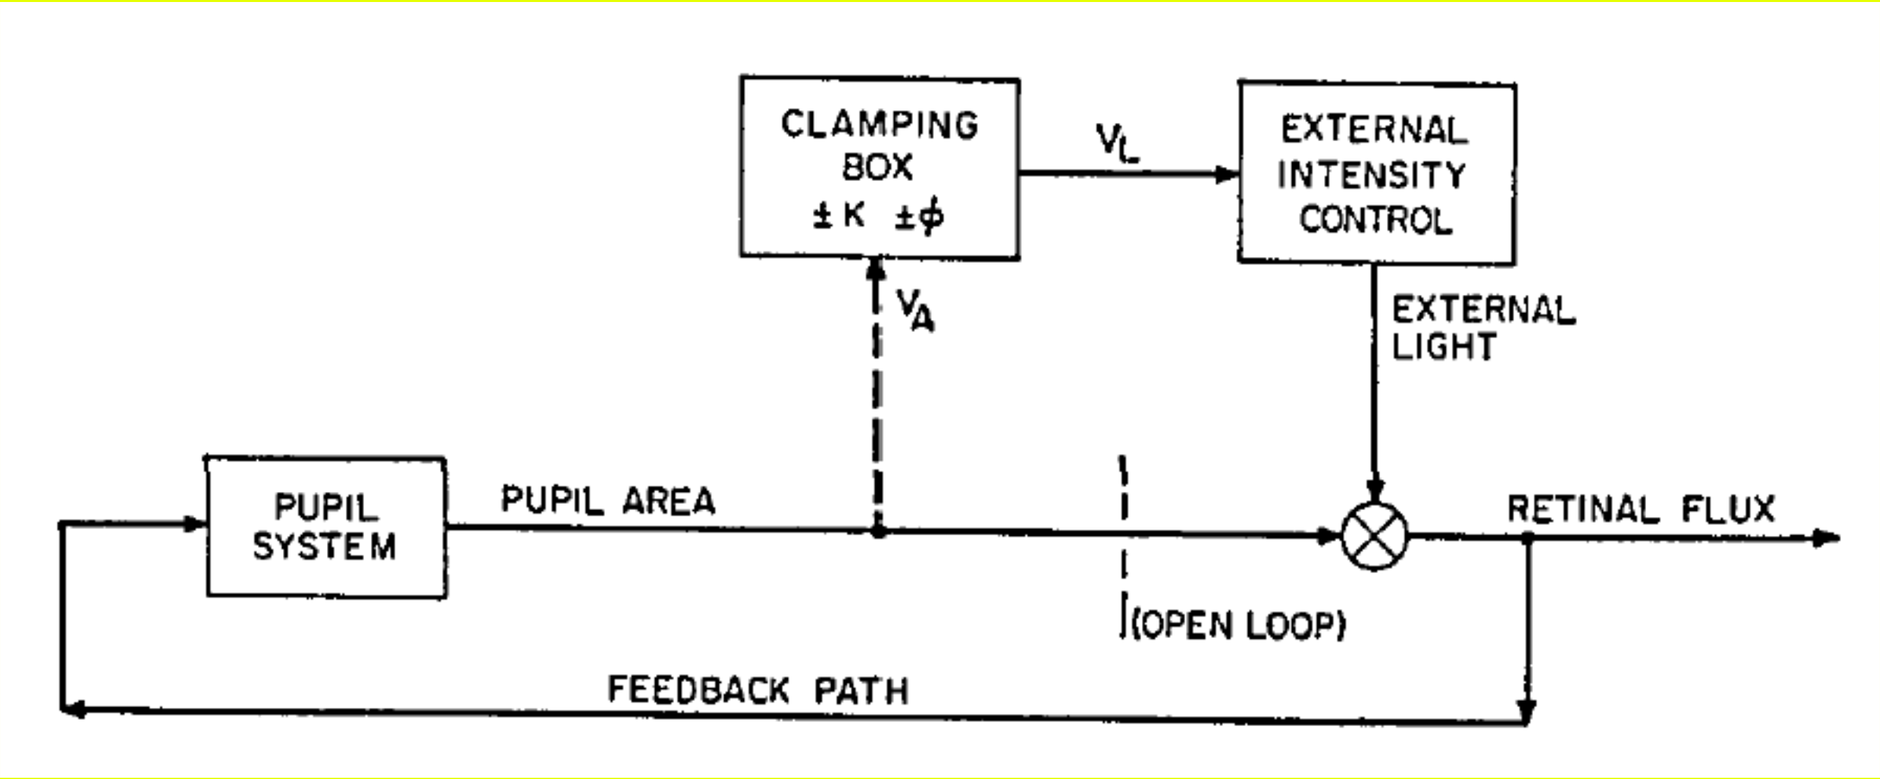

CLAMPING BOX ±K ±ϕ
VL
EXTERNAL INTENSITY CONTROL
VA
EXTERNAL LIGHT
PUPIL SYSTEM
PUPIL AREA
RETINAL FLUX
(OPEN LOOP)
FEEDBACK PATH

Figures from Stark (1962)

The pupil and cognition

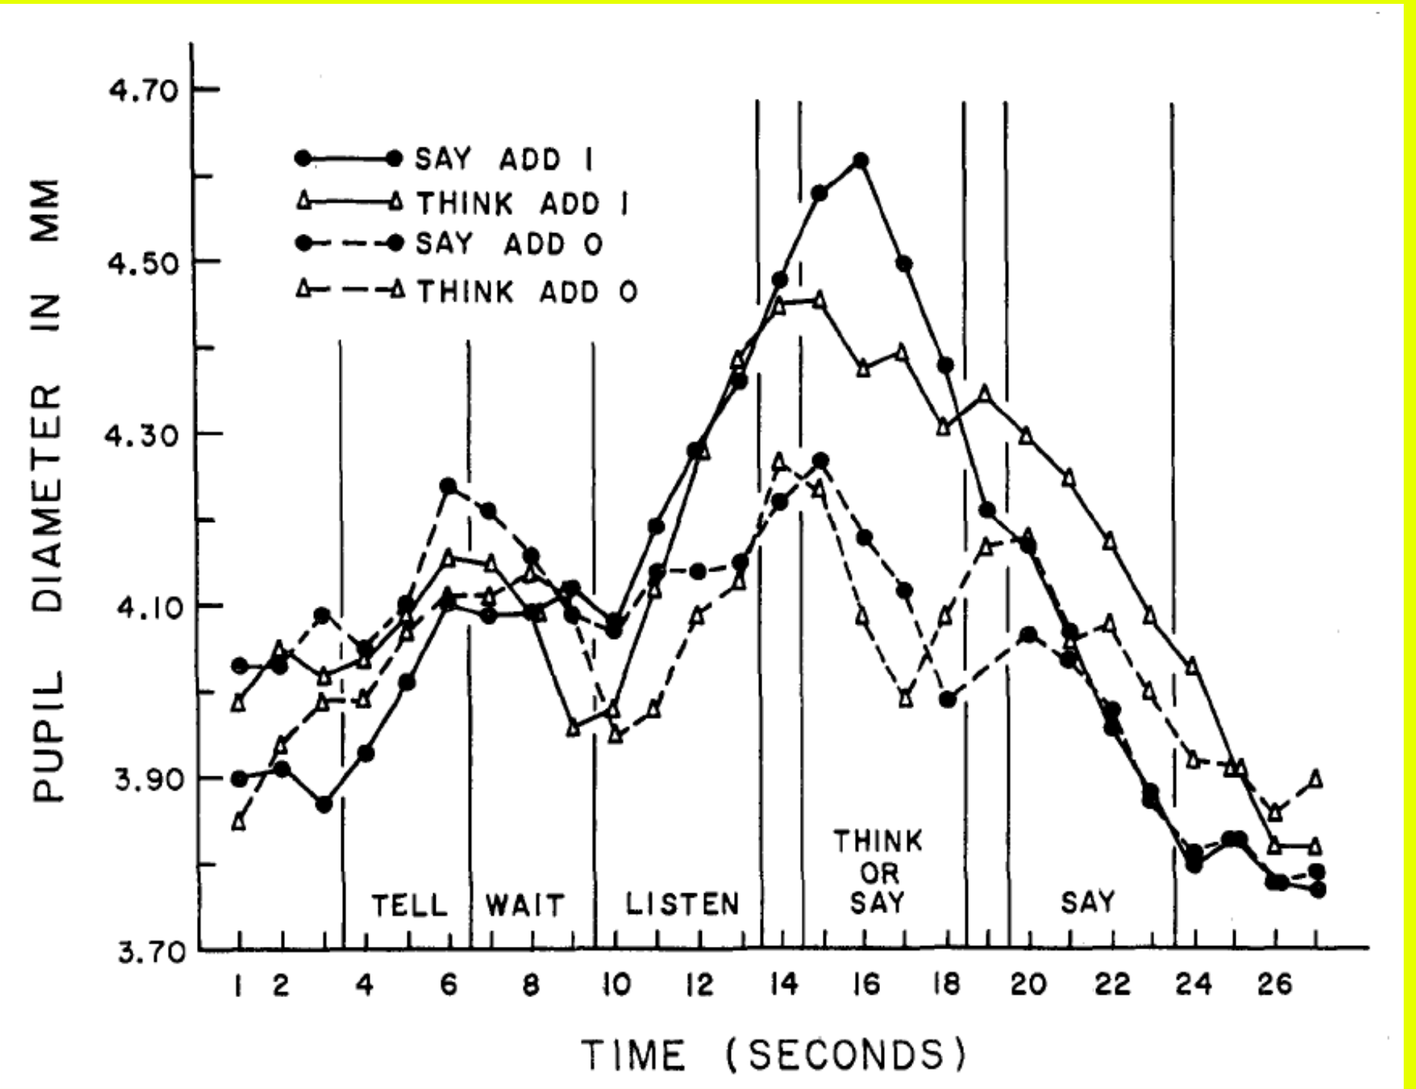

SAY ADD 1
THINK ADD 1
SAY ADD 0
THINK ADD 0
PUPIL DIAMETER IN MM
TELL
WAIT
LISTEN
THINK OR SAY
SAY
TIME (SECONDS)

Figure from Kahneman, Peavler & Onuska (1968)

