A comparison of the dynamics of visually-controlled head and hand movements

Jeffrey B. Mulligan
NASA Ames Research Center

Scott B. Stevenson
Univ. Houston College of Optometry

BACKGROUND AND HISTORY

The eye movement correlogram

When a subject attempts to track a randomly-moving target, high frequency components of the resulting eye movements do not at a first glance appear related to the stimulus waveform. Cross-correlation and signal averaging, however, reveal reliable responses in a method we call the eye movement correlogram.

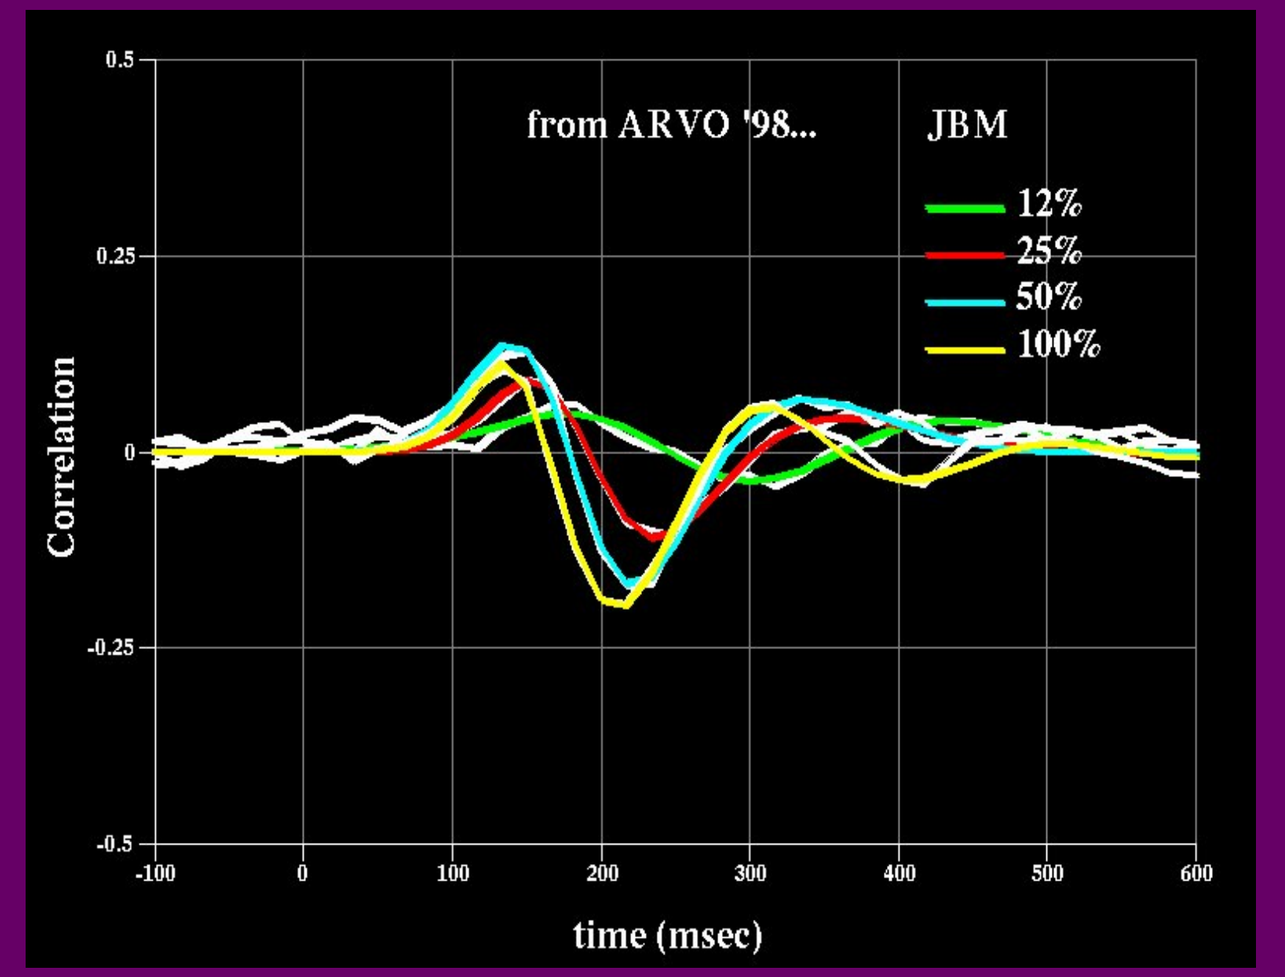
from ARVO '98...
JBM
12%
25%
50%
100%
Correlation
time (msec)

The delayed feedback paradigm

In the delayed feedback paradigm, the visual consequences of an eye movement are artificially delayed by transiently stabilizing the stimulus. Under various simple control laws, oscillations are expected with a linear relation between period and delay, the slope of which depends on the particular control law. In 2007, we implemented delayed visual feedback using a DPI eye-tracker equipped with optical stimulus deflectors. In contrast to monkey data showing period-vs-delay slopes between 2 and 4 (Goldreich, Krauzlis and Lisberger, 1992) , we observed slopes close to 1.5.

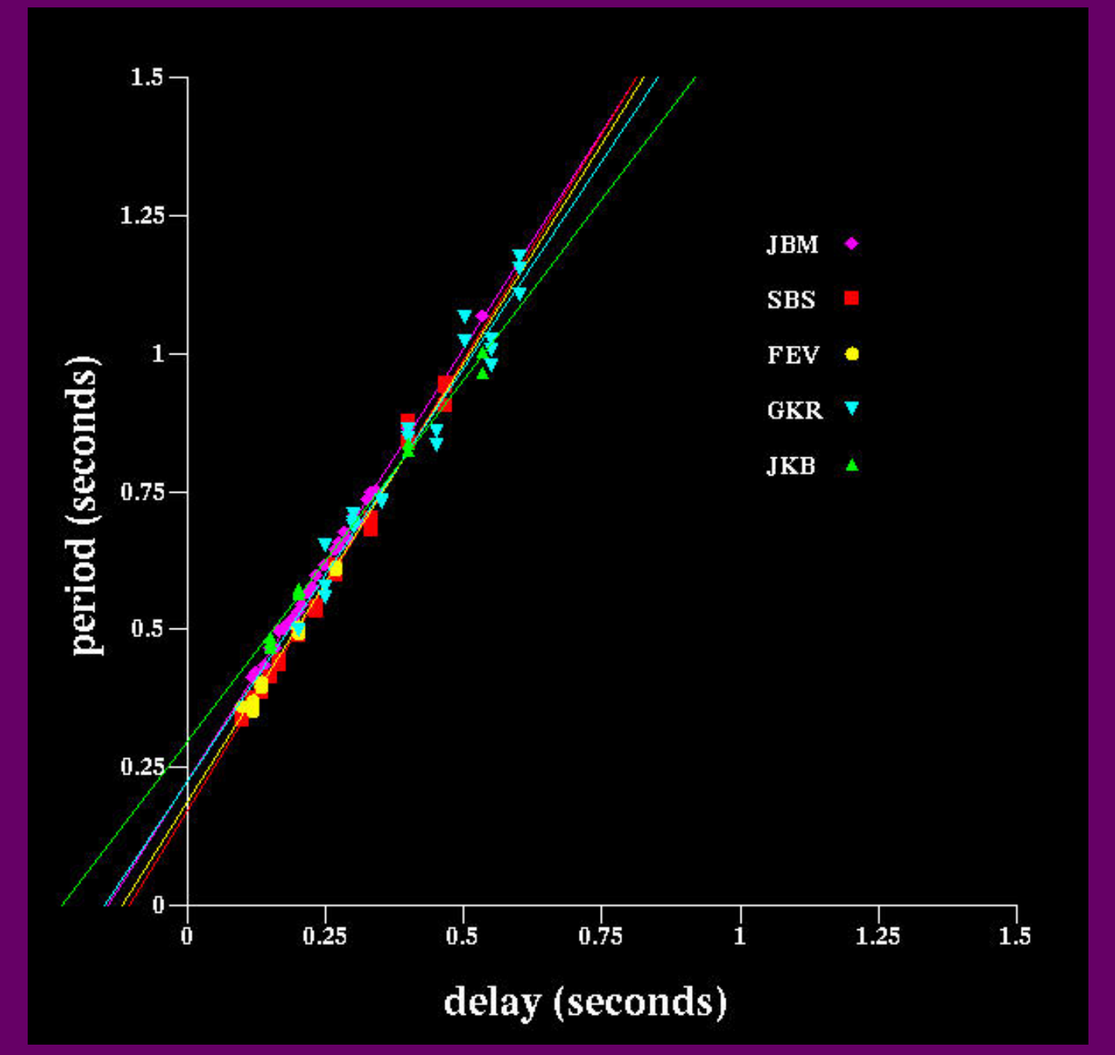
JBM
SBS
FEV
GKR
JKB
period (seconds)
delay (seconds)

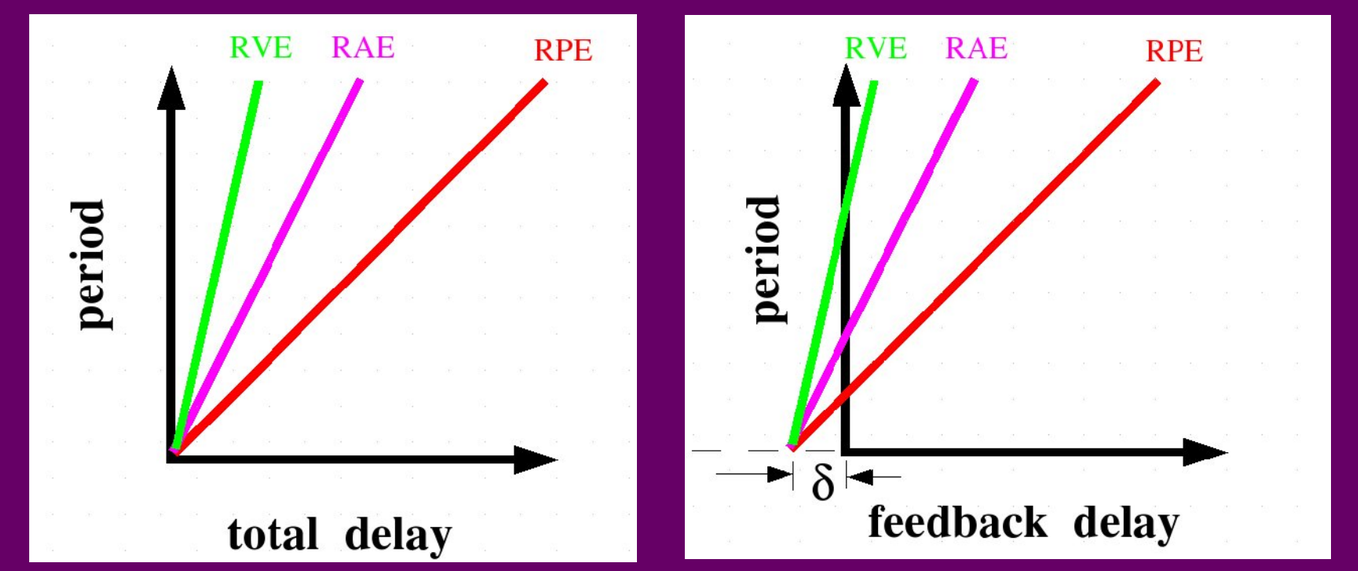
RVE
RAE
RPE
period
total delay
RVE
RAE
RPE
period
δ
feedback delay

The hand movement correlogram

Bruggemann and Stevenson extended the correlogram technique to manual tracking using a joystick in their 2007 OSA Fall Vision Meeting poster. The hand is significantly slowerthan the eye, but the latencies of hand and eye movements are equally affected by manipulations of the visual stimulus.

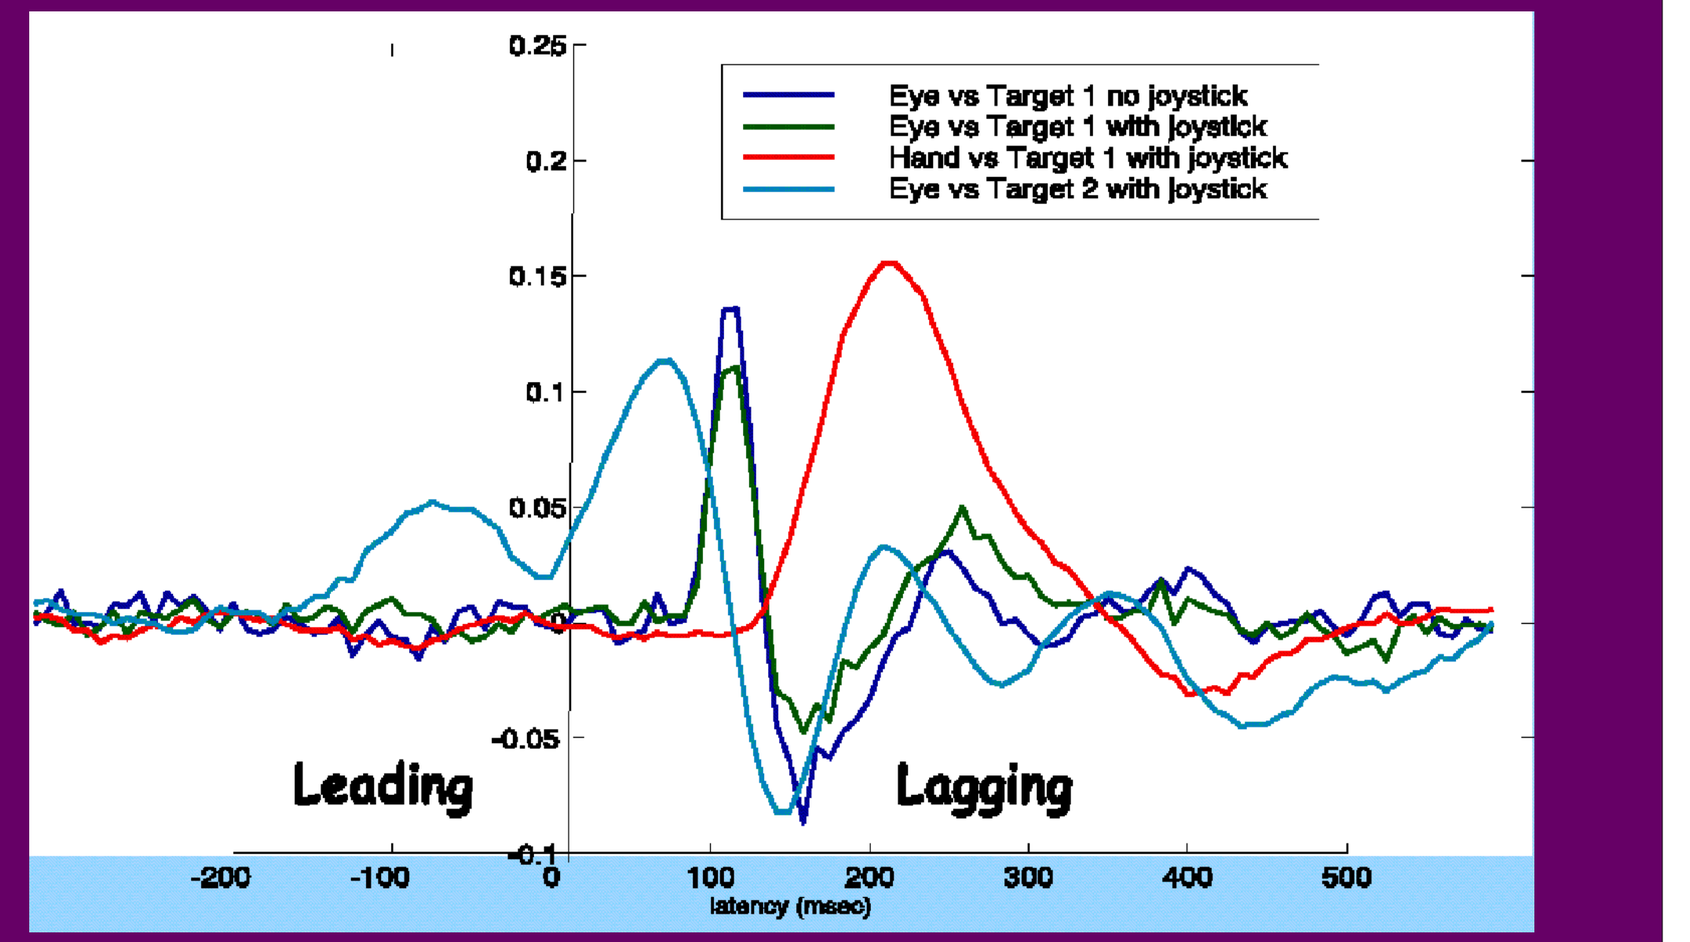
Eye vs Target 1 no joystick
Eye vs Target 1 with joystick
Hand vs Target 1 with joystick
Eye vs Target 2 with joystick
Leading
Lagging
latency (msec)

ABSTRACT

Brueggemann and Stevenson compared tracking of a randomly moving target by eye gaze and hand, using a joystick-controlled cursor (2007 OSA Fall Vision Meeting). In this study, we extended their work by comparing motor control of a tracking cursor using either the hand or the head. A Polhemus Fastrak 6-DOF space tracker was used to provide inputs; in the case of the hand, position was used to control the cursor, with up and right in space naturally mapping to up and right on the screen. In the case of the head, angular measures of pitch and yaw were used, as if a virtual laser pointer were attached to the subject's nose. The primary difference between these two cases is that, in the case of head rotations, the vestibulo-ocular reflex (VOR) causes compensatory eye movements to be made, stabilizing the scene on the retina in spite of the movement of the head. Under normal circumstances, the VOR effectively compensates for head movements, the world appears stable, and similar results are obtained for head and hand correlograms. Observed latencies are in the neighborhood of 400 milliseconds, with the head around 50 milliseconds ~~faster~~ slower than the hand. The method is expected to reveal more striking differences, however, under abnormal gravitational conditions such as those encountered during aircraft maneuvers or space flight, where lack of visual stability is often observed. We have examined adaptation of the VOR (using modified visual feedback) as a possible analog of these conditions. We have also measured period-versus-delay (PVD) functions of oscillations induced by delayed visual feedback. We have previously reported PVD slopes near 1.6 for eye movements, suggesting a control system using both position and velocity inputs. PVD functions obtained for head tracking show steeper slopes, suggesting a different weighting of visual signals is used for control.

BIG QUESTION (not answered today): Can changes in the Vestibulo-Ocular Reflex (VOR) be revealed by a head-pointing task?

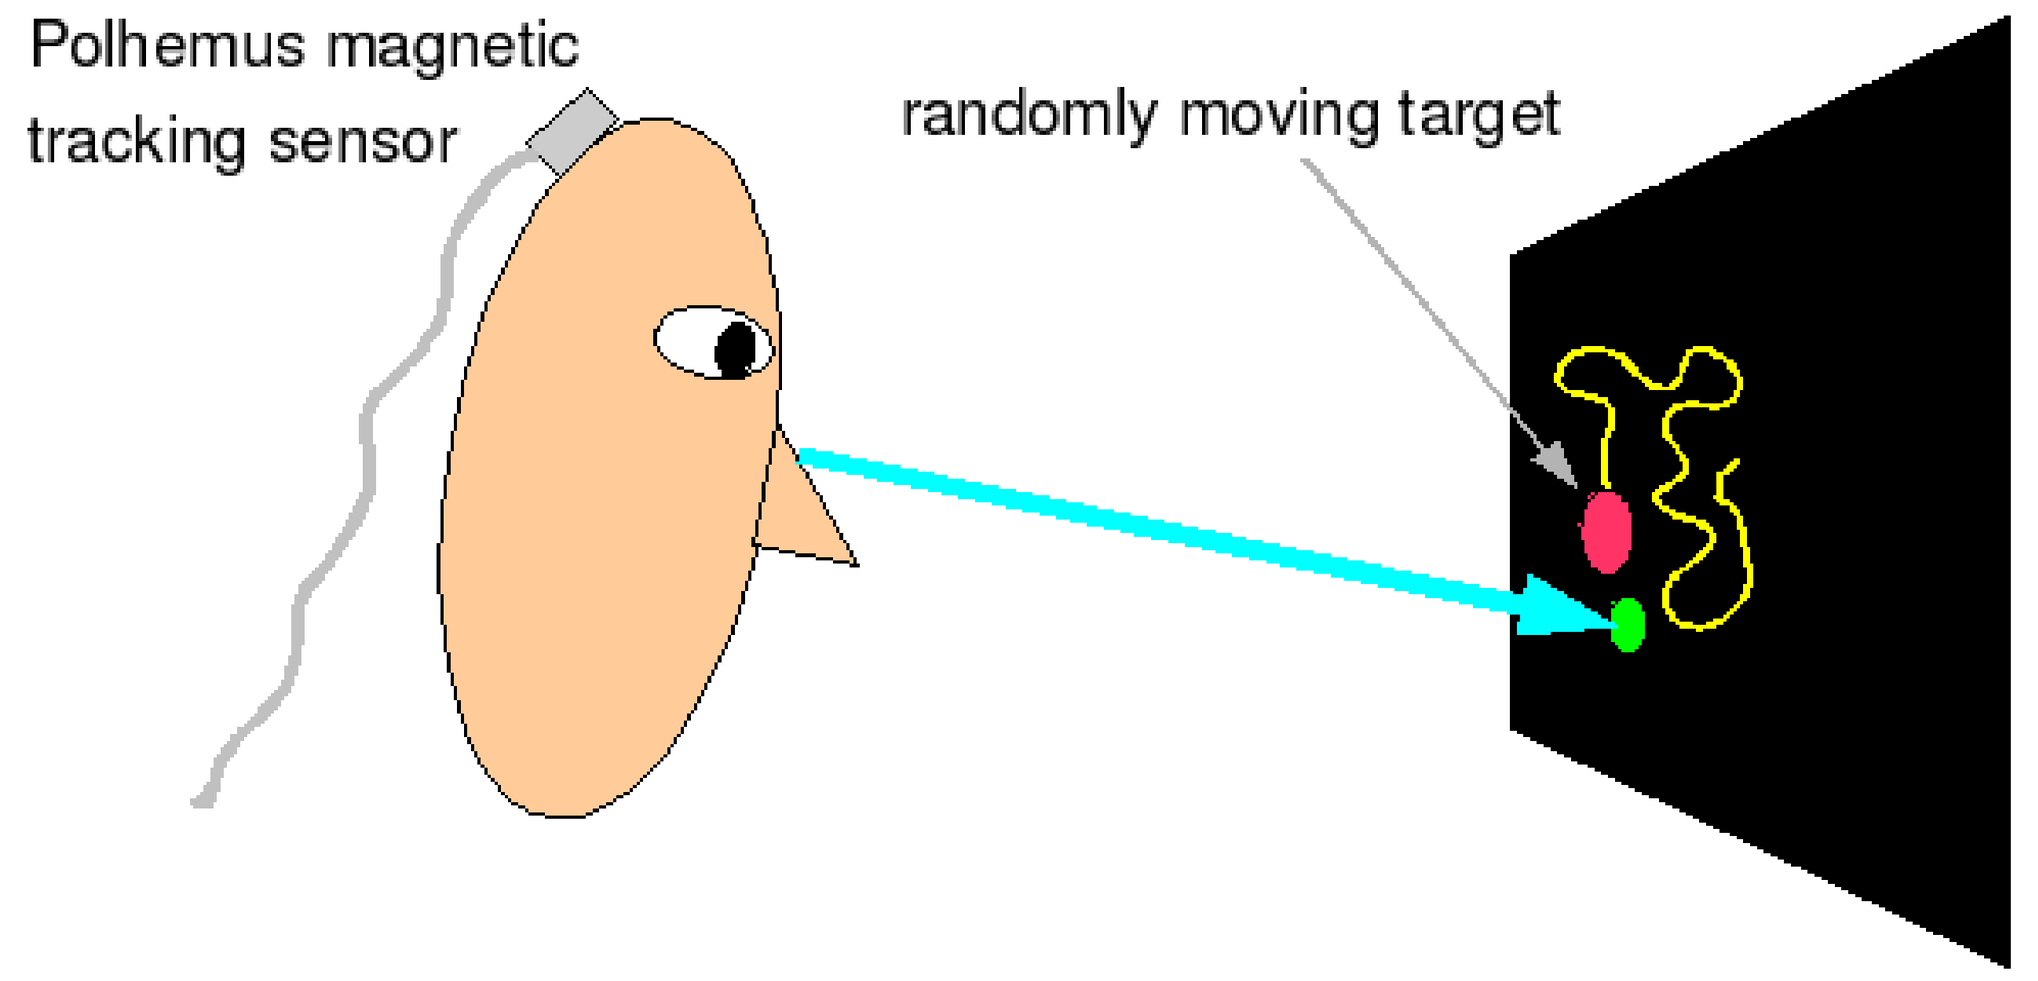
Polhemus magnetic
tracking sensor
randomly moving target