Pupil pattern responses

Pupil grating response

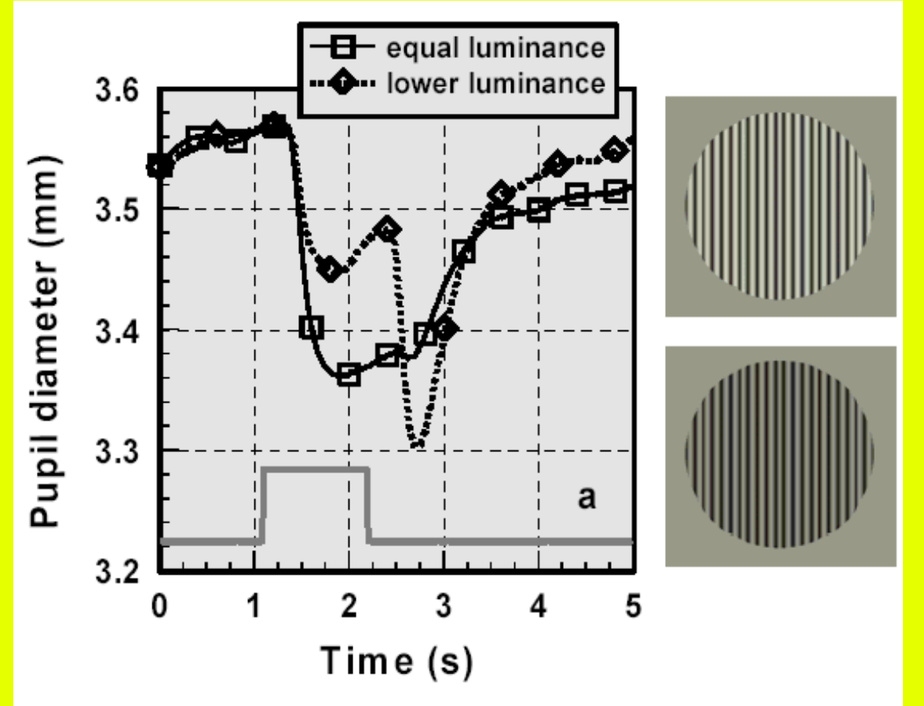

equal luminance
lower luminance
Pupil diameter (mm)
Time (s)
a

Pupil color response

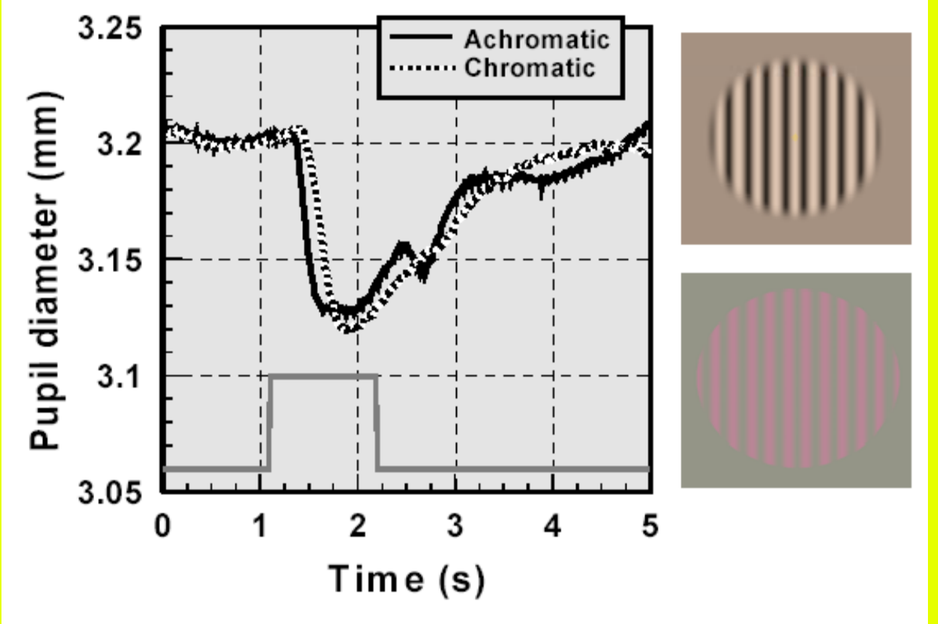

Achromatic
Chromatic
Pupil diameter (mm)
Time (s)

Figures from Barbur (2004)

Melanopsin and ipRGC's

Sustained pupil response in macaque mimics activity of ipRGC's

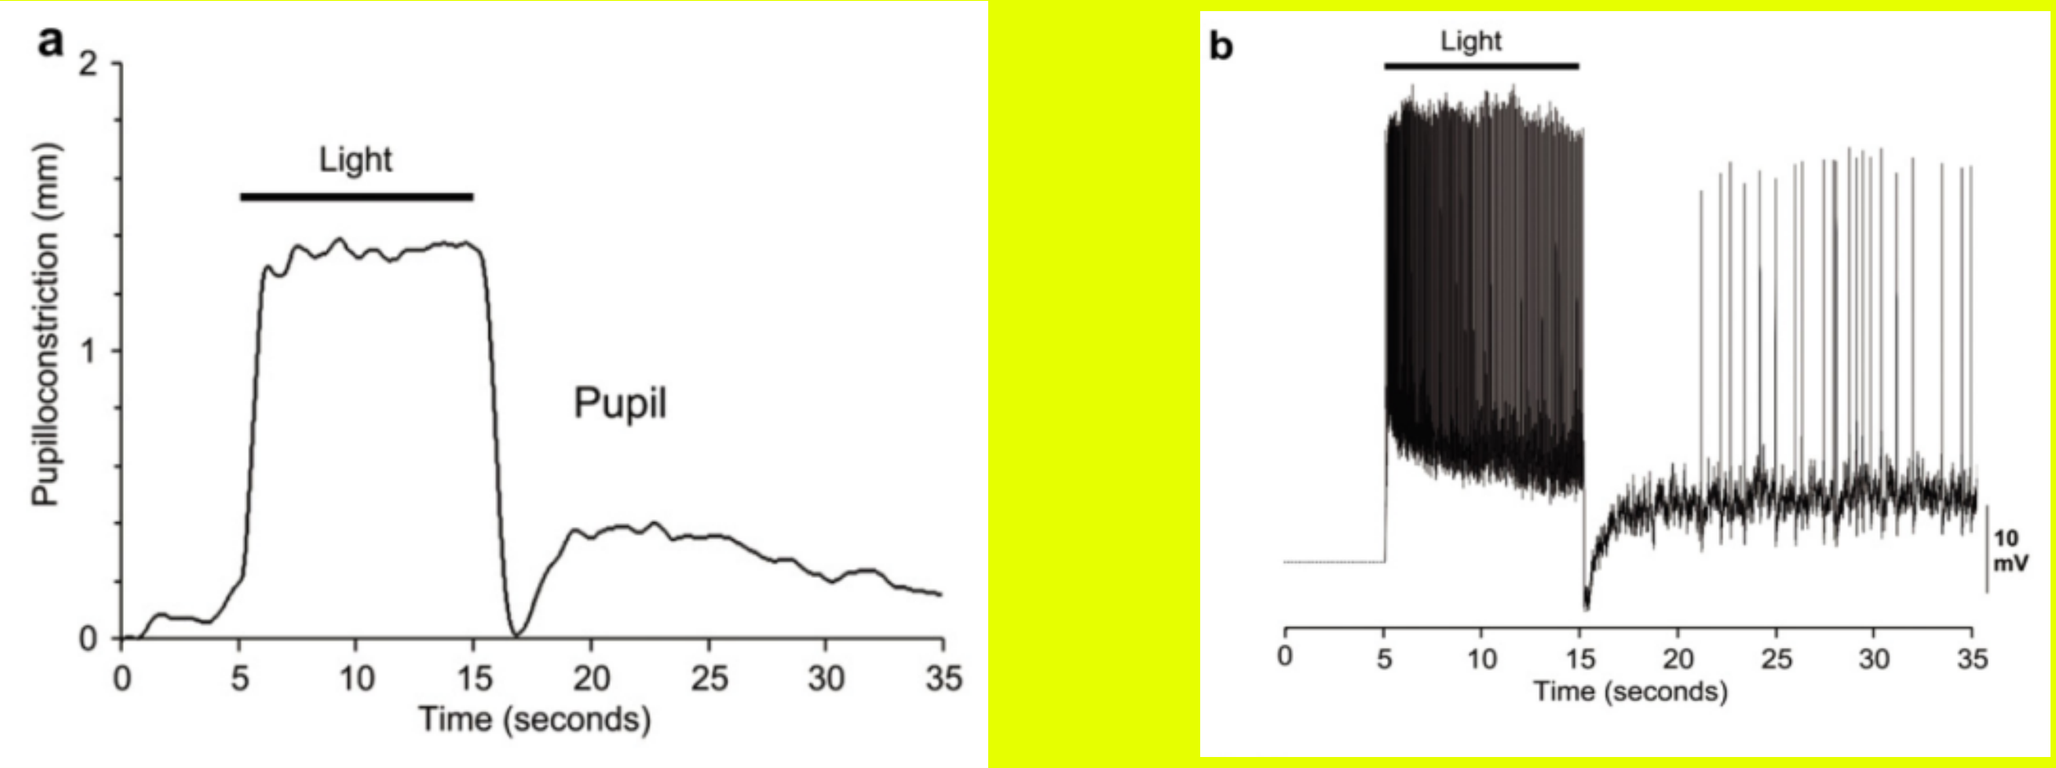

a
Light
Pupil
Pupilloconstriction (mm)
Time (seconds)
b
Light
10 mV
Time (seconds)

Figures from Gamlin *et al.* (2007)