head orientation controls position of tracking cursor

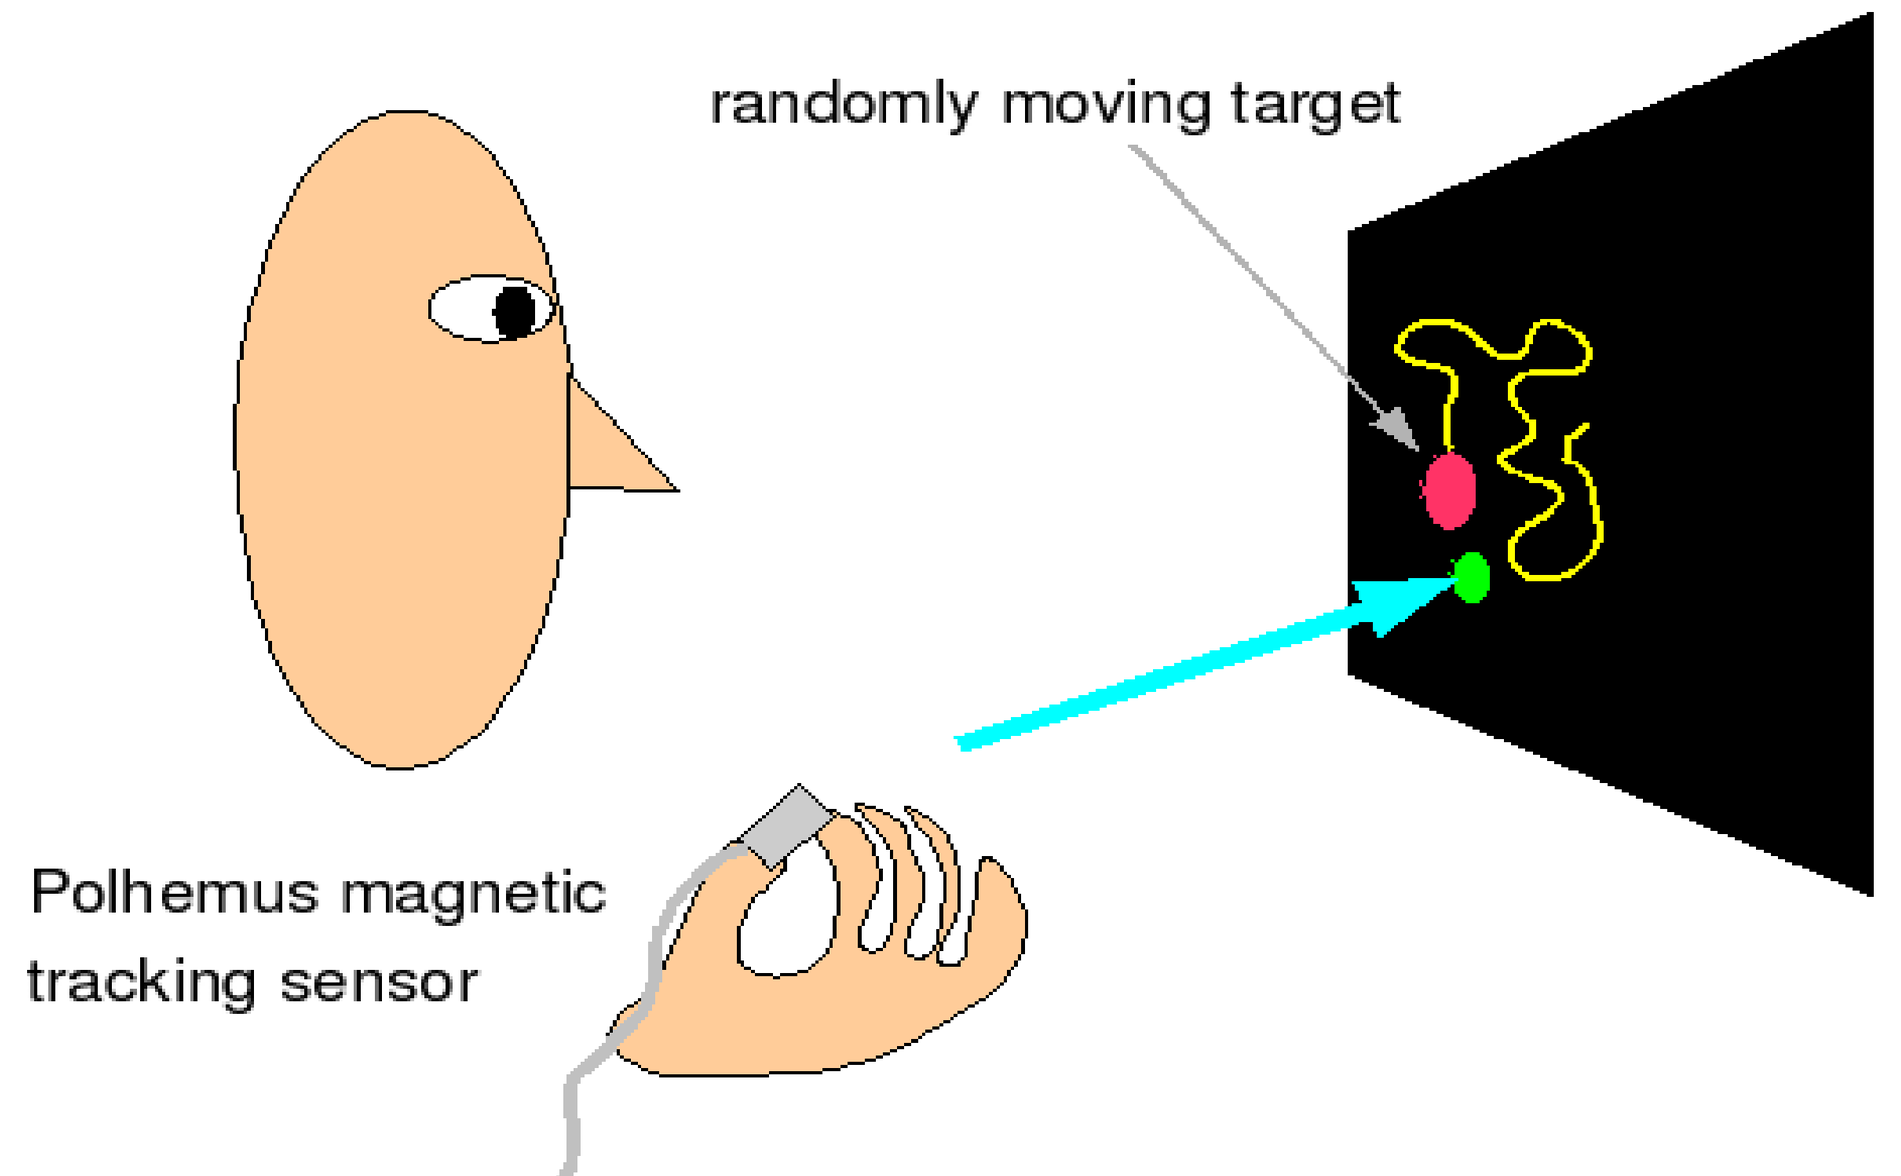
randomly moving target
Polhemus magnetic
tracking sensor

hand position controls position of tracking cursor

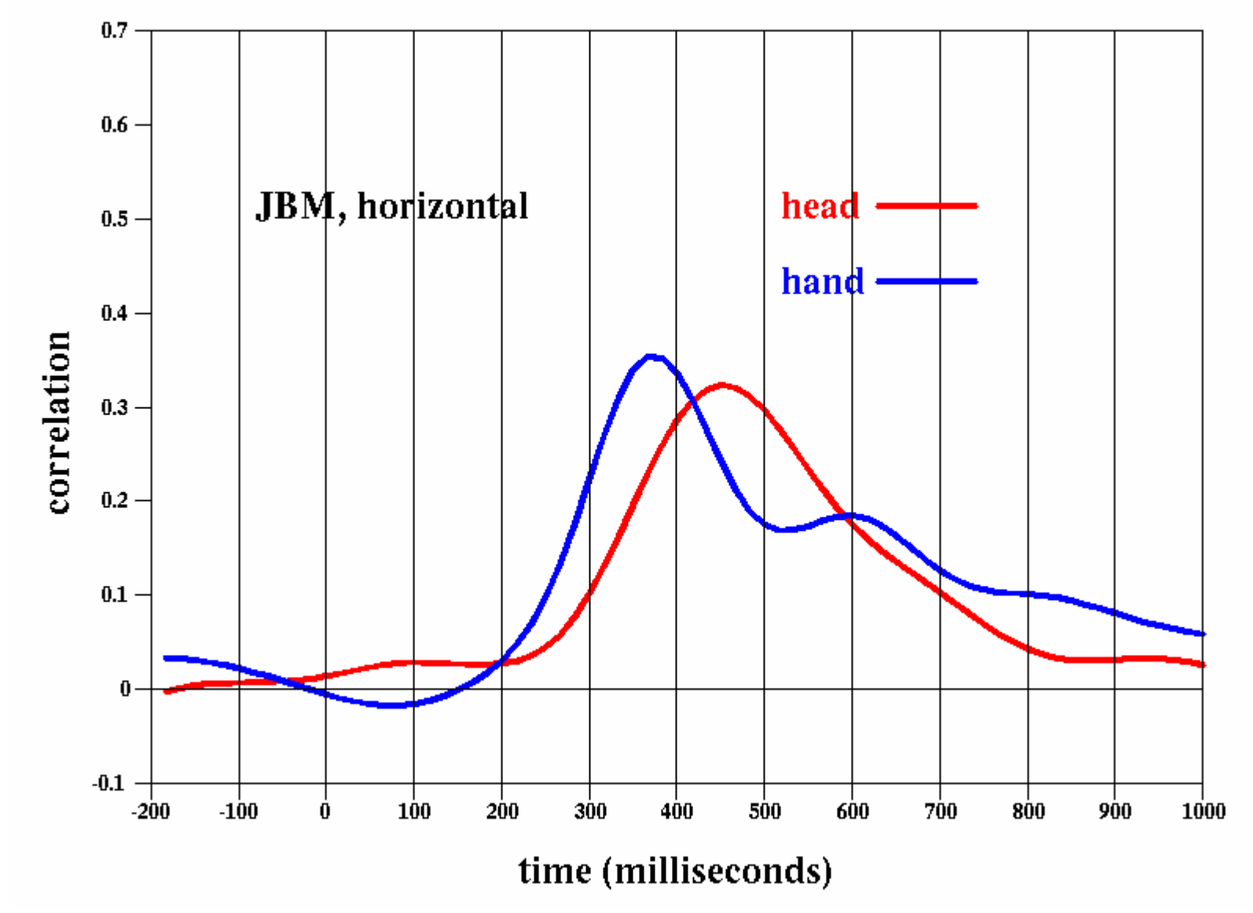
JBM, horizontal
head
hand
correlation
time (milliseconds)

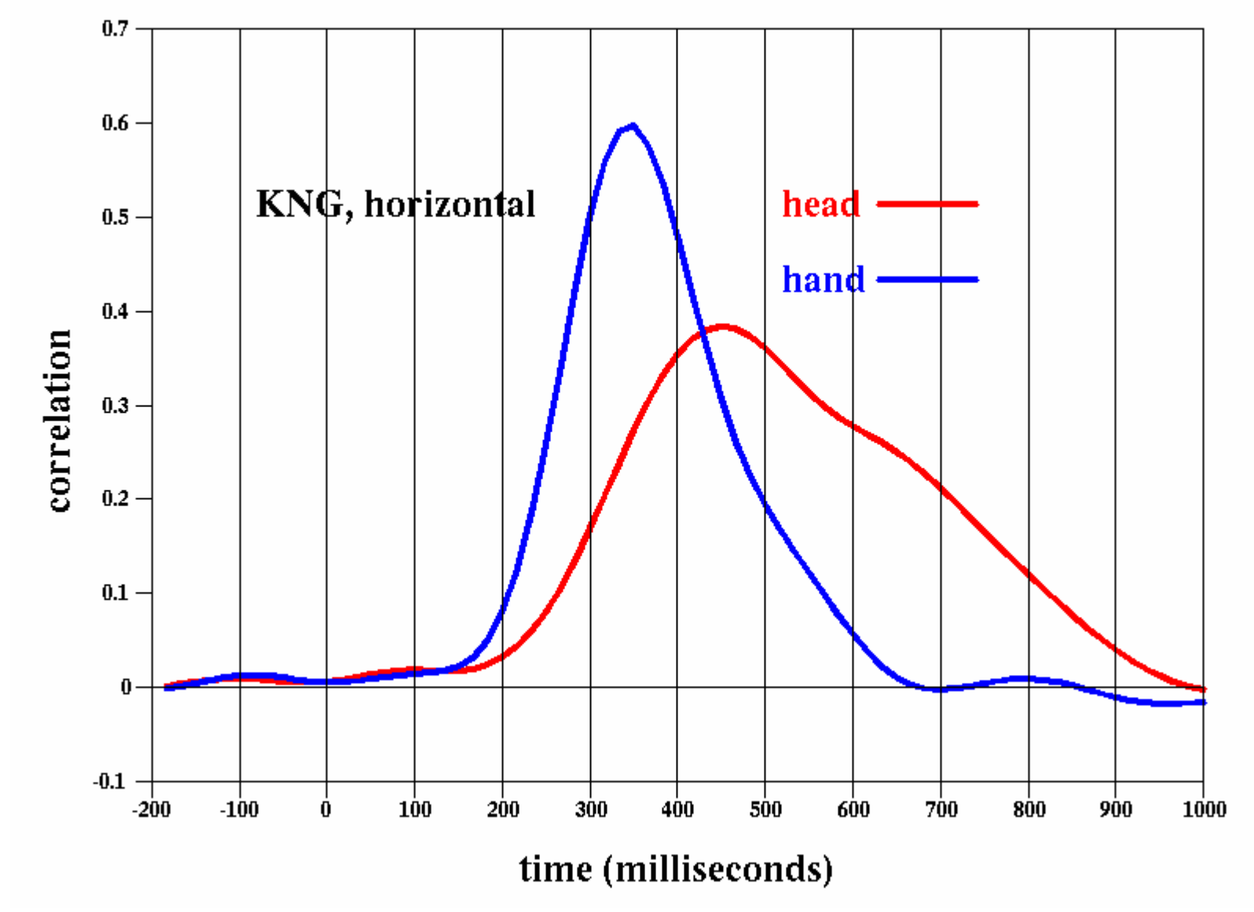
KNG, horizontal
head
hand
correlation
time (milliseconds)