Melanopsin action spectrum from sustained pupil response in human

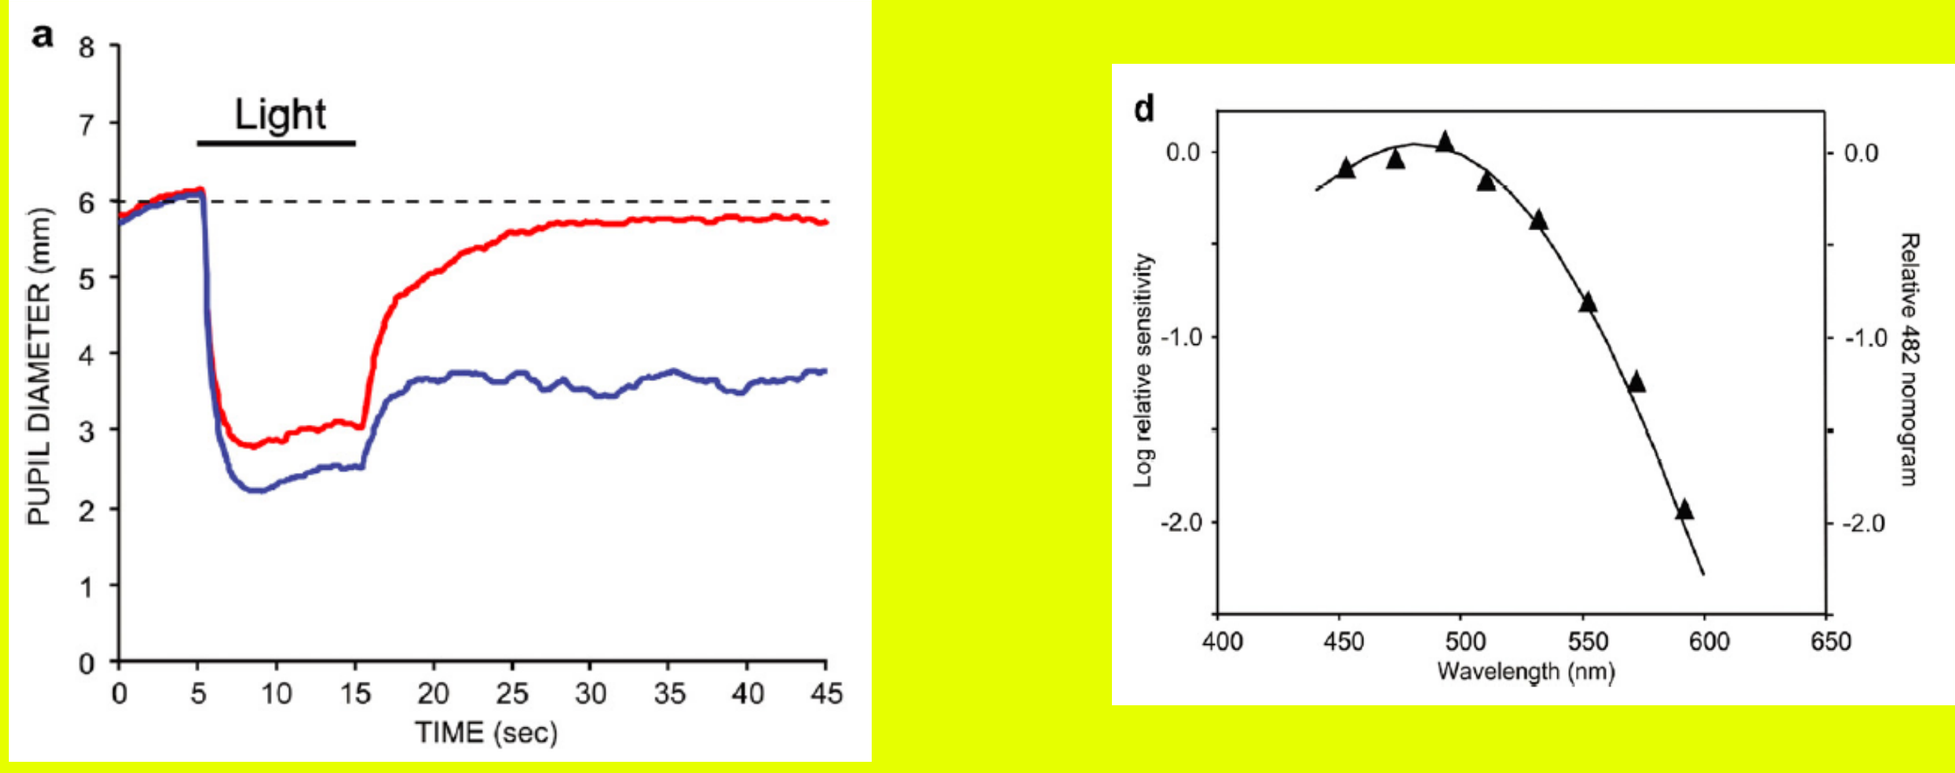

a
Light
PUPIL DIAMETER (mm)
TIME (sec)
d
Log relative sensitivity
Relative 482 nomogram
Wavelength (nm)

Figures from Gamlin *et al.* (2007)

ABSTRACT

Stark (1962) demonstrated vigorous pupil oscillations by illuminating the retina with a beam of light focussed to a small spot near the edge of the pupil. Small constrictions of the pupil then are sufficient to completely block the beam, amplifying the normal relationship between pupil area and retinal illuminance. In addition to this simple and elegant method, Stark also investigated more complex feedback systems using an electronic "clamping box" which provided arbitrary gain and phase delay between a measurement of pupil area and an electronically controlled light source. We have replicated Stark's results using a video-based pupillometer to control the luminance of a display monitor. Pupil oscillations were induced by imposing a linear relationship between pupil area and display luminance, with a variable delay. Slopes of the period-vs-delay function for 3 subjects are close to the predicted value of 2 (1.96-2.39), and the implied delays range from ~~254 to 376~~ 508 to 652 milliseconds. Our setup allows us to extend Stark's work by investigating a broader class of stimuli.

BIG QUESTIONS:

How can we use pupil measurements to assess operator state (fatigue, alertness)?

What are the effects of complex visual signals on the pupil?

Technical Challenges

State-related pupil changes are small, and may be swamped by other effects as well as random fluctuations. Challenges exist both in the measurement and interpretation of the signals. As can be seen in the upper right figure in the panel to the leftt, the transient pupil dilation resulting from mental effort has a peak amplitude of approximately half a millimeter.

Technical approach

In this work we seek to increase our understanding of the pupillary system using system identification methods first applied in this context by Stark in 1962. Stark studied the pupil by creating a situation in which the pupil system ran “open loop,” by focussing a beam of light down to a small spot at the pupil, so that changes in pupil size did not influence the amount of light falling on the retina (see lower left figures on panel to the left) . We accomplished a similar effect by using a real-time video pupillometer to control the intensity of a video display.

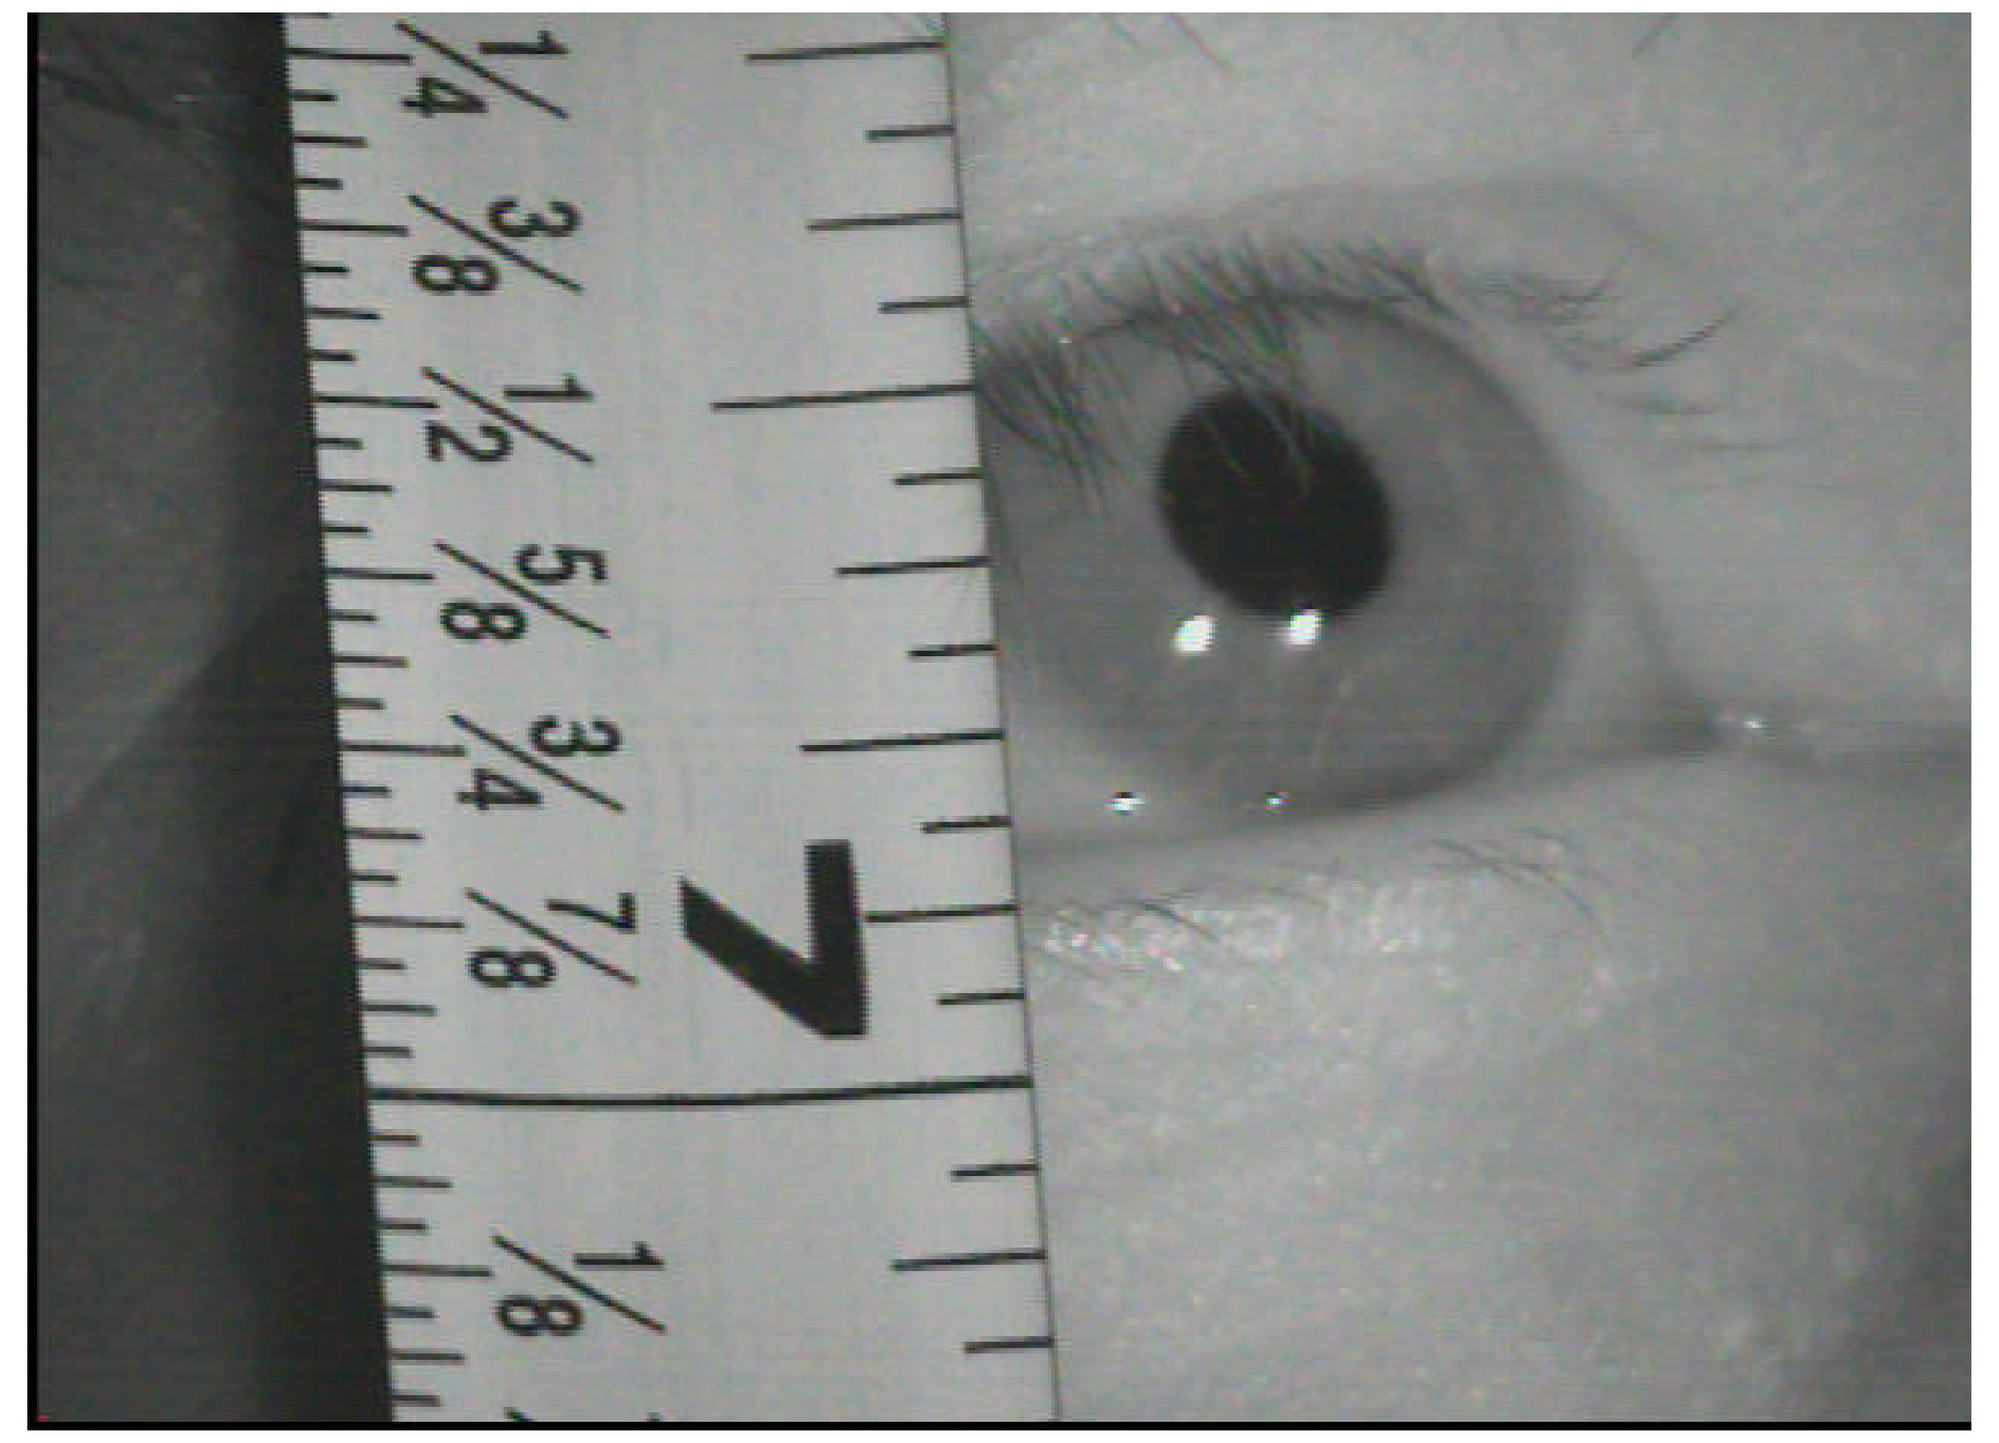

Typical video image used for pupillometry: The camera is a SONY EVI-D70 pan-tilt-zoom camera, located at about arm's length from the subject, set to maximum optical zoom and fitted with an infrared filter and a +1 diopter close-up lens. The illumination is provided by two groups of near-infrared LED's (seen as the two highlights reflected in the cornea).

Results: Oscillations produced by delayed feedback

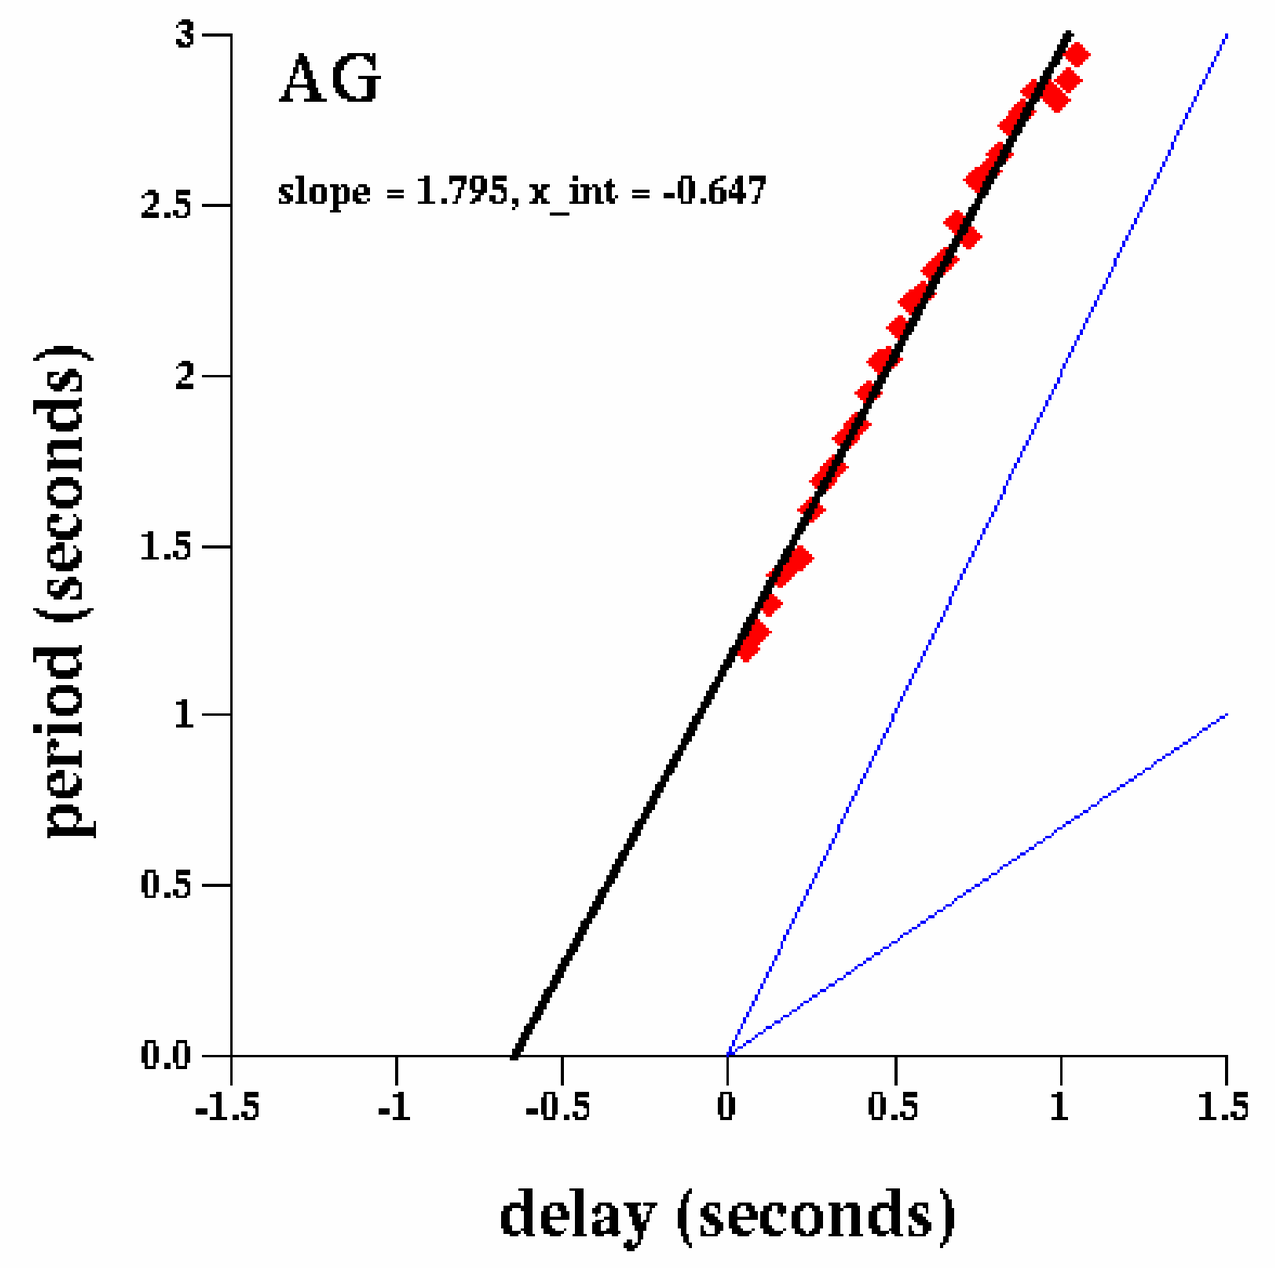

AG
slope = 1.795, x_int = -0.647
period (seconds)
delay (seconds)