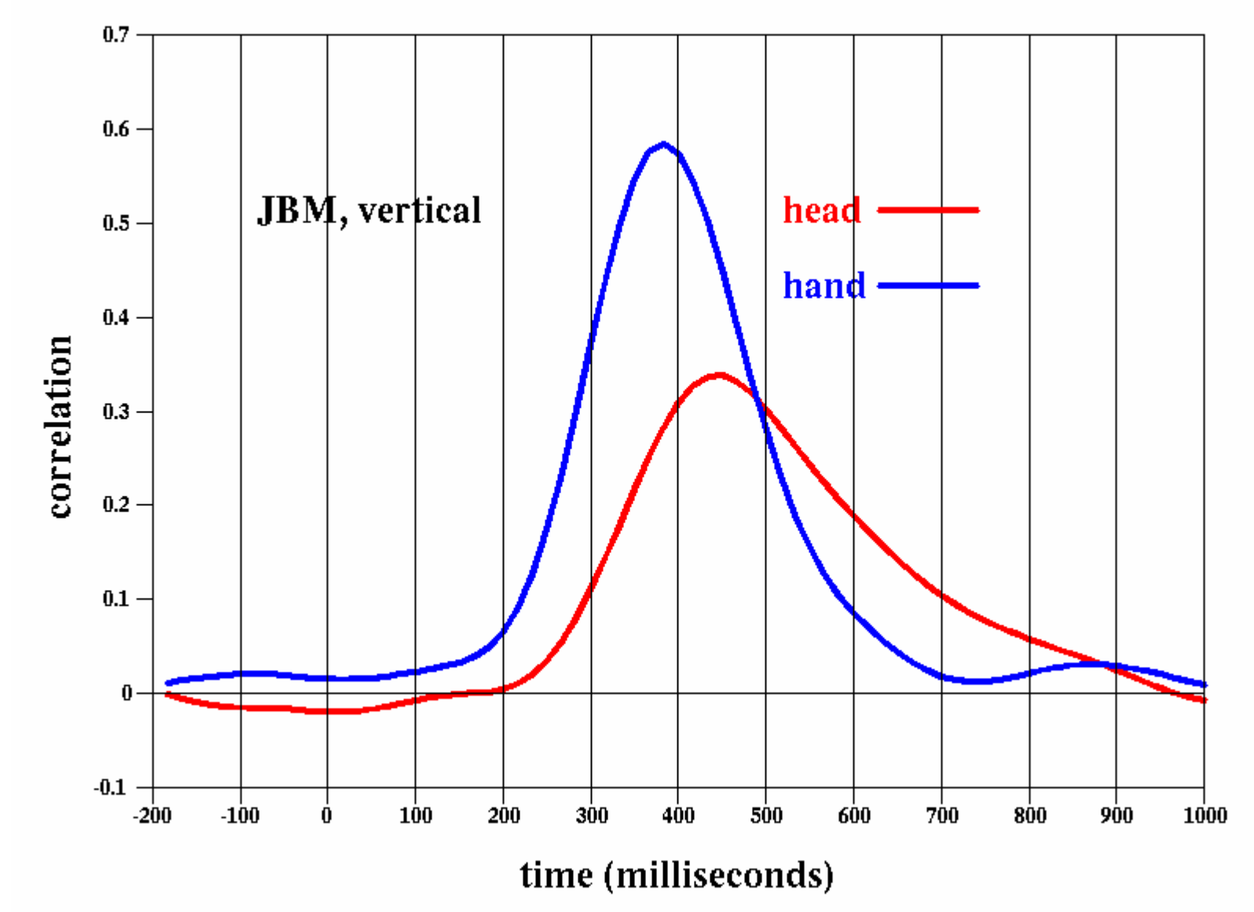
JBM, vertical
head
hand
correlation
time (milliseconds)

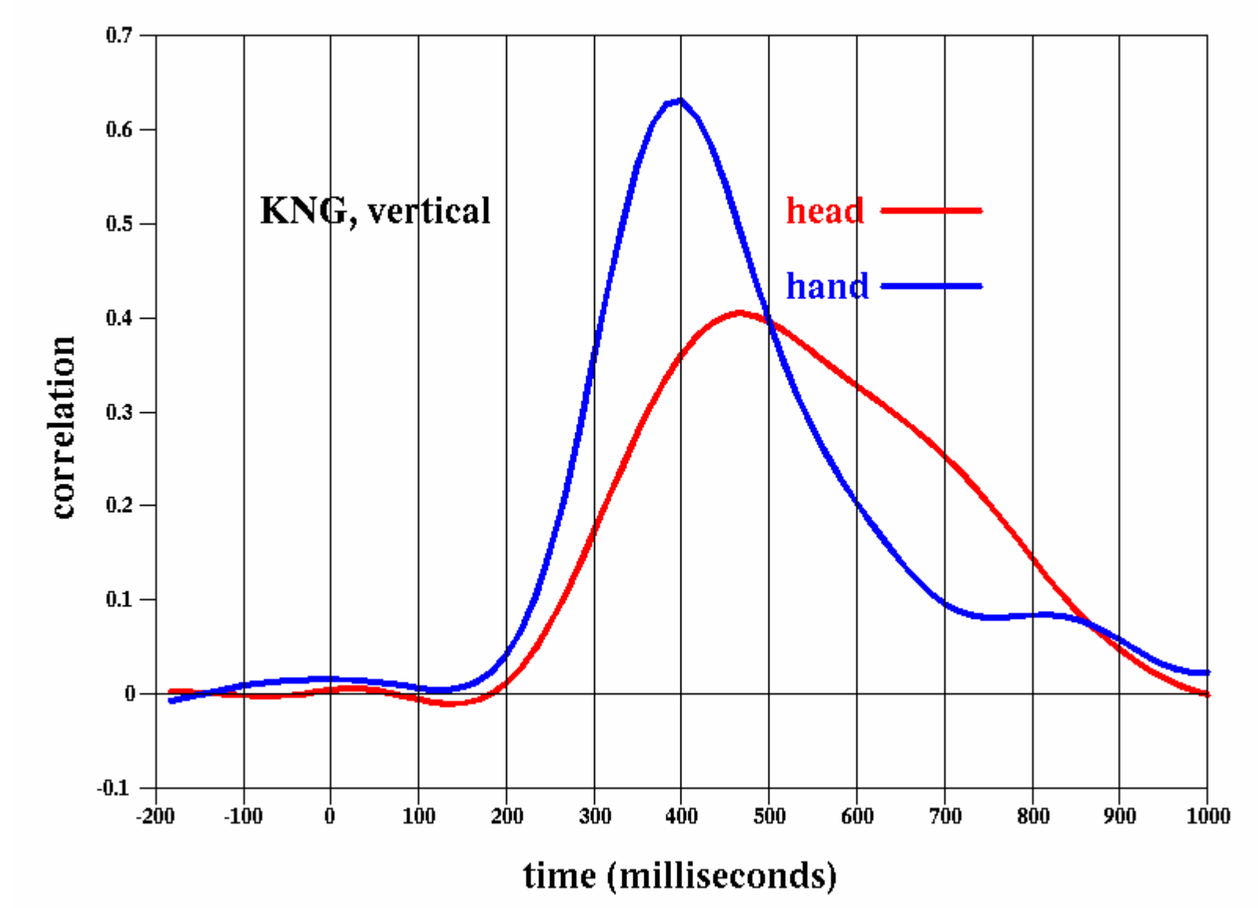
KNG, vertical
head
hand
correlation
time (milliseconds)

Simulation of VOR gain changes

The action of the VOR is to maintain steady gaze in the presence of head movements. The VOR gain can be adapted by the introduction of head-slaved target motion of the visual field. Here we simulate VOR adaptation by feeding the cursor movement back into the target motion (with no delay). While this does not adapt the VOR (due to the presence of unmodified peripheral stimuli), it does produce subtle changes in the correlograms.

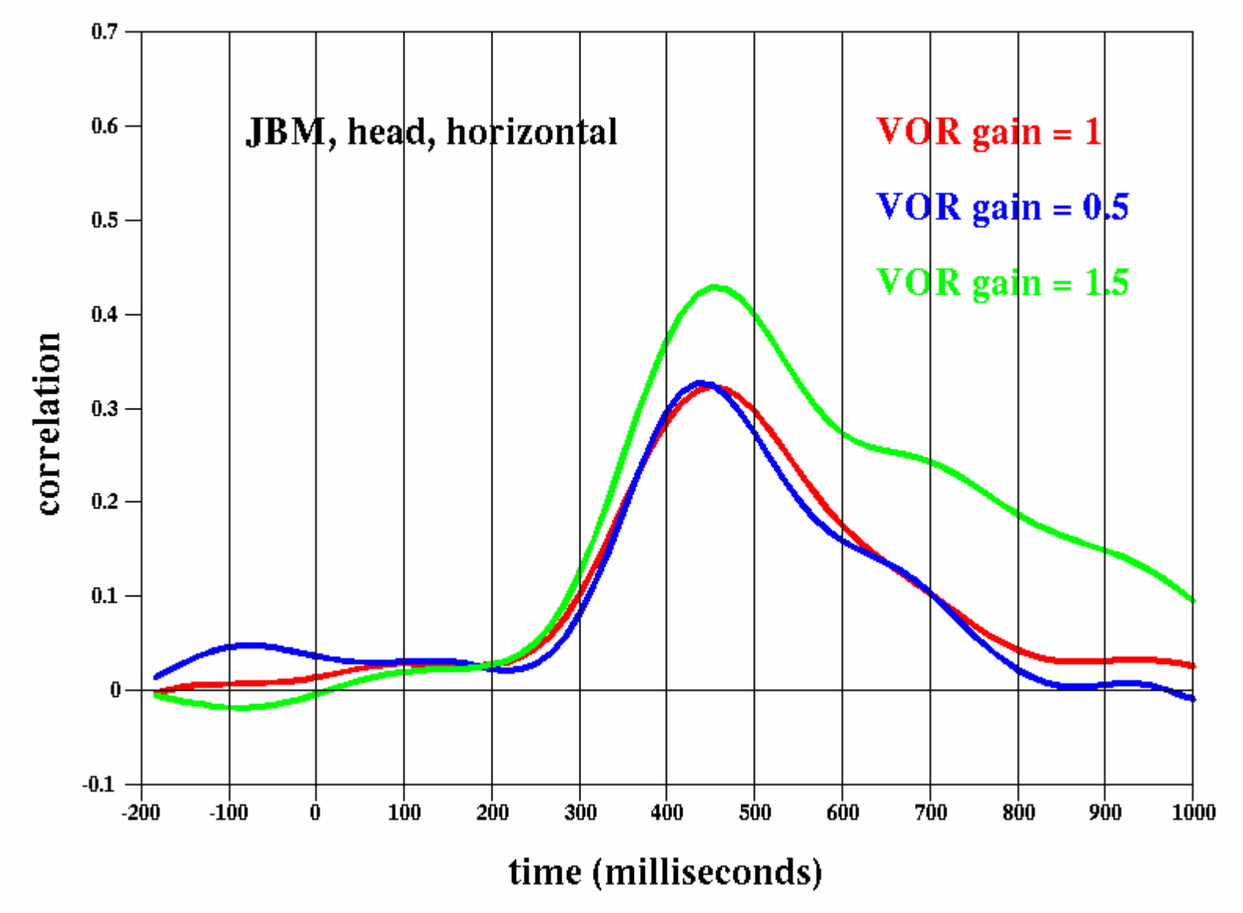
JBM, head, horizontal
VOR gain = 1
VOR gain = 0.5
VOR gain = 1.5
correlation
time (milliseconds)

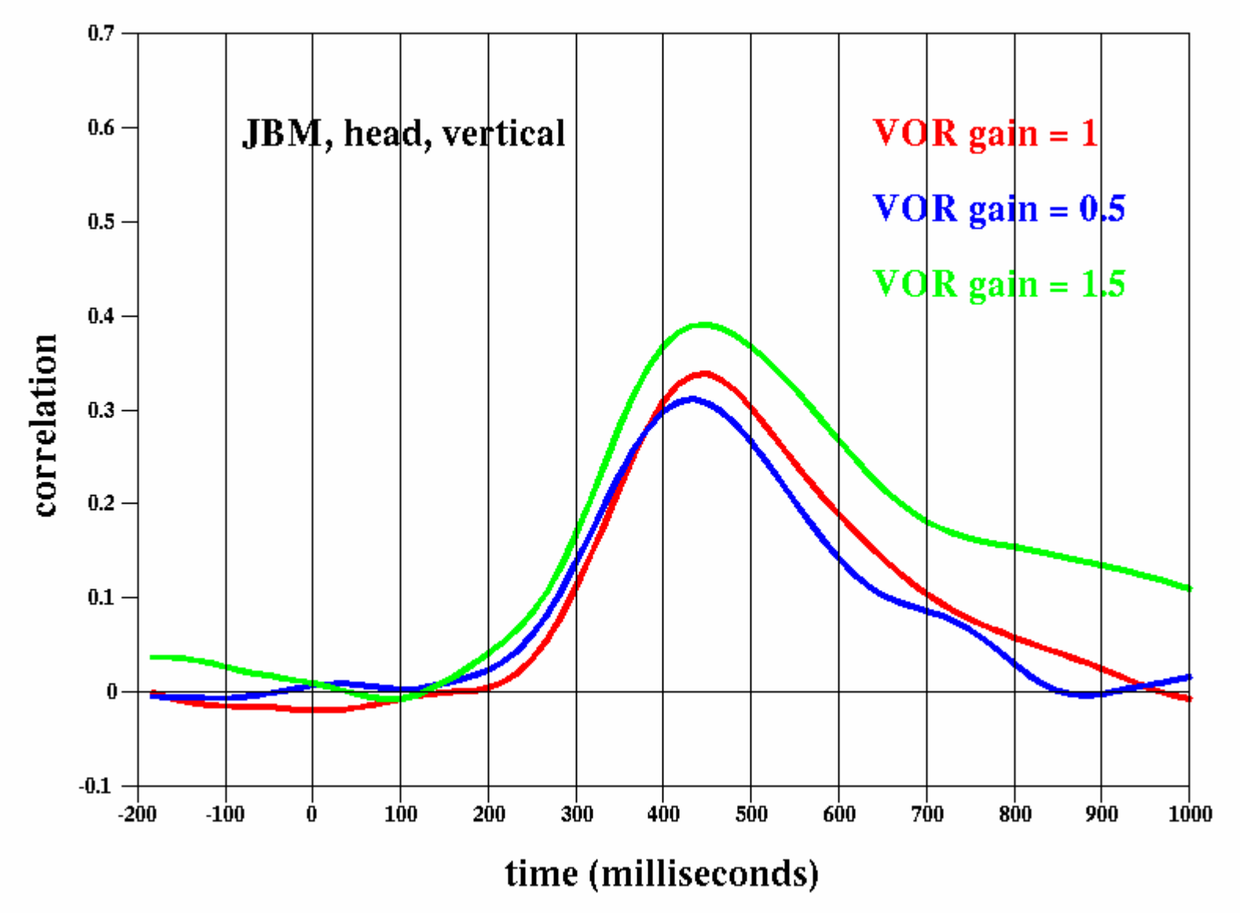
JBM, head, vertical
VOR gain = 1
VOR gain = 0.5
VOR gain = 1.5
correlation
time (milliseconds)