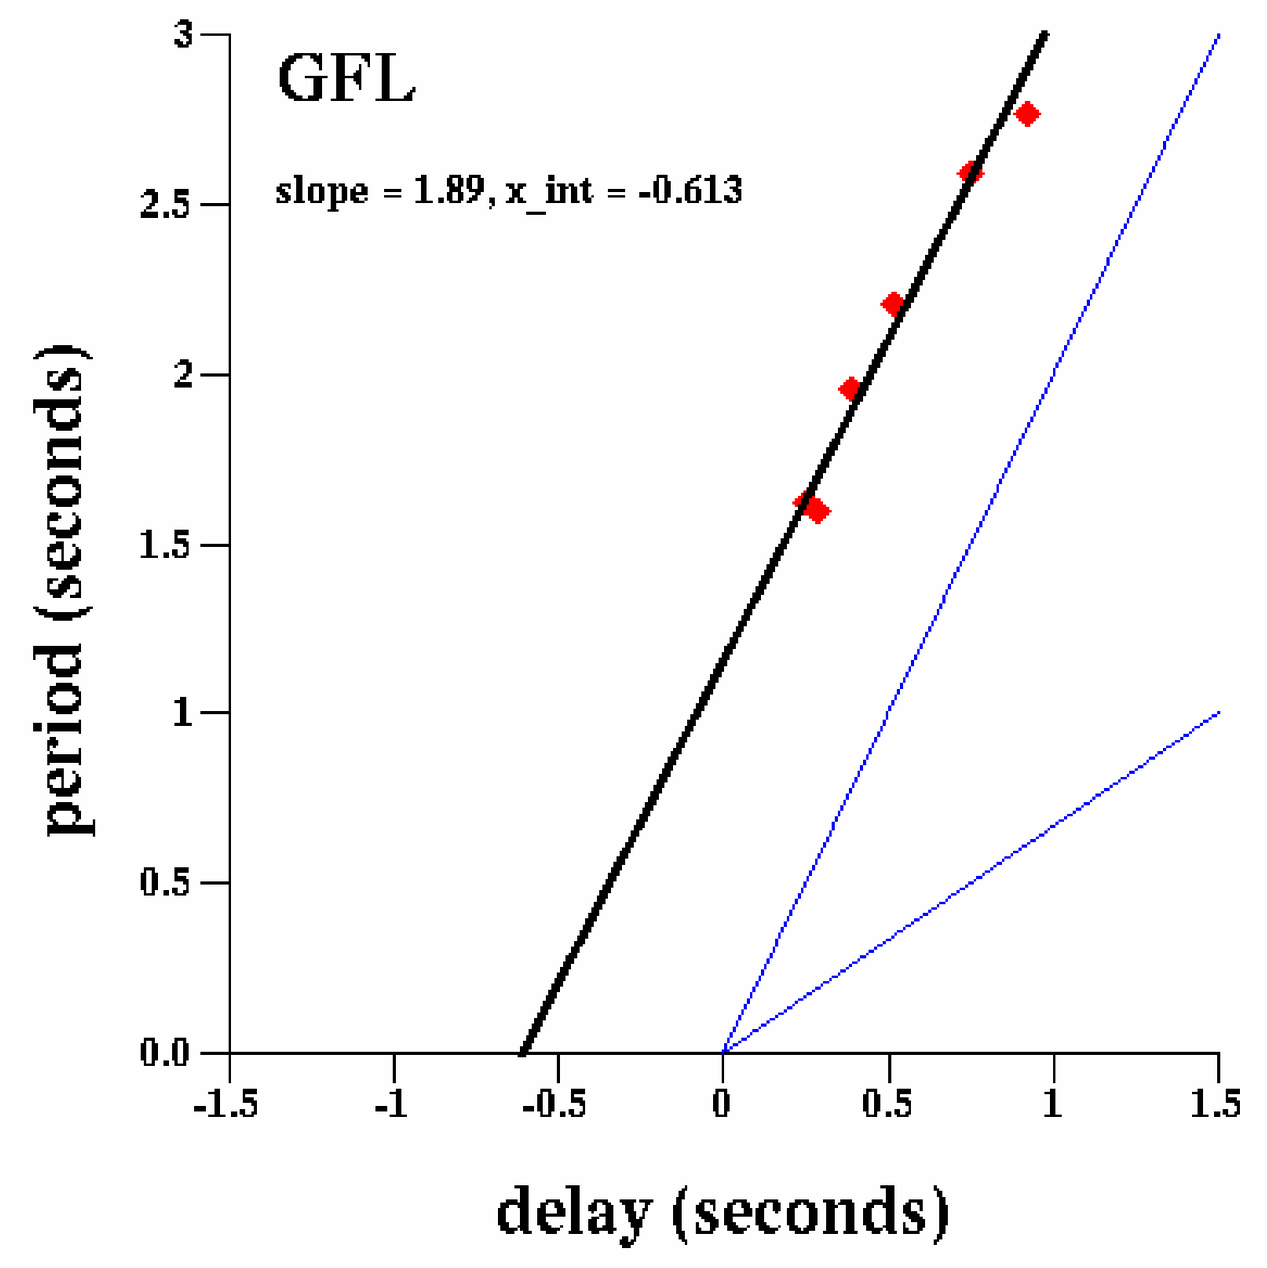

GFL
slope = 1.89, x_int = -0.613
period (seconds)
delay (seconds)

Pupil oscillations were induced by stimulating the eye with a light intensity proportional to pupil area, with a variable delay. A simple control law predicts period-vs-delay slopes of 2, as seen in data from two subjects (above). Subject JBM (below) show two branches; the lower branch (predicted slope=2/3) occurs at longer delays when an unstable pupil becomes entrained with an extra cycle of phase delay. The upper left panel shows a 4 DOF fit in which the two branches are fit independently, while in the upper right panel the fits were constrained to share a common x-intercept. The lower two panels show 1 DOF fits in which the slopes were constrained to the theoretical predictions, and only the x-intercept was free to vary. The two lower panels show two replications, with a 5% discrepancy in their estimates of the internal delay.

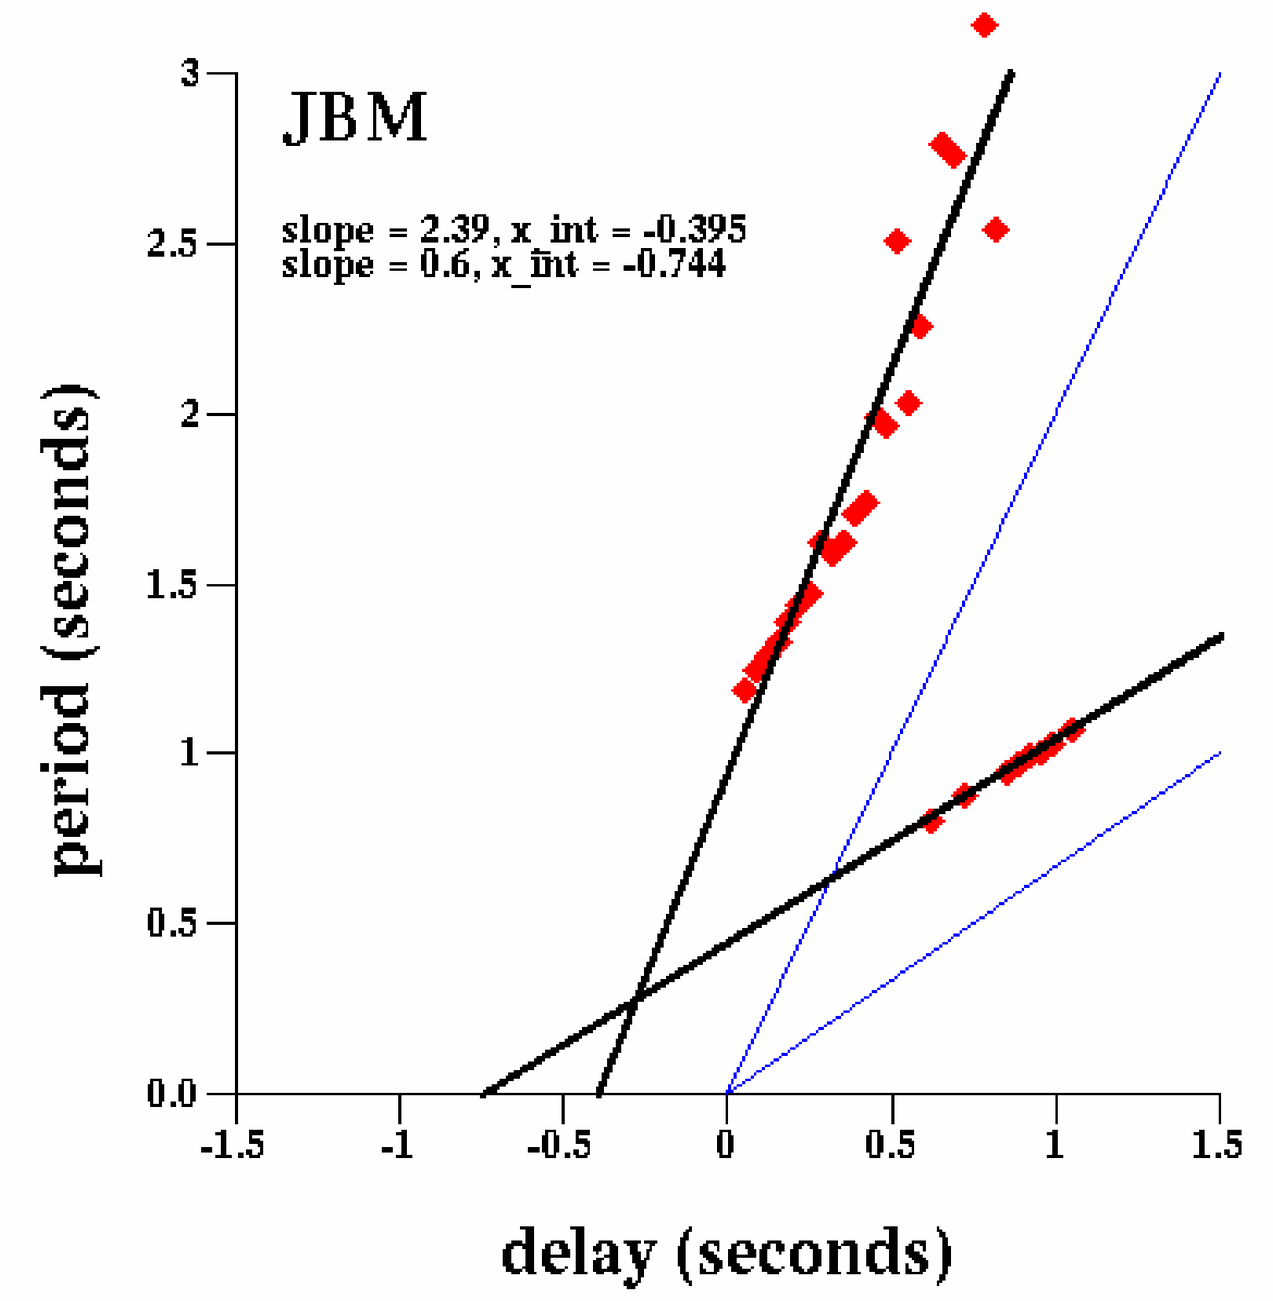

JBM
slope = 2.39, x_int = -0.395
slope = 0.6, x_int = -0.744
period (seconds)
delay (seconds)