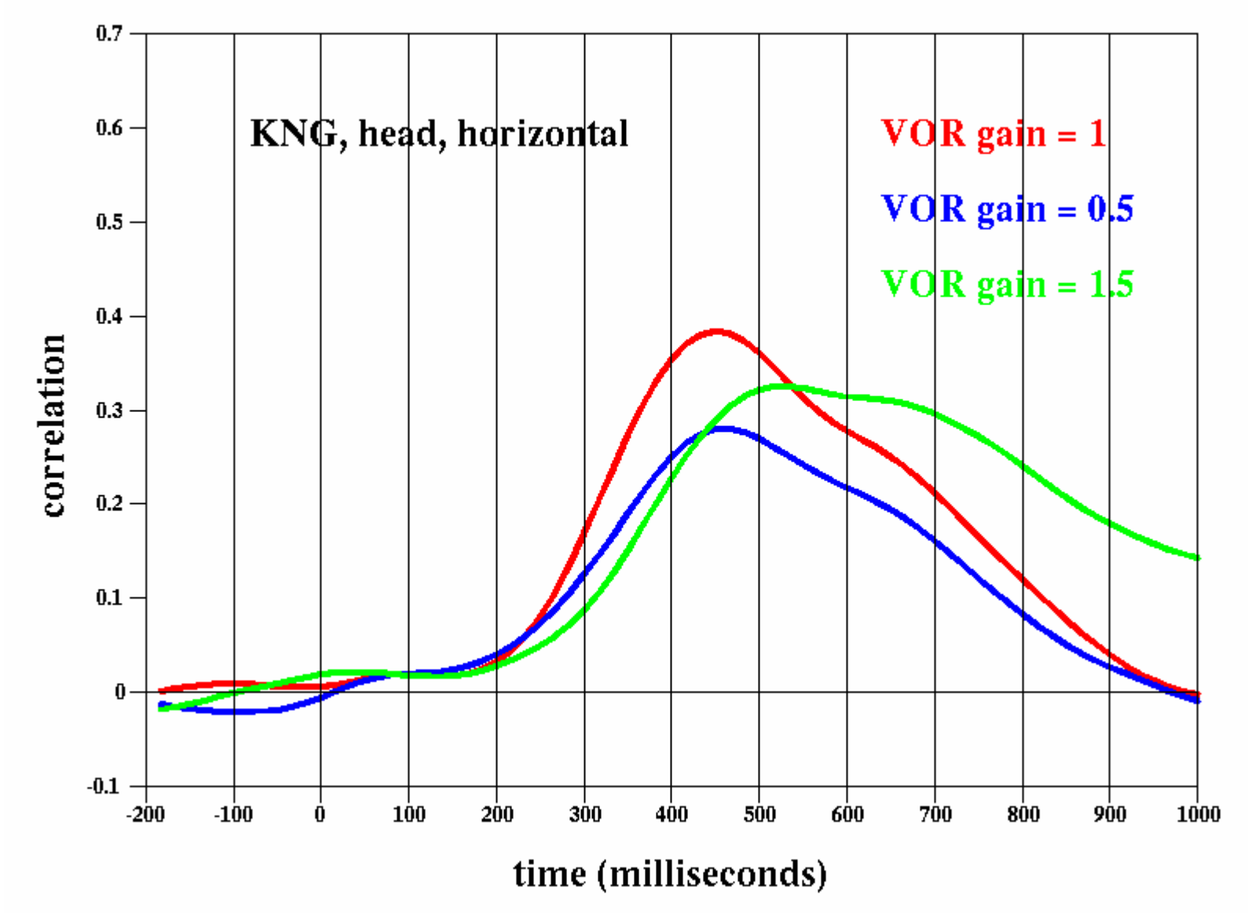
KNG, head, horizontal
VOR gain = 1
VOR gain = 0.5
VOR gain = 1.5
correlation
time (milliseconds)

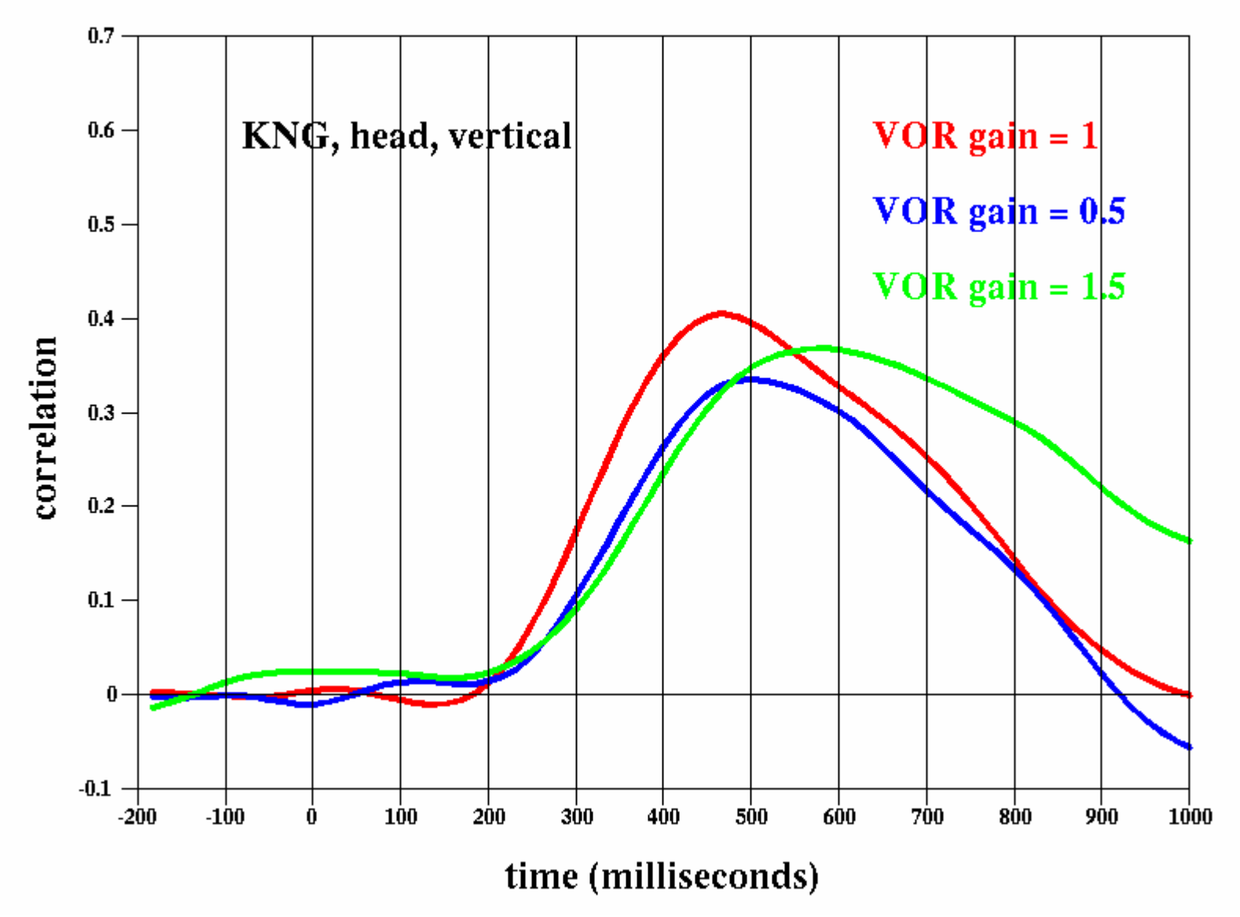
KNG, head, vertical
VOR gain = 1
VOR gain = 0.5
VOR gain = 1.5
correlation
time (milliseconds)

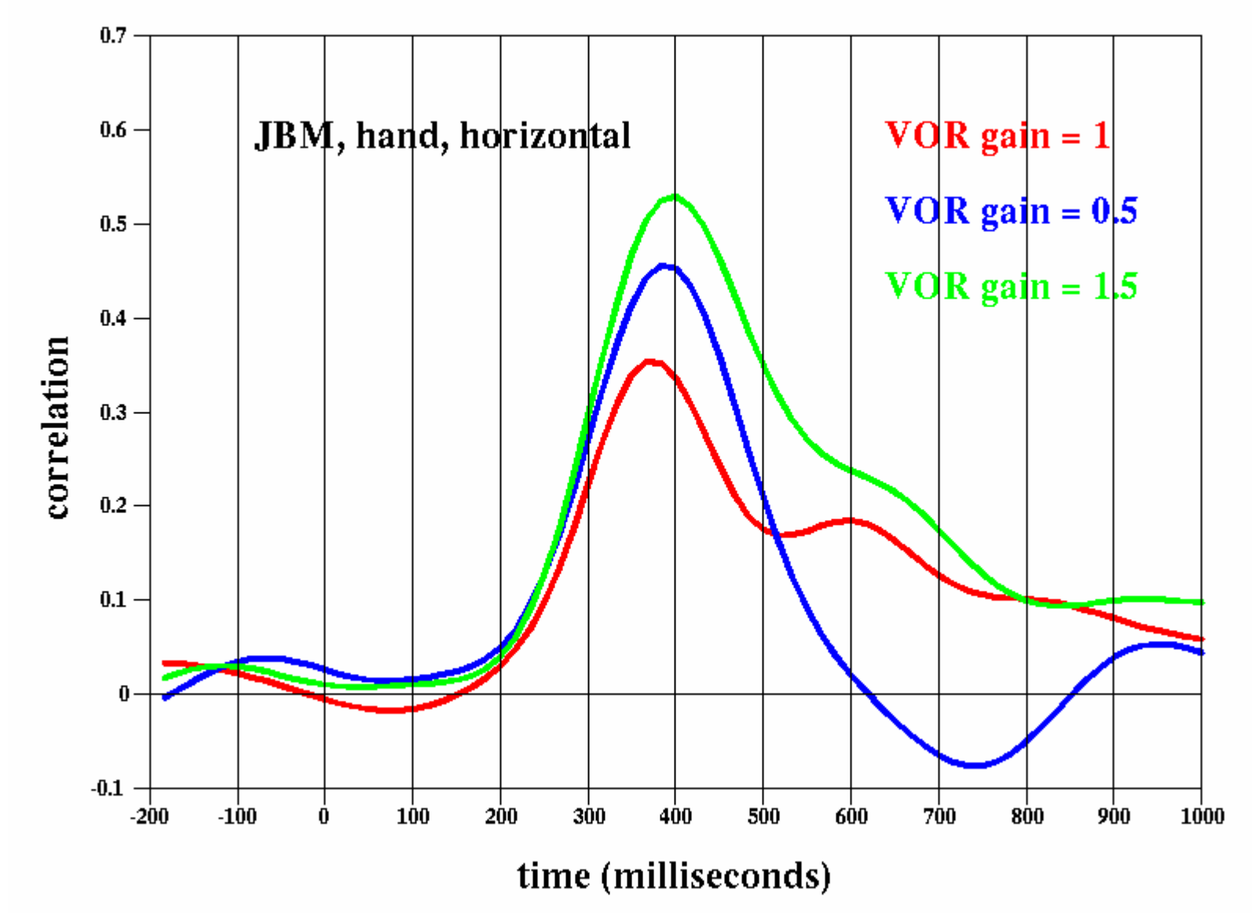
JBM, hand, horizontal
VOR gain = 1
VOR gain = 0.5
VOR gain = 1.5
correlation
time (milliseconds)