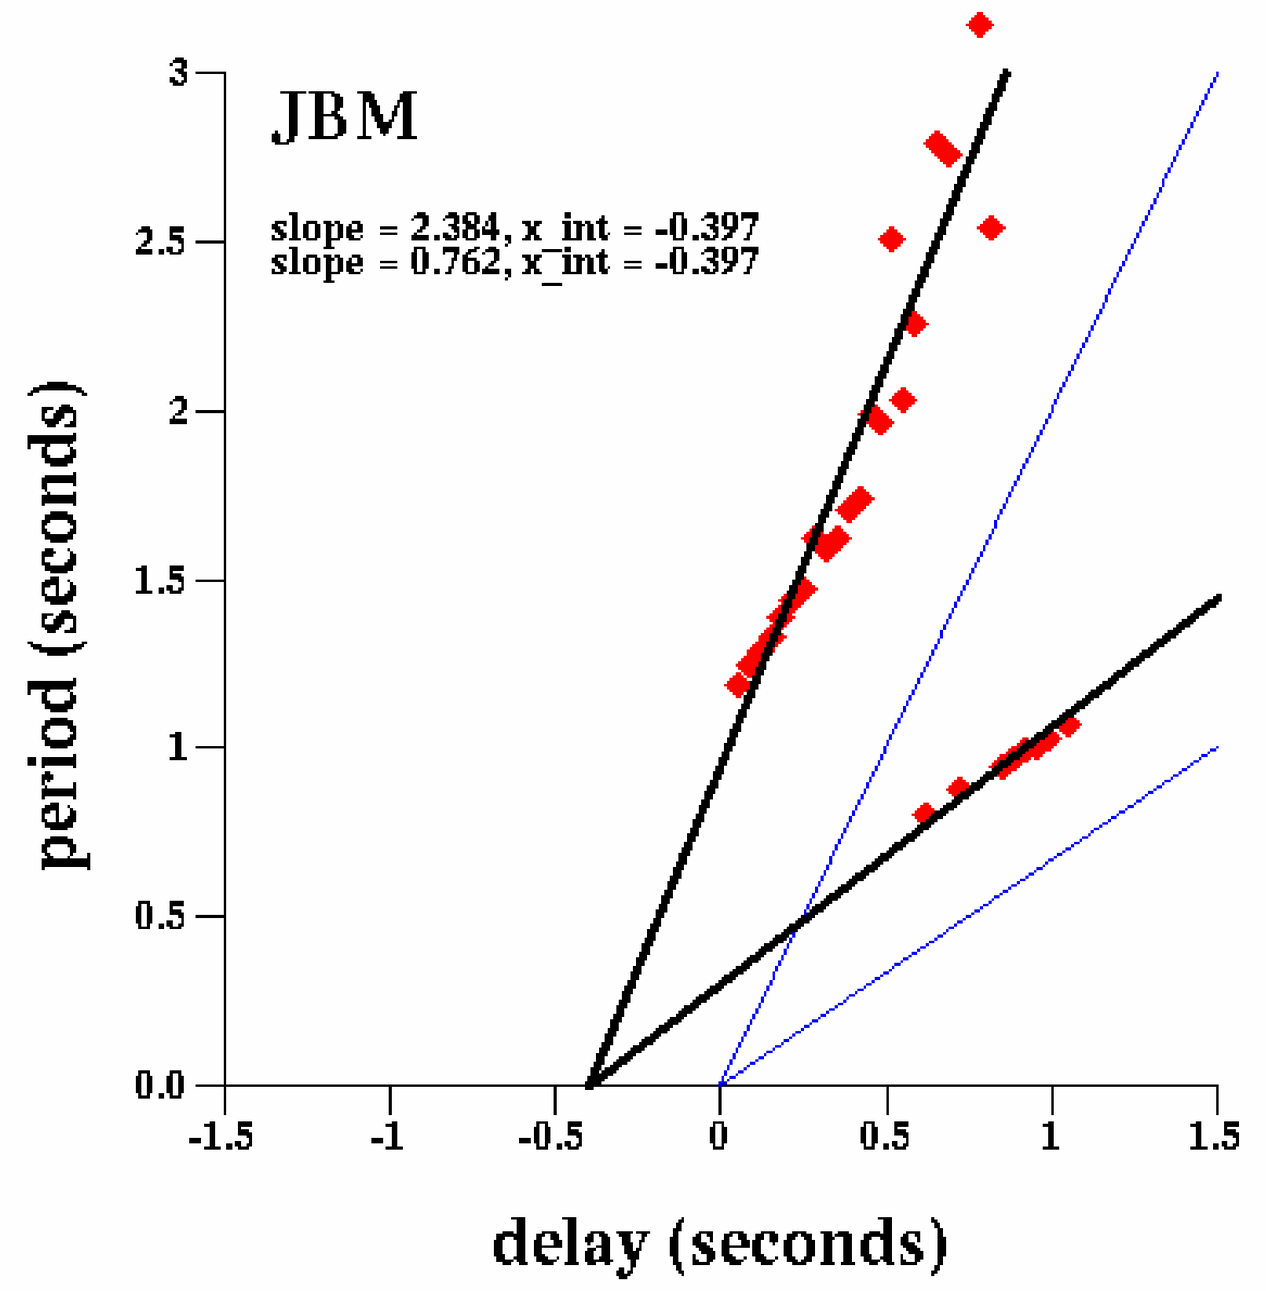

JBM
slope = 2.384, x_int = -0.397
slope = 0.762, x_int = -0.397
period (seconds)
delay (seconds)

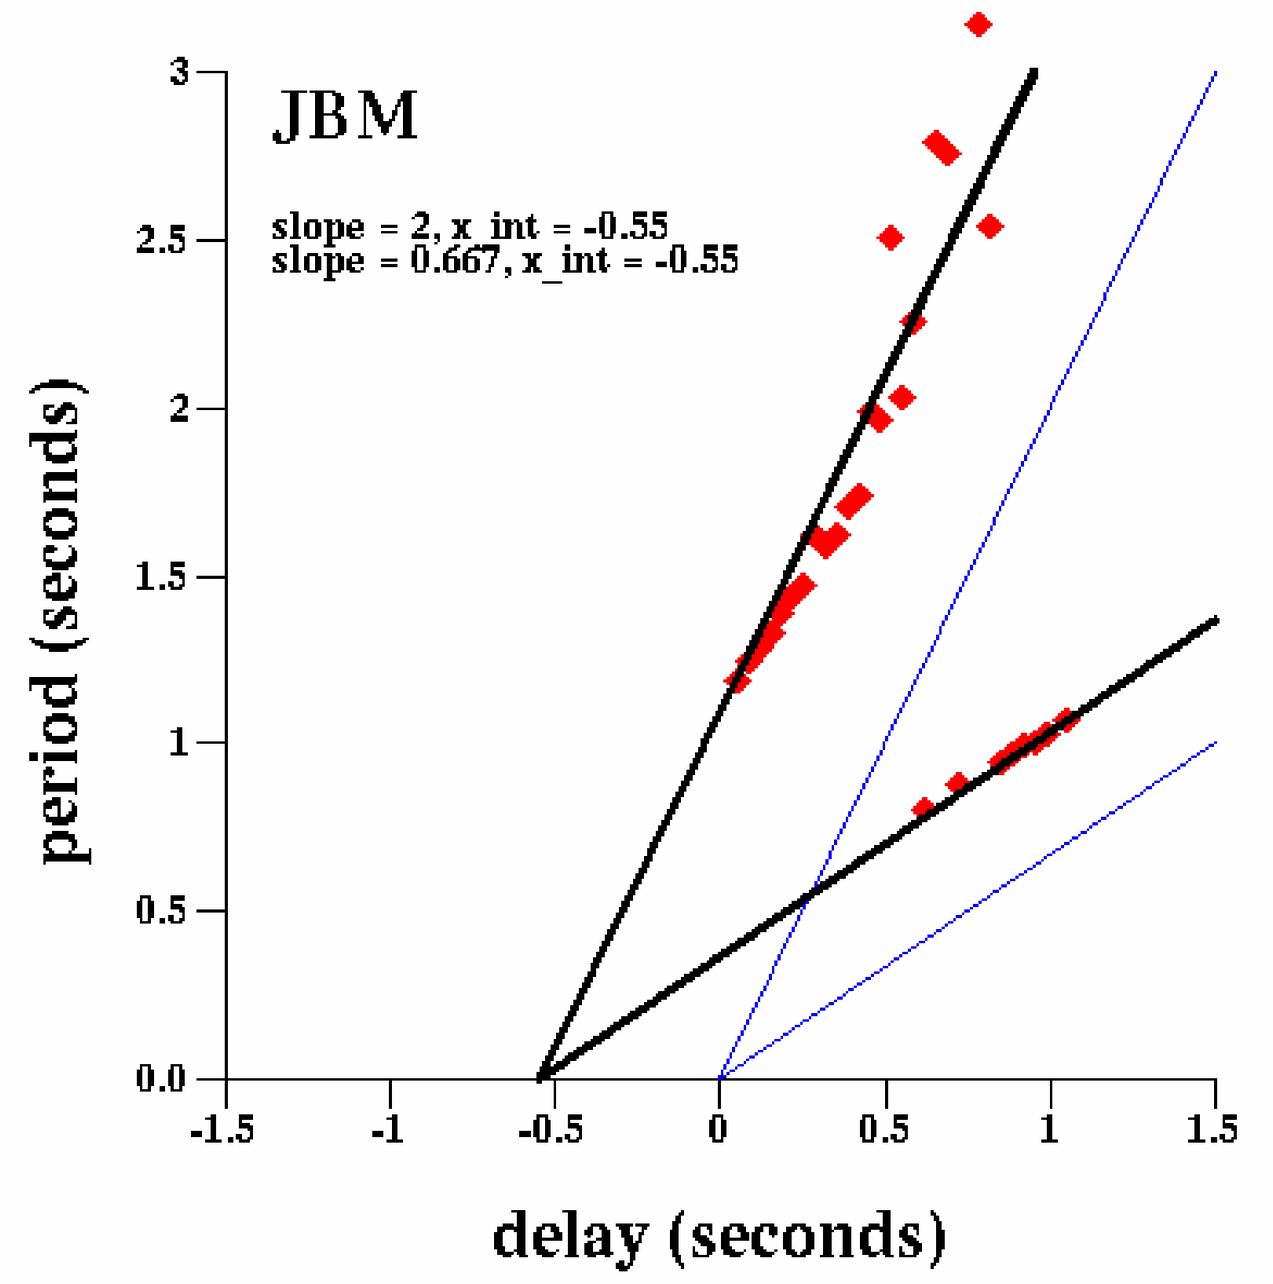

JBM
slope = 2, x_int = -0.55
slope = 0.667, x_int = -0.55
period (seconds)
delay (seconds)

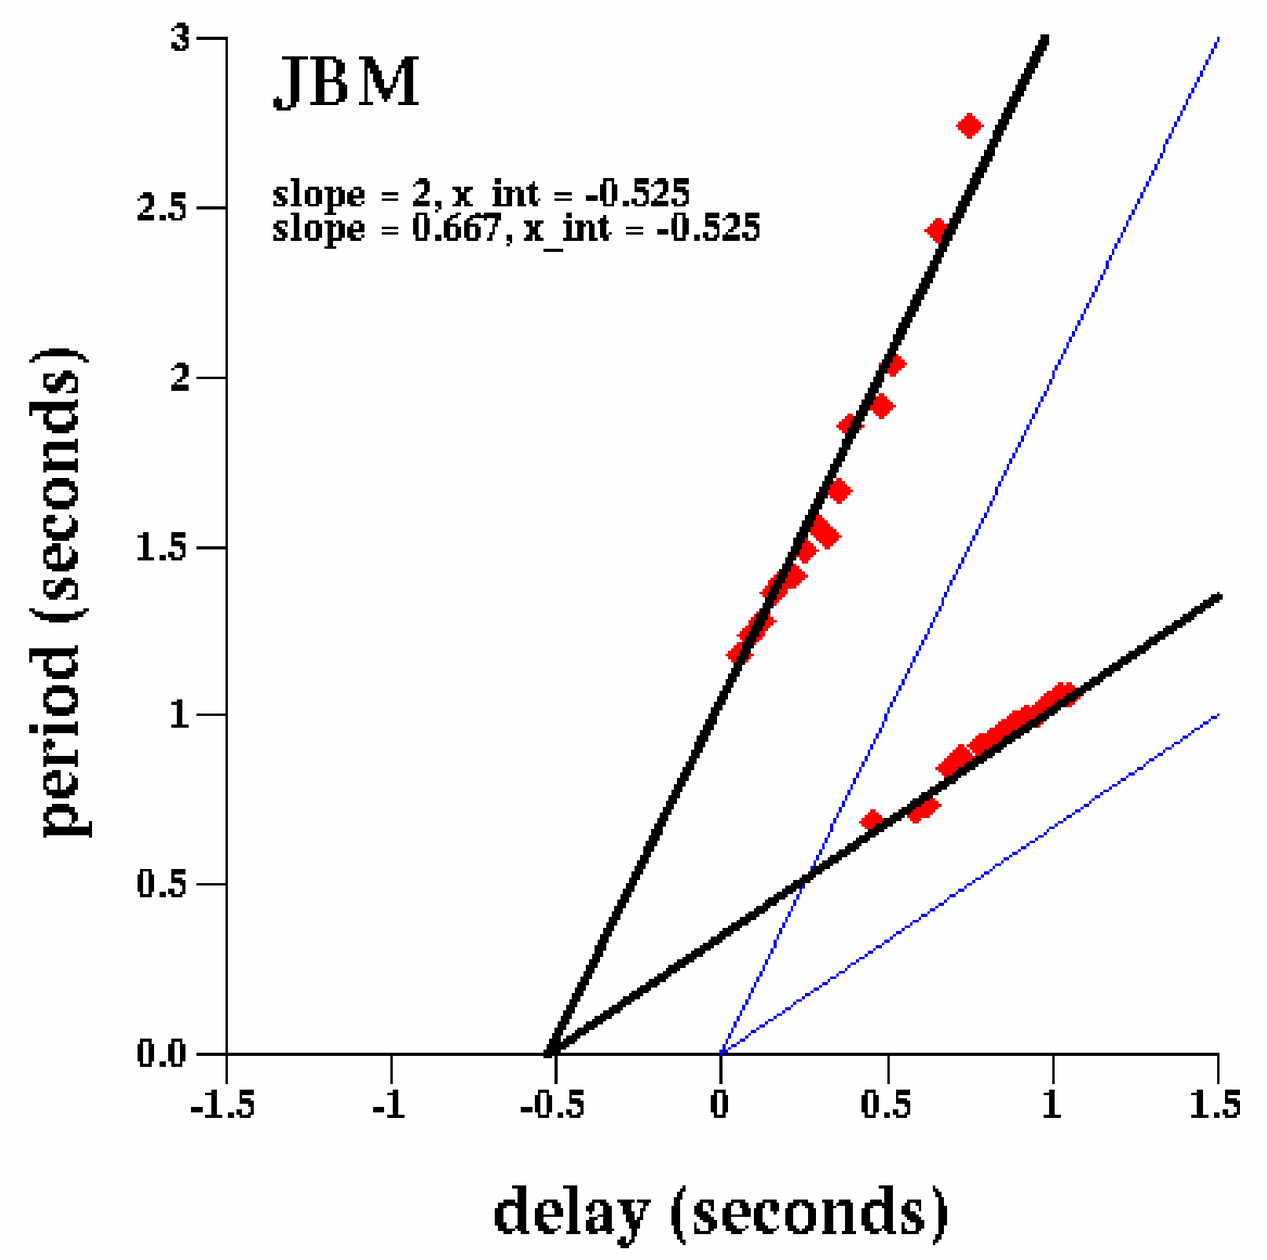

JBM
slope = 2, x_int = -0.525
slope = 0.667, x_int = -0.525
period (seconds)
delay (seconds)

More results: white noise analysis

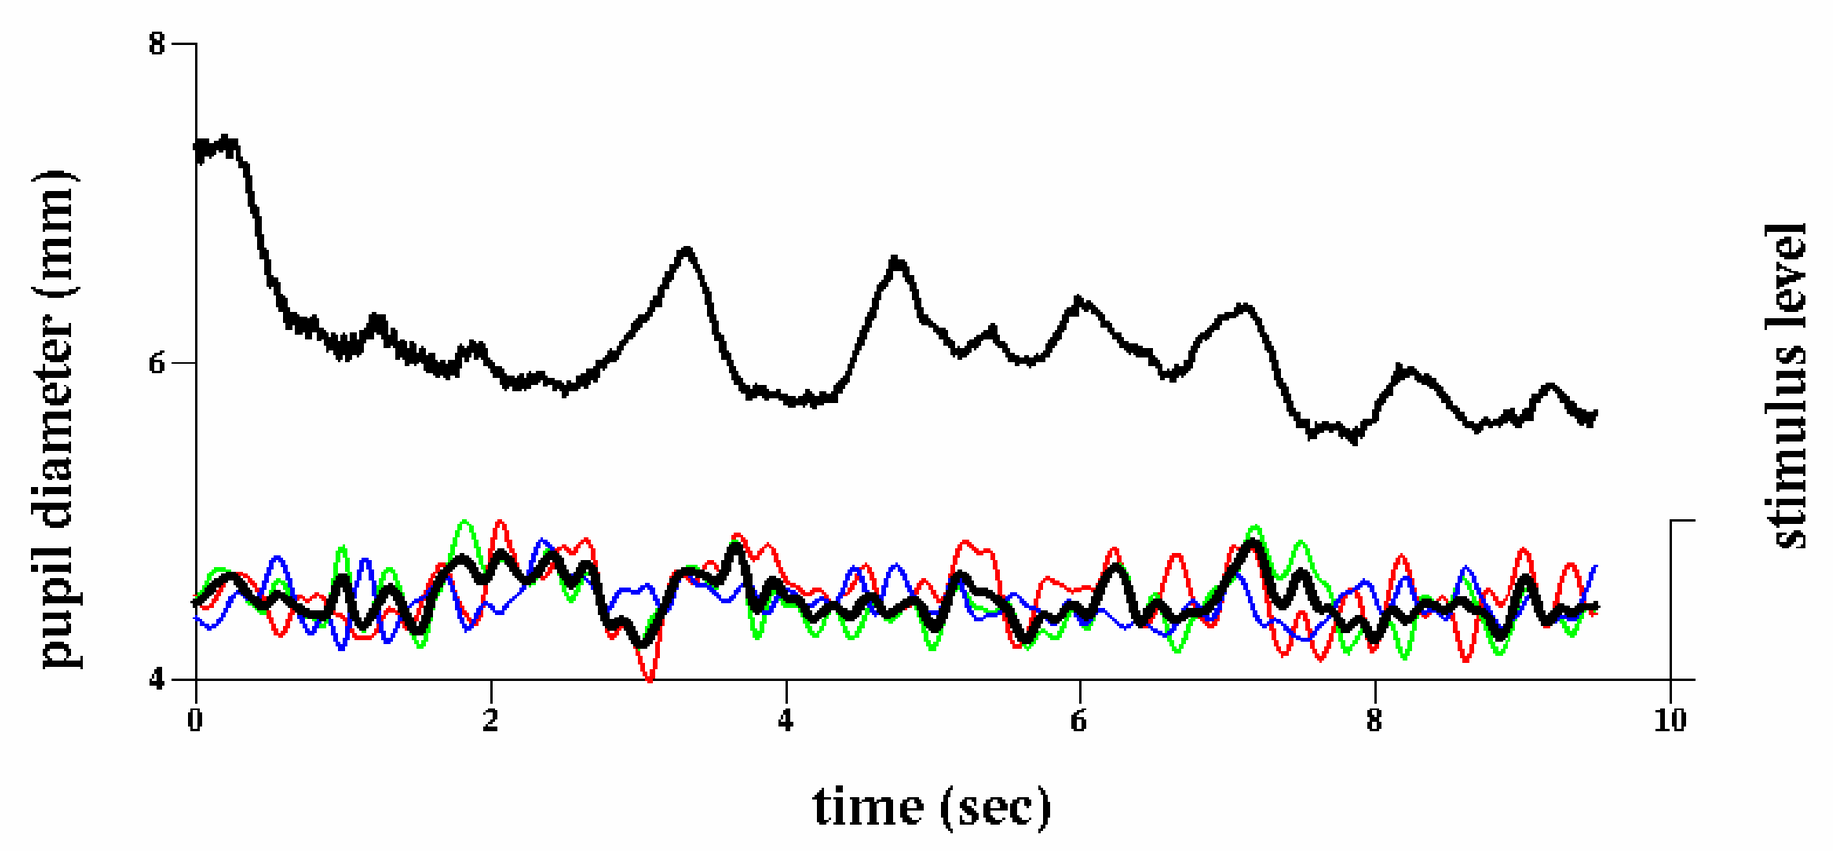

pupil diameter (mm)
stimulus level
time (sec)

In addition to replicating Stark's results using open-loop stimulation and delayed feedback, we are extending the use of white-noise analysis to the case of full-color stimuli, in the hope of identifying a component due to ipRGC's with a different spectral and temporal signature. The figure above shows the pupil response (upper black trace) to independent random signals applied to the red, blue and green components of a video screen (luminance shown in black). This is a pilot experiment intended to demonstrate proof-of-concept; in order to produce a stimulus capable of independently stimulating the three cone photoreceptors, rods and ipRGC's, a stimulator with at least 5 primaries will be required, perhaps by combining two video displays with different primaries using a beam-splitter.