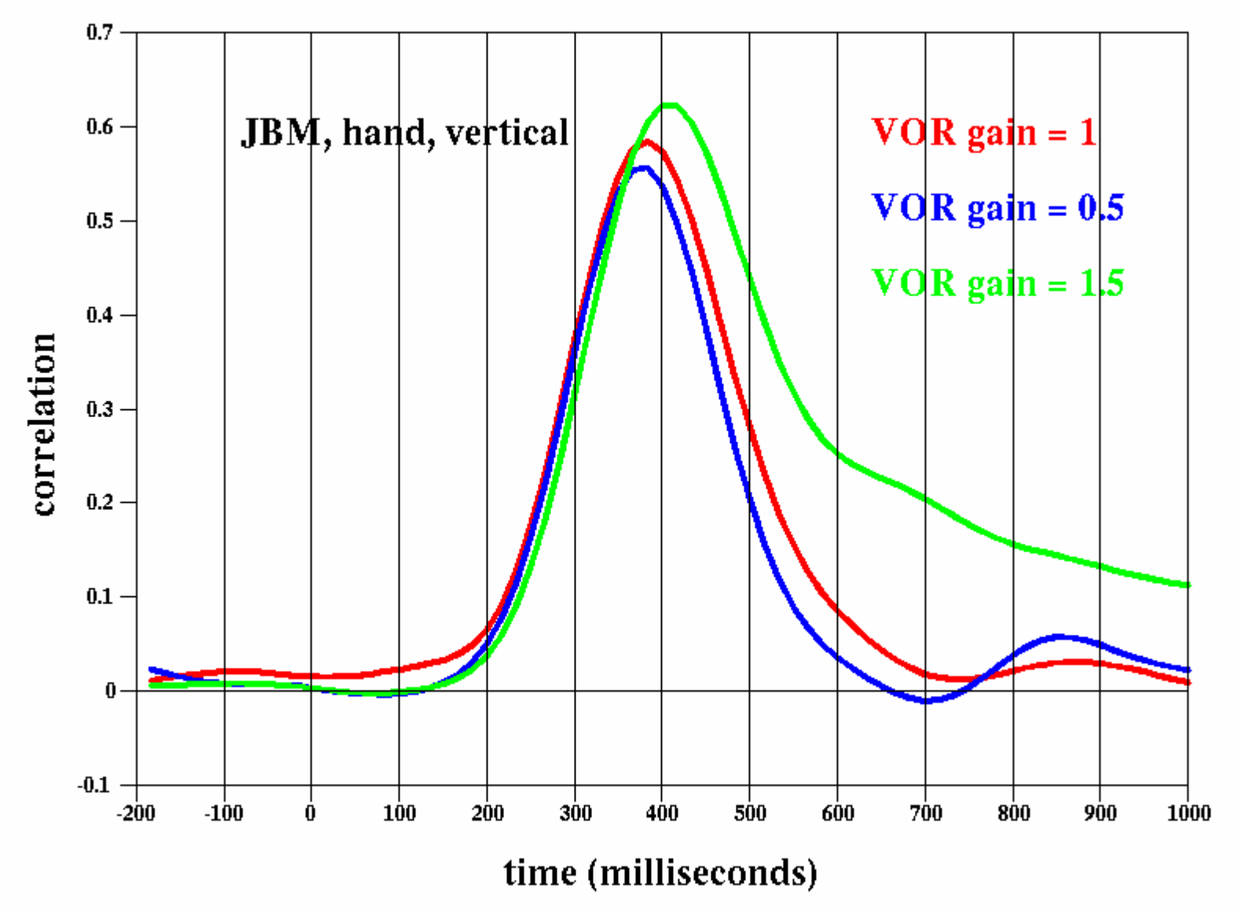
JBM, hand, vertical
VOR gain = 1
VOR gain = 0.5
VOR gain = 1.5
correlation
time (milliseconds)

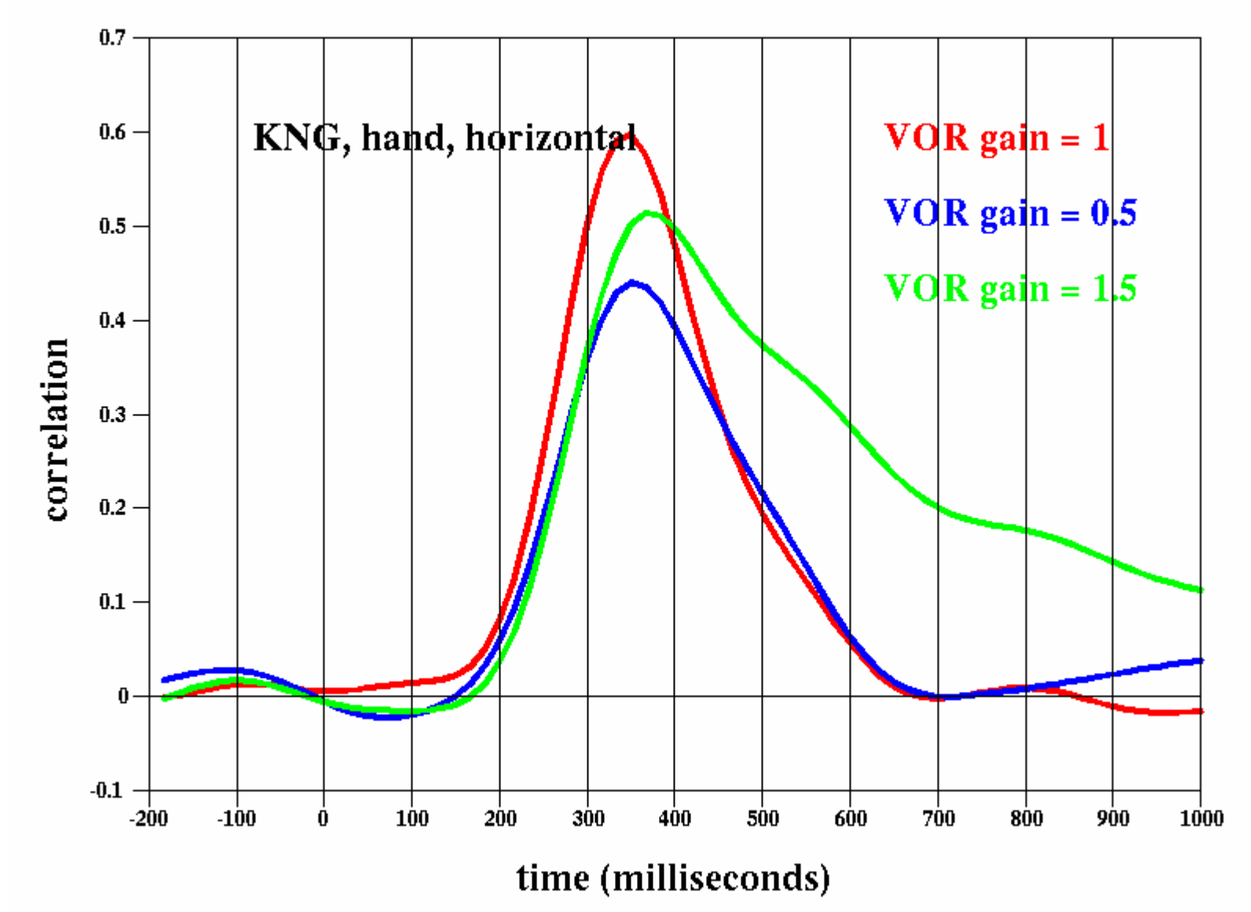
KNG, hand, horizontal
VOR gain = 1
VOR gain = 0.5
VOR gain = 1.5
correlation
time (milliseconds)

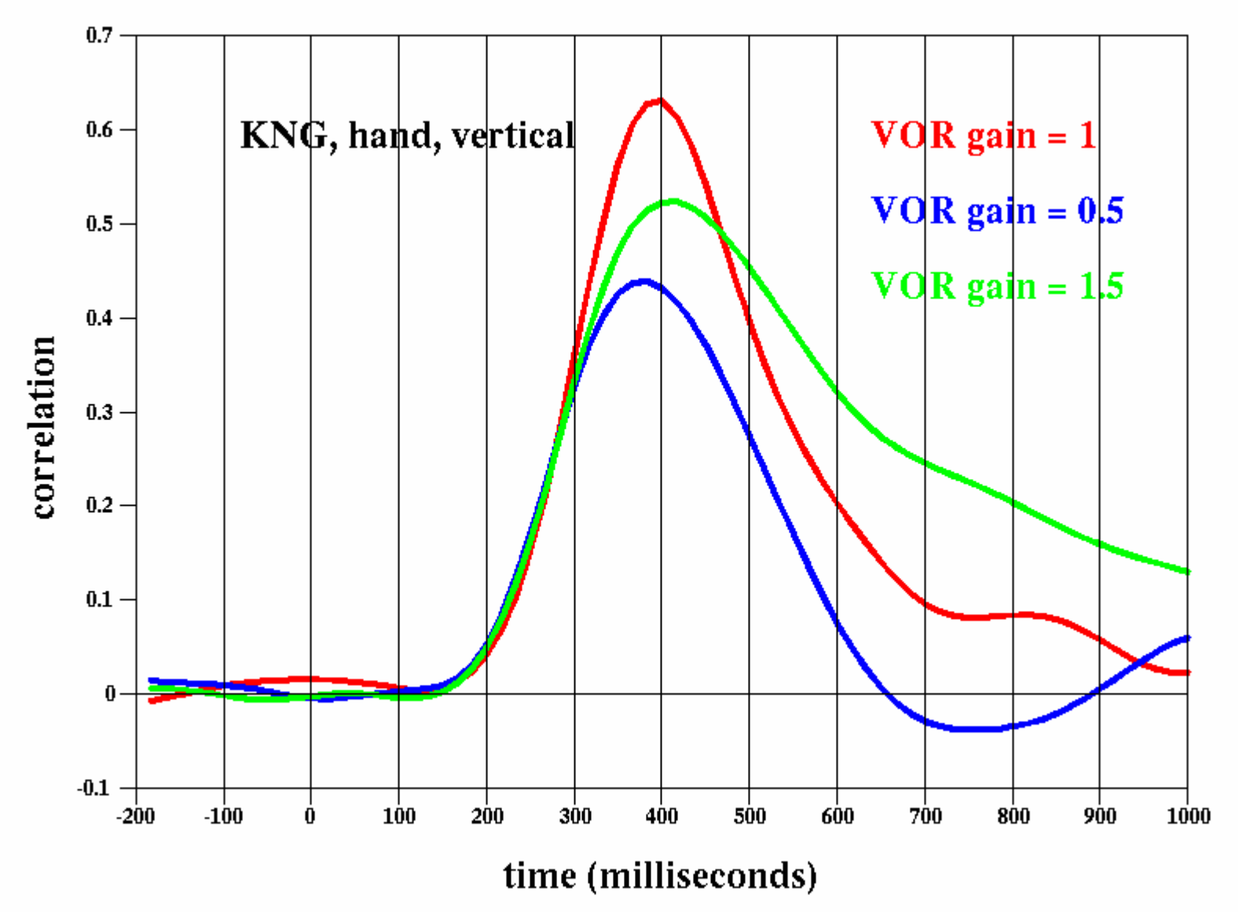
KNG, hand, vertical
VOR gain = 1
VOR gain = 0.5
VOR gain = 1.5
correlation
time (milliseconds)

Incorporation of delayed feedback

In our earlier work, we observed spontaneous oscillations which arose when the eye attempted to track delayed versions of small fixation errors. Here, for the first time, we incorporate delayed feedback with the random motion tracking task.

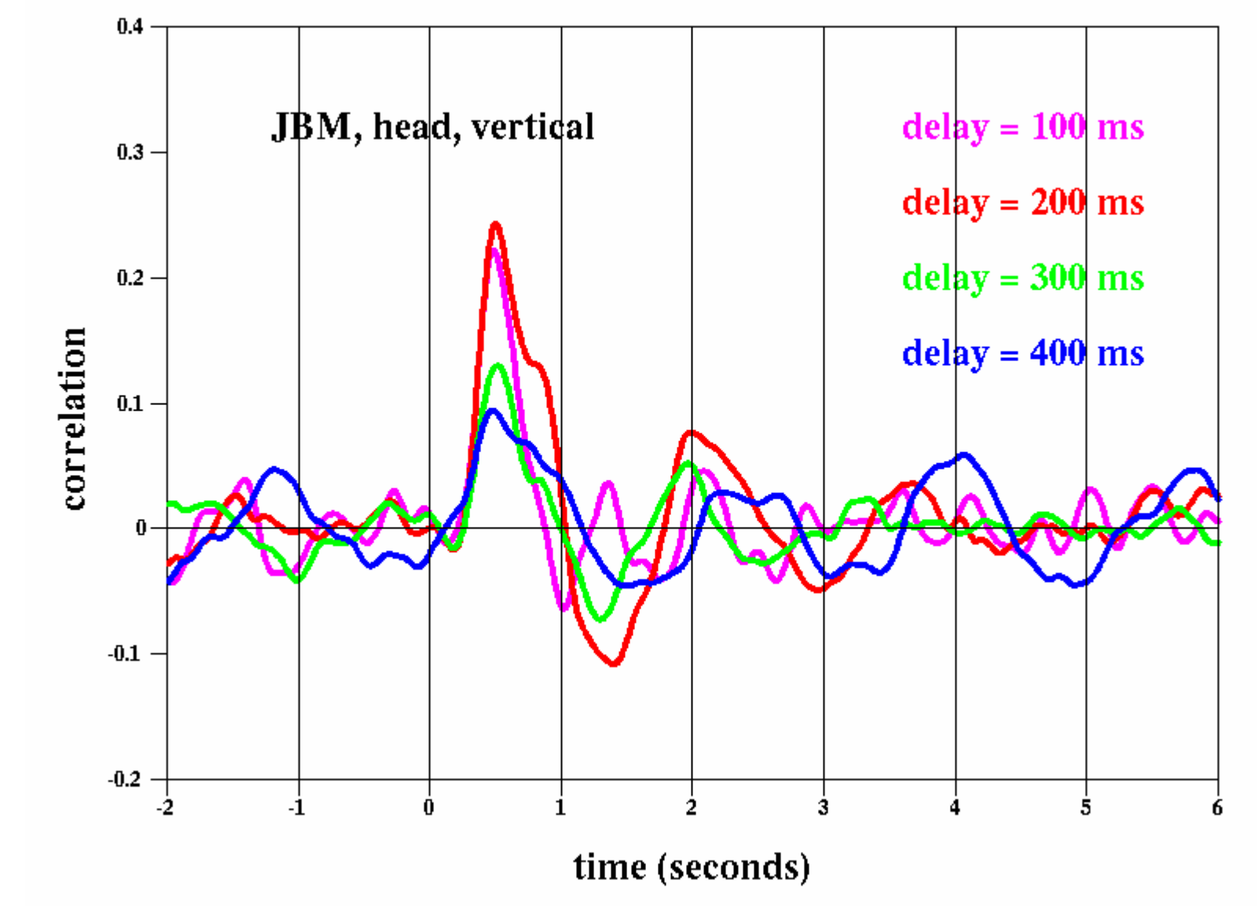
JBM, head, vertical
delay = 100 ms
delay = 200 ms
delay = 300 ms
delay = 400 ms
correlation
time (seconds)

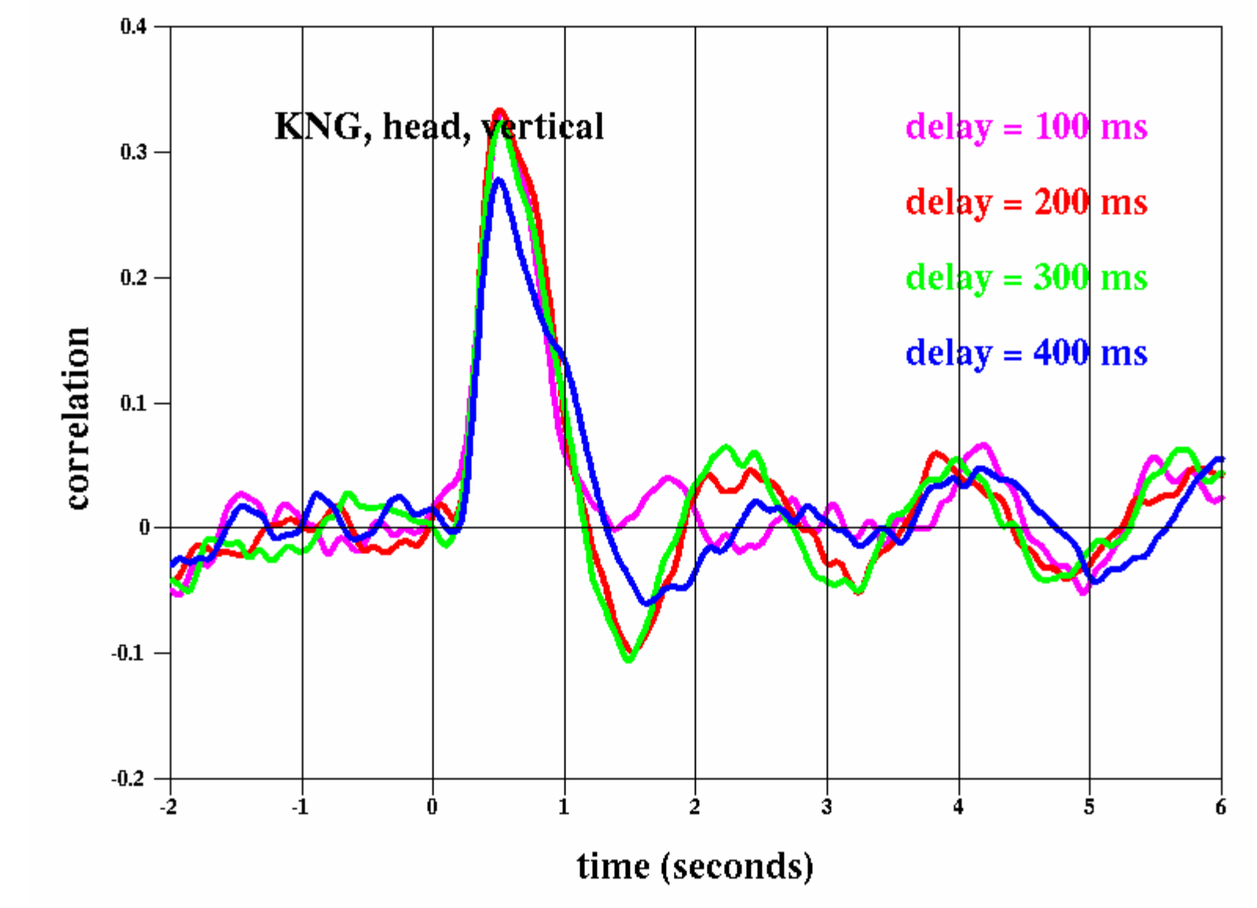
KNG, head, vertical
delay = 100 ms
delay = 200 ms
delay = 300 ms
delay = 400 ms
correlation
time (seconds)

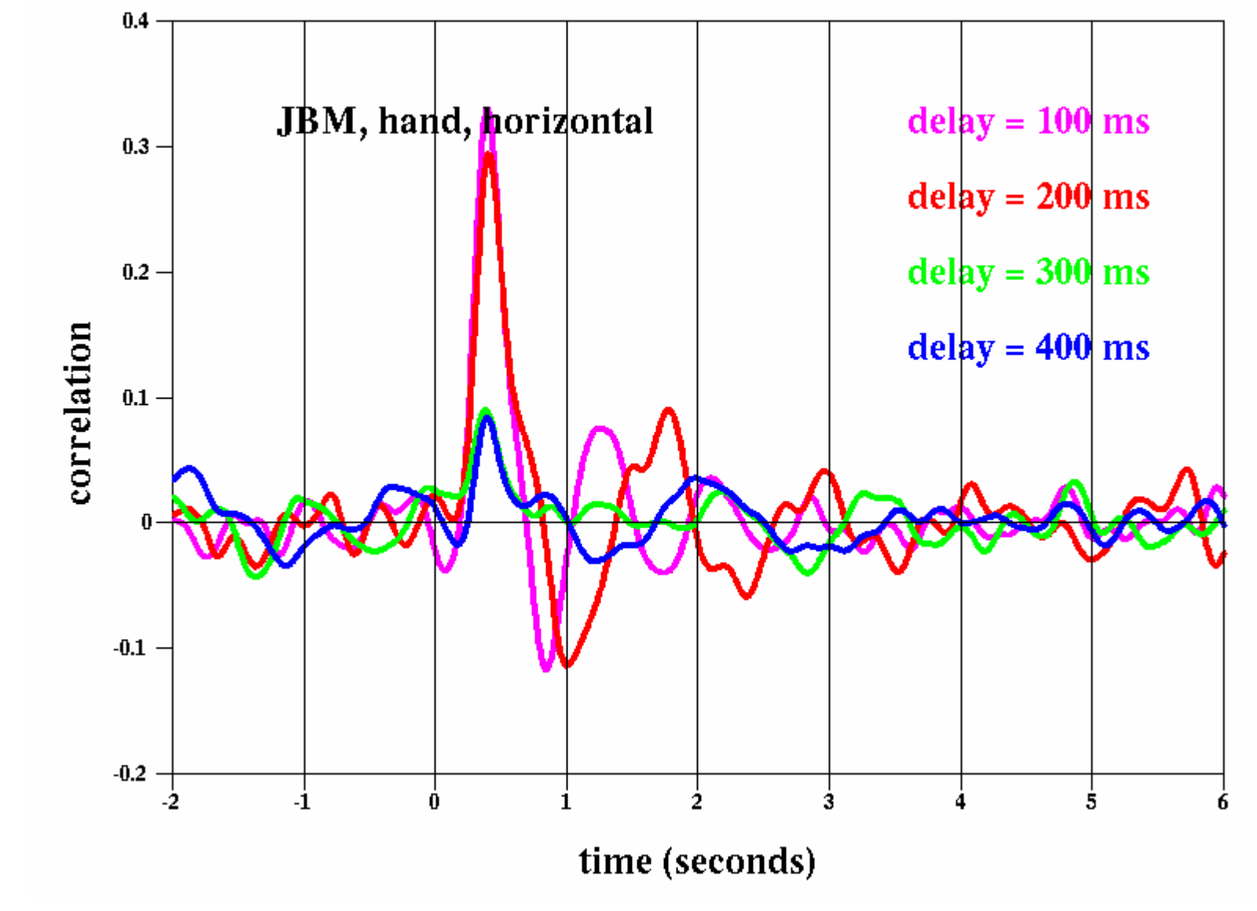
JBM, hand, horizontal
delay = 100 ms
delay = 200 ms
delay = 300 ms
delay = 400 ms
correlation
time (seconds)

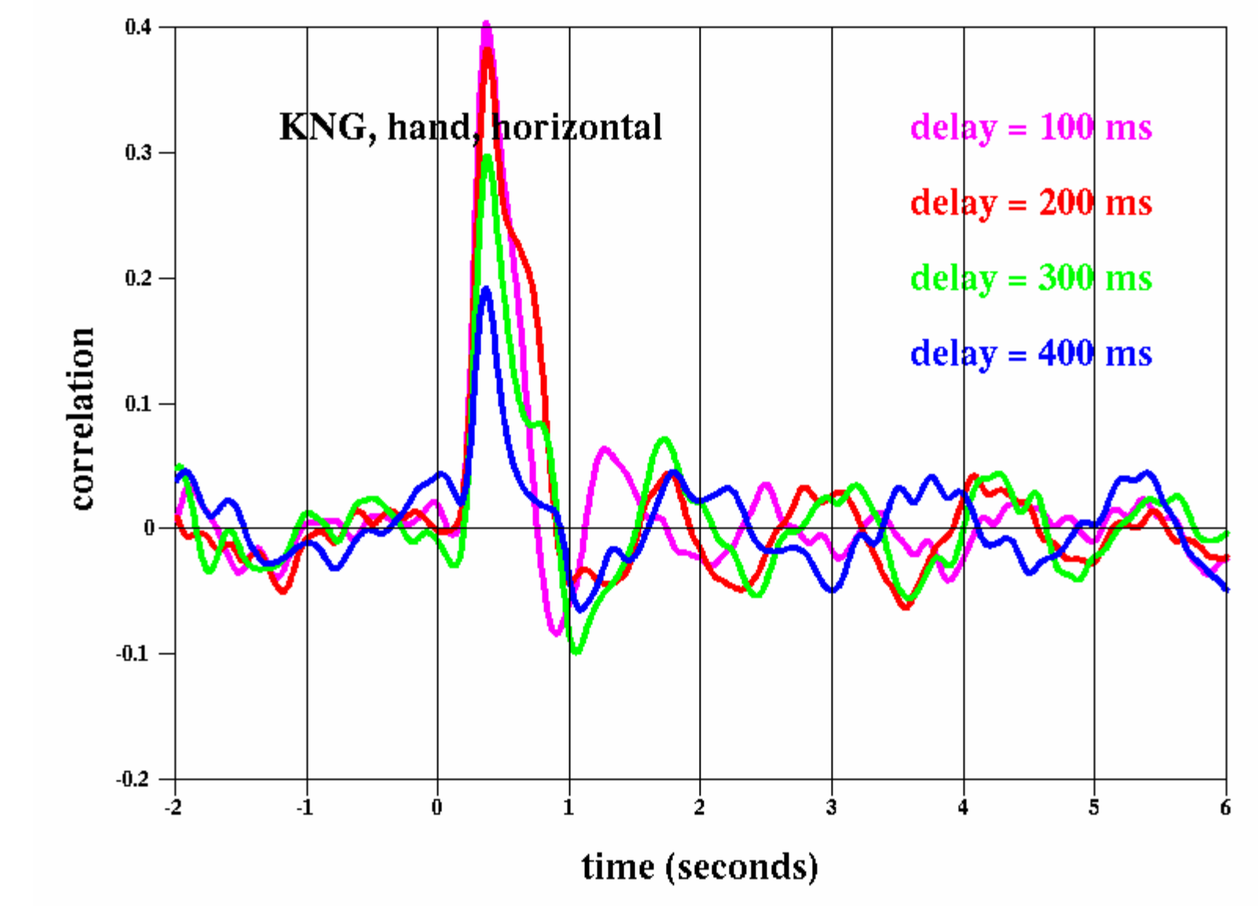
KNG, hand, horizontal
delay = 100 ms
delay = 200 ms
delay = 300 ms
delay = 400 ms
correlation
time (seconds)