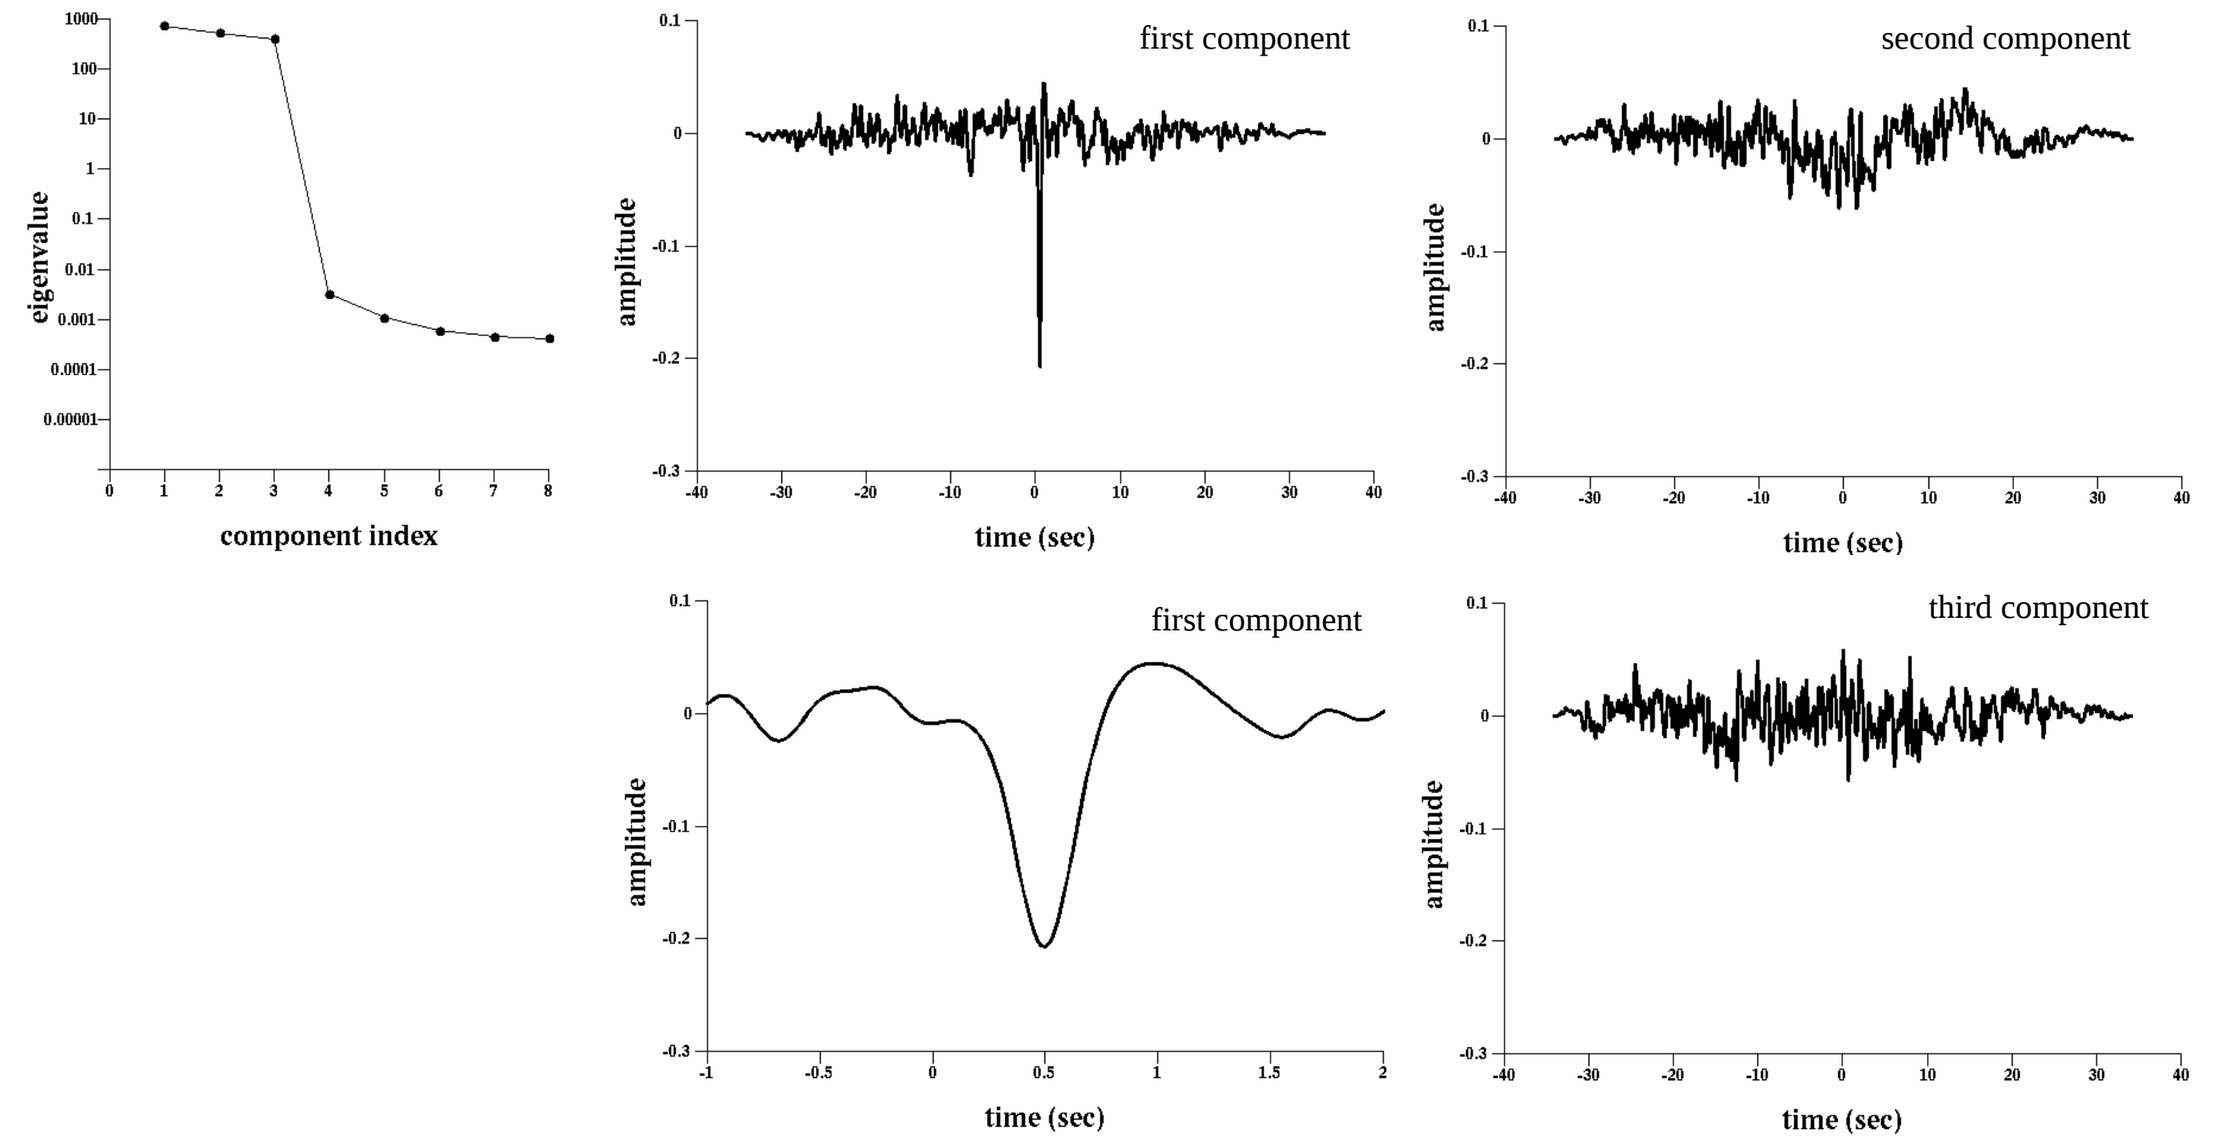

eigenvalue
component index
first component
second component
first component
third component
amplitude
time (sec)

The data were analyzed by cross-correlating the response with the projection of the stimulus in a variety of color directions. Principle Components Analysis (PCA) was then applied to the tableau of resulting kernels. Because the inherent dimensionality of the stimulus space is 3, only the first 3 components have eigenvalues that are larger than the noise floor. These are shown above; the phosphor loadings on the first component correspond to the luminance direction, while the second two components appear to soak up the rest of the noise without displaying any clear causality. The latency of the peak of the first component is in good agreement with the estimate obtained from the period-vs-delay functions.

Conclusion

Disappointingly, we failed to observe pupil oscillations when using delayed feedback to control grating contrast (rather than luminance), although the efforts to date fall somewhat short of heroic. Similarly, to date the extended white noise analysis has not revealed mechanisms beyond the basic luminance input to the pupil system. Given the extremely long sustained response seen in ipRGC's, this method may not be the most efficient.

References

Barbur, JL and Stockman, A (2010) Photopic, Mesopic, and Scotopic Vision and Changes in Visual Performance. In: Dartt, D and Besharse, J and Dana, R, (eds.) *Encyclopedia of the Eye.* (323 - 331). Academic Press: Oxford.

Kahneman, D., Peavler, W. S., and Onuska, L. (1968) Effects of verbalization and incentive on the pupil response to mental activity. *Canadian Journal of Psychology*, **22**:186-196.

Gamlin, P. D. R., McDougal, D. H., Pokorny, J., Smith, V. C., Yau, K., and Dacey, D. (2007) Human and macaque pupil responses driven by melanopsin-containing retinal ganglion cells. *Vision Research* **47**:946-954.

Stark, L. (1962) Environmental Clamping of Biological Systems: Pupil Servomechanism. *Journal of the Optical Society of America*, **52**:925-929.

Supported by the System-wide Safety Assurance Technologies (SSAT) project of NASA's Aviation Safety Program.