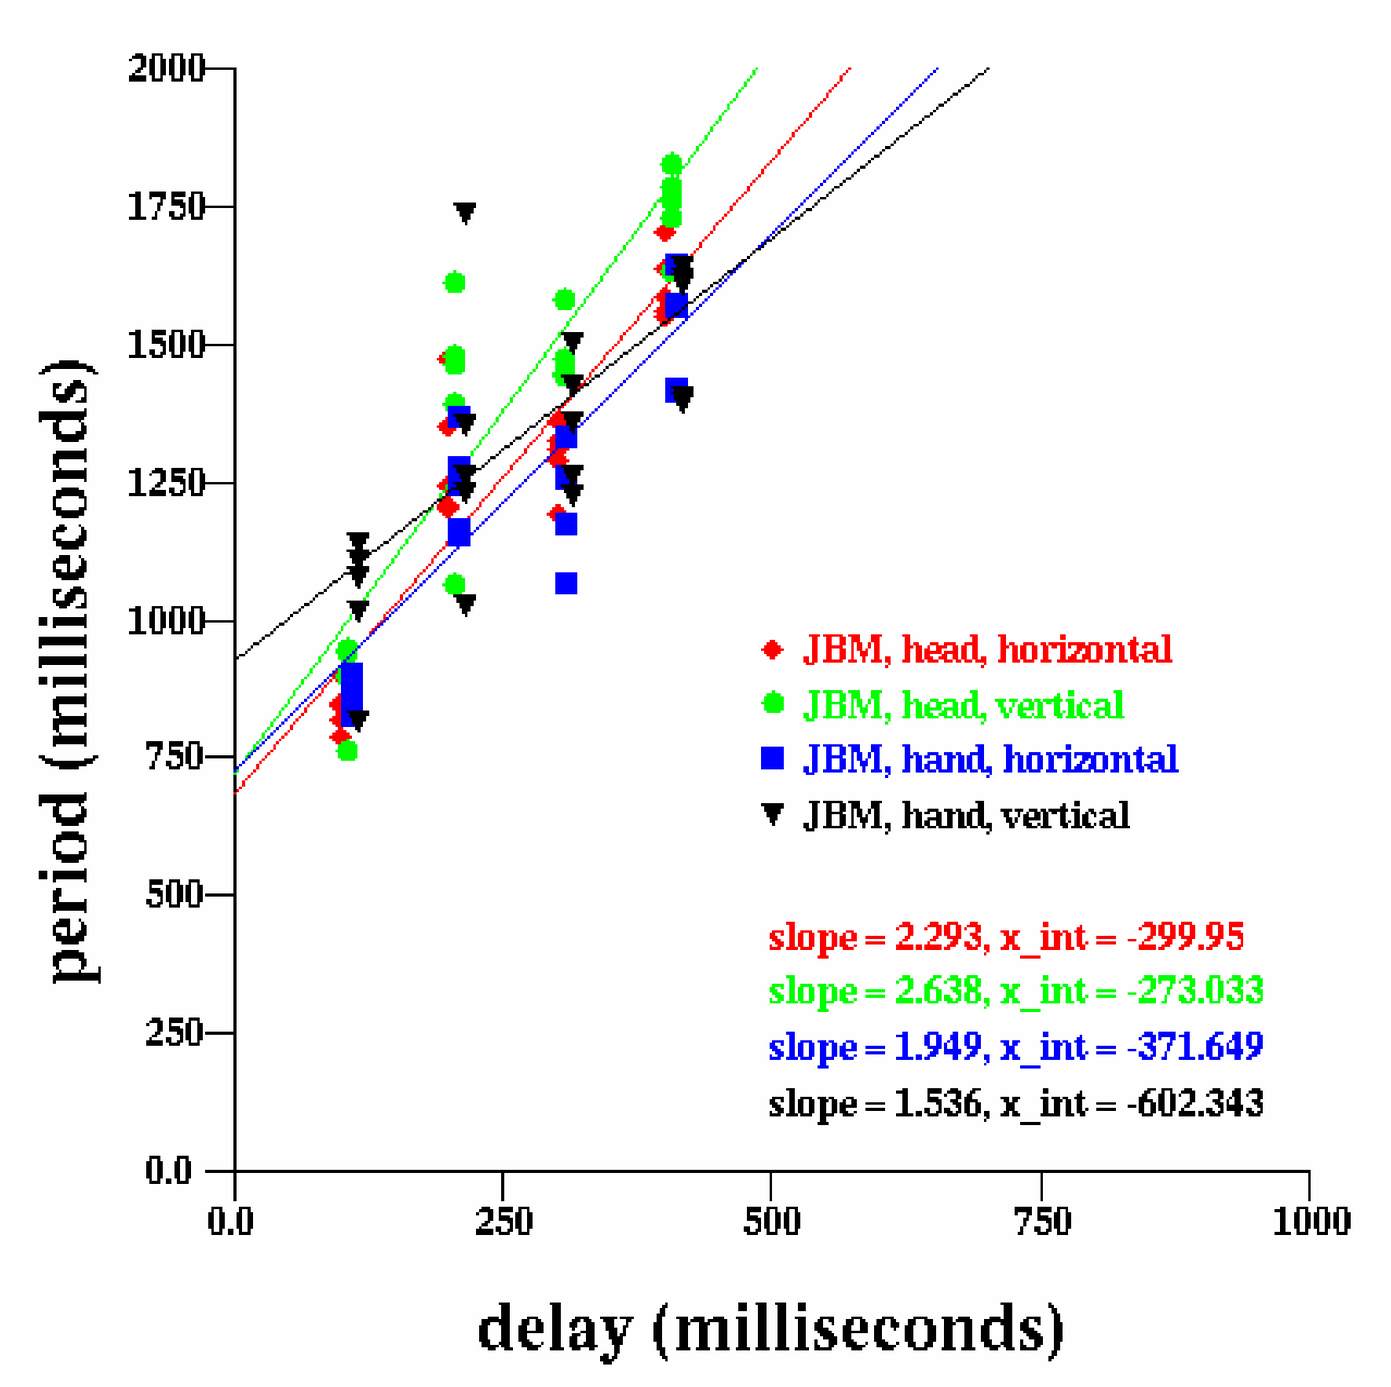
JBM, head, horizontal
JBM, head, vertical
JBM, hand, horizontal
JBM, hand, vertical
slope = 2.293, x_int = -299.95
slope = 2.638, x_int = -273.033
slope = 1.949, x_int = -371.649
slope = 1.536, x_int = -602.343
period (milliseconds)
delay (milliseconds)

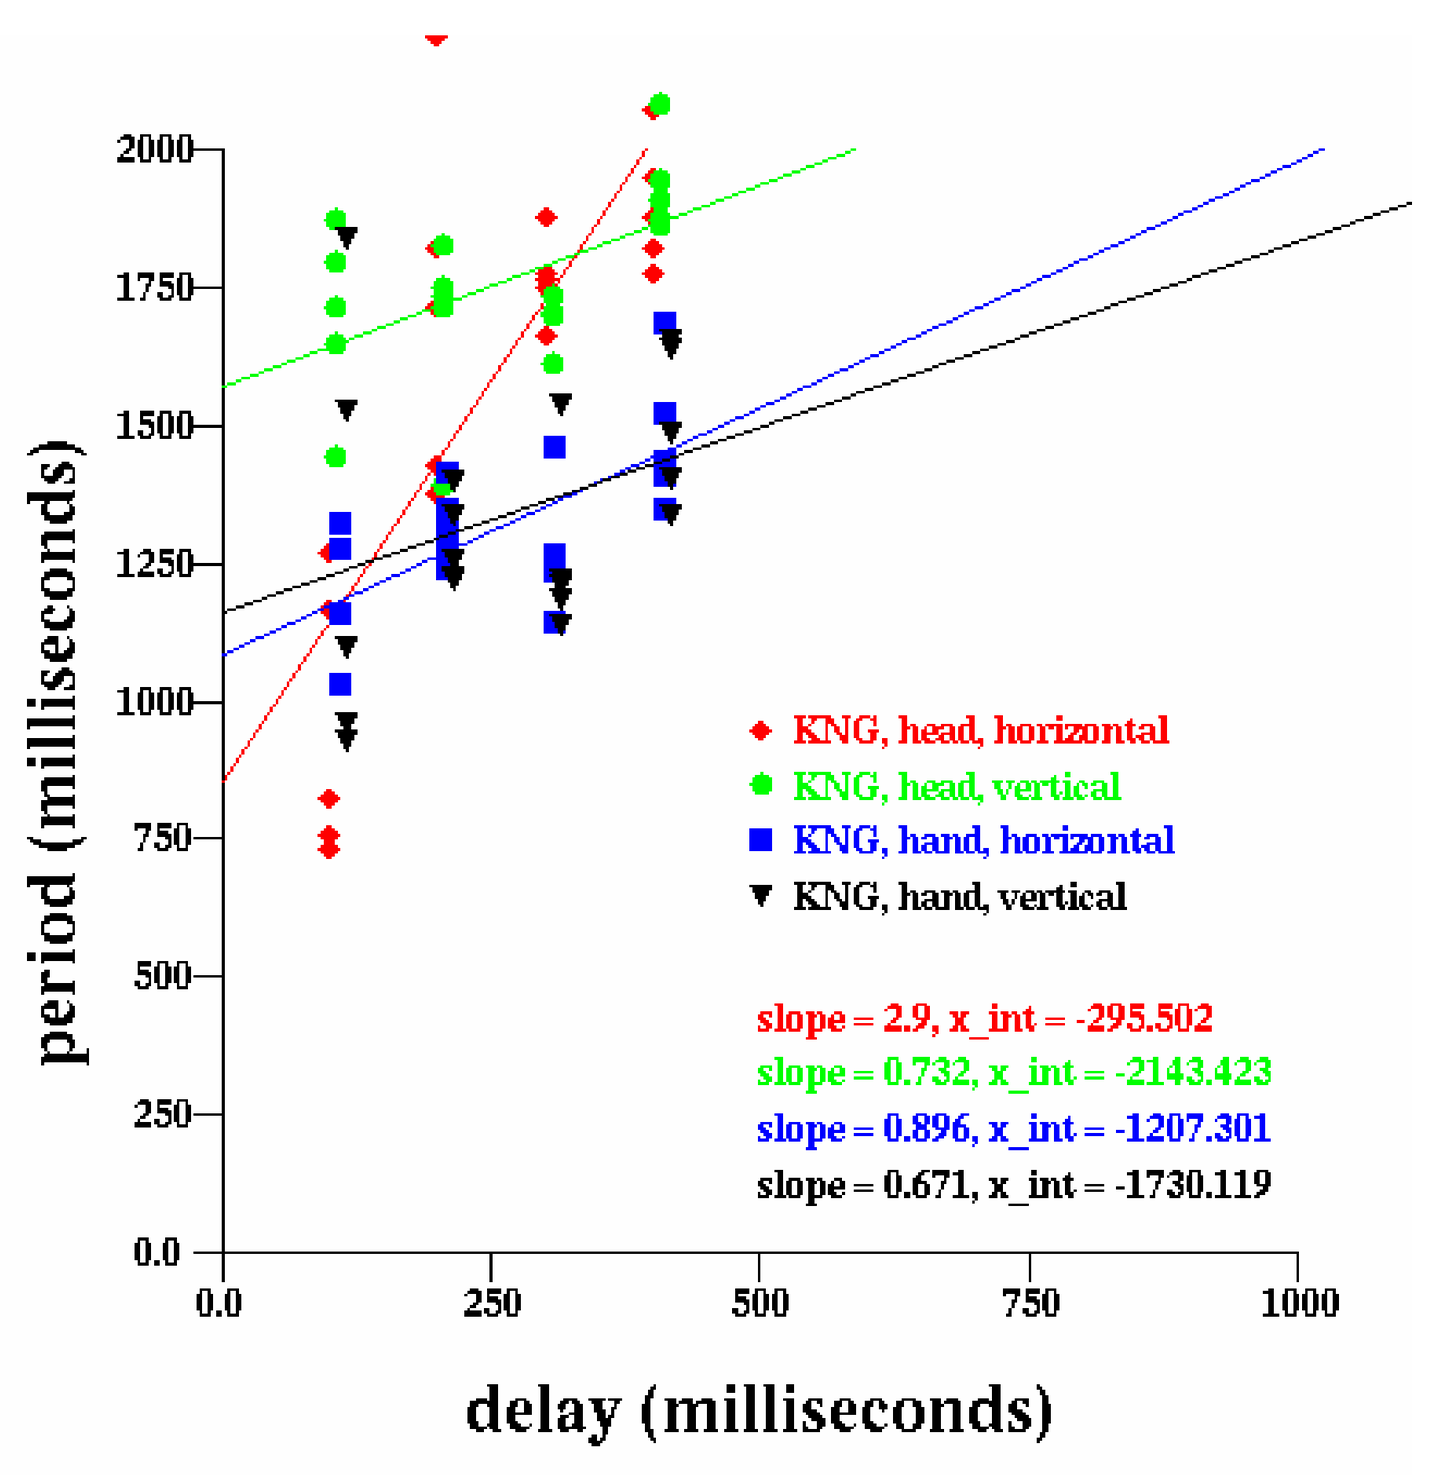
KNG, head, horizontal
KNG, head, vertical
KNG, hand, horizontal
KNG, hand, vertical
slope = 2.9, x_int = -295.502
slope = 0.732, x_int = -2143.423
slope = 0.896, x_int = -1207.301
slope = 0.671, x_int = -1730.119
period (milliseconds)
delay (milliseconds)

Acknowledgments: Supported by NASA's Aviation Safety and Airspace Systems programs. Thanks to Kevin Gabayan for assistance with data collection.